Bria-OTS+: A Versatile Therapeutic Platform for Inducing Anti-Cancer Immunity

Miguel Lopez-Lago[1], Pravin Kesarwani[1], Vikas Bhardwaj[1], Xiaoyi Zheng[1], Sagarika Pachhal[1], Patience Cournoo[1], Renee Cortez[1], George Woodfield[1], Charles L. Wiseman[1], William V. Williams[1]

[1]BriaCell Therapeutics Corp. Philadelphia, PA

AACR 2025 - # 3553

BACKGROUND AND OBJECTIVES

Despite advances, achieving curative cancer therapies remains challenging due to the complex, multi-stage nature of the disease. Recently, therapeutic cancer vaccines have shown promising efficacy, driven by advances in immune profiling, antigen selection, immunotherapy combinations, mRNA technology, and delivery platforms. However, challenges persist, including identifying highly immunogenic tumor antigens, overcoming T-cell immune escape, and achieving scalable, cost-effective manufacturing.

To address these, BriaCell is developing a gene-modified allogeneic tumor cell platform to stimulate both innate and adaptive immunity. This platform provides a broad range of tumor antigens and includes immune-stimulatory factors to induce a comprehensive immune response. Initial results with the SV-BR-1-GM (Bria-IMT) breast cancer cell line, expressing GM-CSF, have shown promise. Bria-OTS+ further enhances immune activation by expressing additional cytokines, co-stimulatory factors, and HLA alleles across tumor cells, offering a personalized, adaptable immunotherapy platform.

METHODS

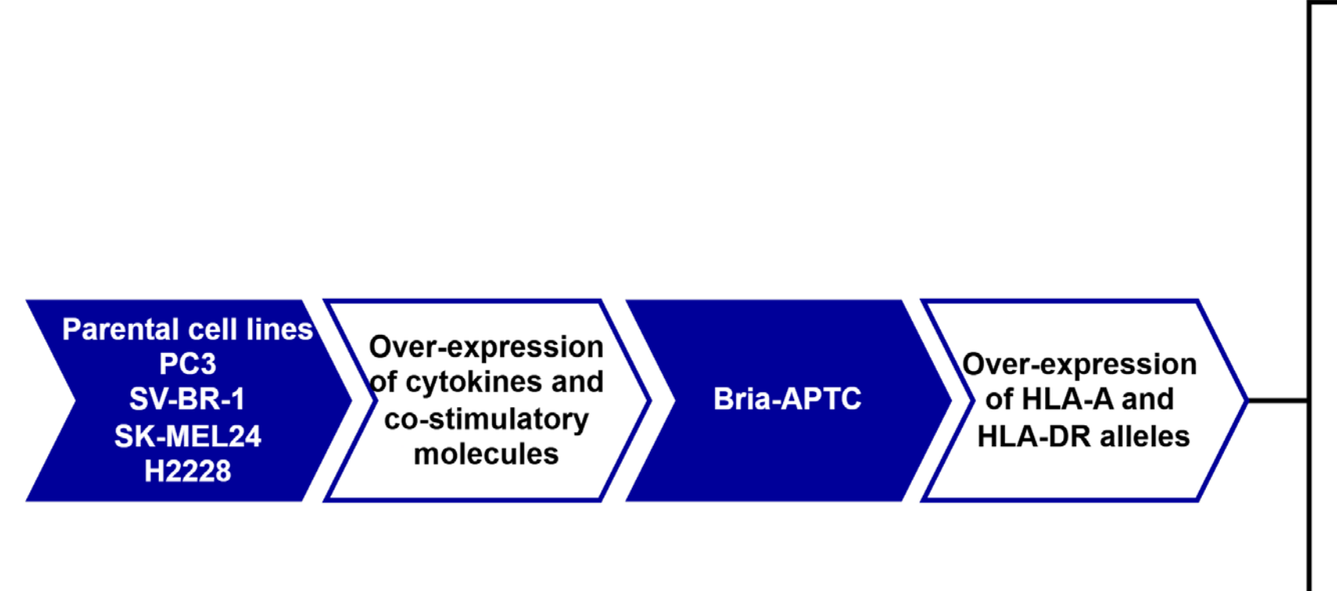
Parental cell lines
PC3
SV-BR-1
SK-MEL24
H2228
Over-expression of cytokines and co-stimulatory molecules
Bria-APTC
Over-expression of HLA-A and HLA-DR alleles

	HLA class I	HLA class II	Co-stimulatory molecules	Cytokines
Bria-PROS1+ Bria-BRES1+ Bria-MEL1+ Bria-LUNG1+	A*01:01, A*68:01	DRB3*02:02, DRB5*01:01	CD40 (only Bria-MEL1+ and Bria-LUNG1+), CD80, CD86	GM-CSF, IFNa, IL7 IL12
Bria-PROS2+ Bria-BRES2+ Bria-MEL2+ Bria-LUNG2+	A*02:01, A*11:01	DRB4*01:01, DRB3*01:01	CD40 (only Bria-MEL2+ and Bria-LUNG2+), CD80, CD86	GM-CSF, IFNa, IL7 IL12
Bria-PROS3+ Bria-BRES3+ Bria-MEL3+ Bria-LUNG3+	A*03:01, A*23:01	DRB5*01:02, DRB3*03:01	CD40 (only Bria-MEL3+ and Bria-LUNG3+), CD80, CD86	GM-CSF, IFNa, IL7 IL12
Bria-PROS4+ Bria-BRES4+ Bria-MEL4+ Bria-LUNG4+	A*24:02, A*33:03	DRB5*02:02, DRB3*01:01	CD40 (only Bria-MEL4+ and Bria-LUNG4+), CD80, CD86	GM-CSF, IFNa, IL7 IL12

Engineering of Bria-OTS+ Cell Lines: *The Bria-OTS+ cell lines were developed from various types of cancer, including breast cancer (SV-BR-1), prostate cancer (PC3), melanoma (SK-MEL-24), and lung cancer (NCI-H2228), selected for their expression of a unique gene immune signature initially characterized in SV-BR-1 cells. These cell lines were further enhanced to boost their antigen presentation ability and stimulate the immune response by genetically engineering them to express co-stimulatory molecules and immunomodulatory cytokines, resulting in the formation of antigen presenting tumor cells (APTC). Furthermore, to create a semi-allogeneic cell therapy with extensive applicability, the HLA allele expression repertoire of SV-BR-1 was broadened. Population analysis indicated that a configuration of four cell lines, each bearing a combination of two HLA-A and two HLA-DRB3/4/5 alleles, could potentially provide at least one HLA match for 99% of the population. This includes a 92% probability of matching at Class I HLA-A alleles and a 98% likelihood at Class II HLA-DRB3/4/5 alleles.*

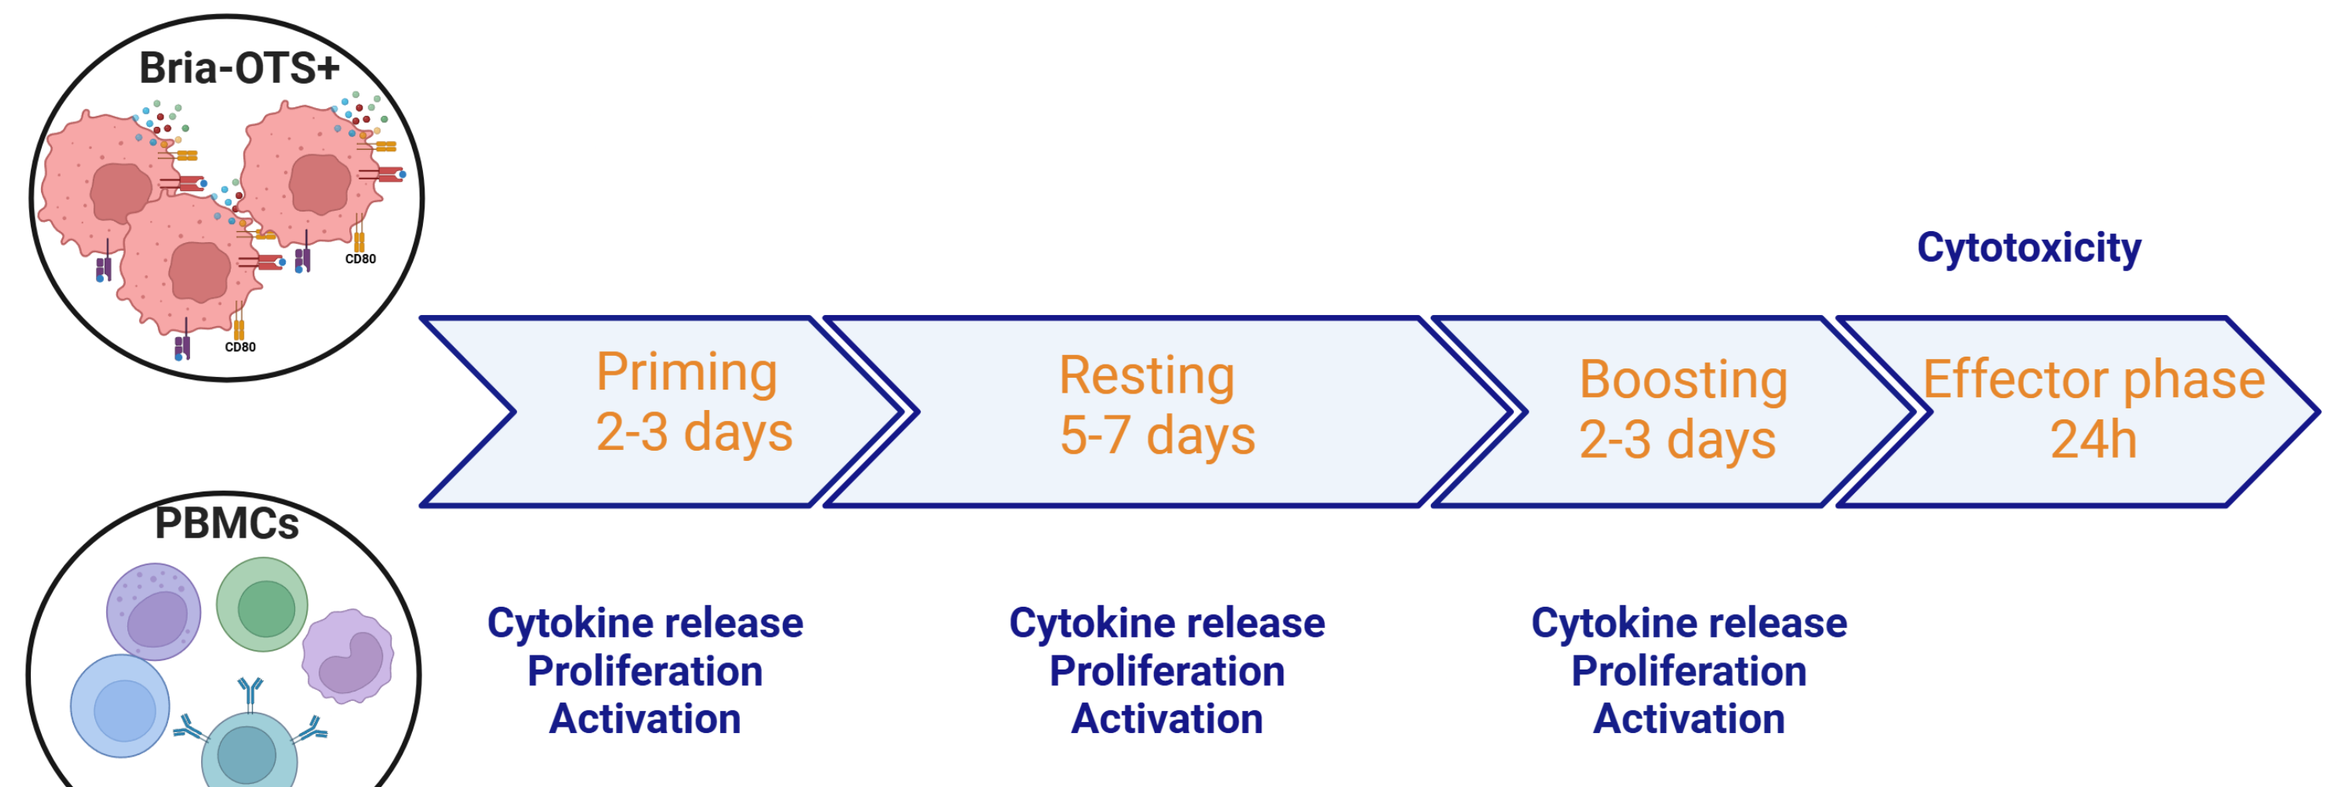
Bria-OTS+
PBMCs
Cytotoxicity
Priming 2-3 days
Resting 5-7 days
Boosting 2-3 days
Effector phase 24h
Cytokine release Proliferation Activation
Cytokine release Proliferation Activation
Cytokine release Proliferation Activation

In vitro vaccination assay using Bria-OTS+ cells. *Bria-OTS+ cells, are co-cultured with peripheral blood mononuclear cells (PBMCs) in an* ***in vitro vaccination*** *assay to induce an anti-tumor immune response. The activation protocol consists of four sequential phases: (1)* ***Priming (2–3 days)****, where PBMCs initially interact with Bria-OTS+ cells, leading to cytokine release (primarily IFNγ, IL-2), proliferation, and activation of immune cells; (2)* ***Resting (5–7 days)****, allowing expansion and maturation of activated T cells and other immune populations; (3)* ***Boosting (2–3 days)****, where a second exposure to Bria-OTS+ cells further enhances immune activation and expands tumor-specific T cell populations; and (4)* ***Effector phase (24 hours)****, where cytotoxic activity against target tumor cells is assessed through cell killing assays. Throughout the process, immune activation is characterized by cytokine release, proliferation, and cellular activation markers. The final effector phase evaluates the cytotoxic potential of the activated immune cells, primarily CD8+ T cells and NK cells, against target tumor cells.*

RESULTS

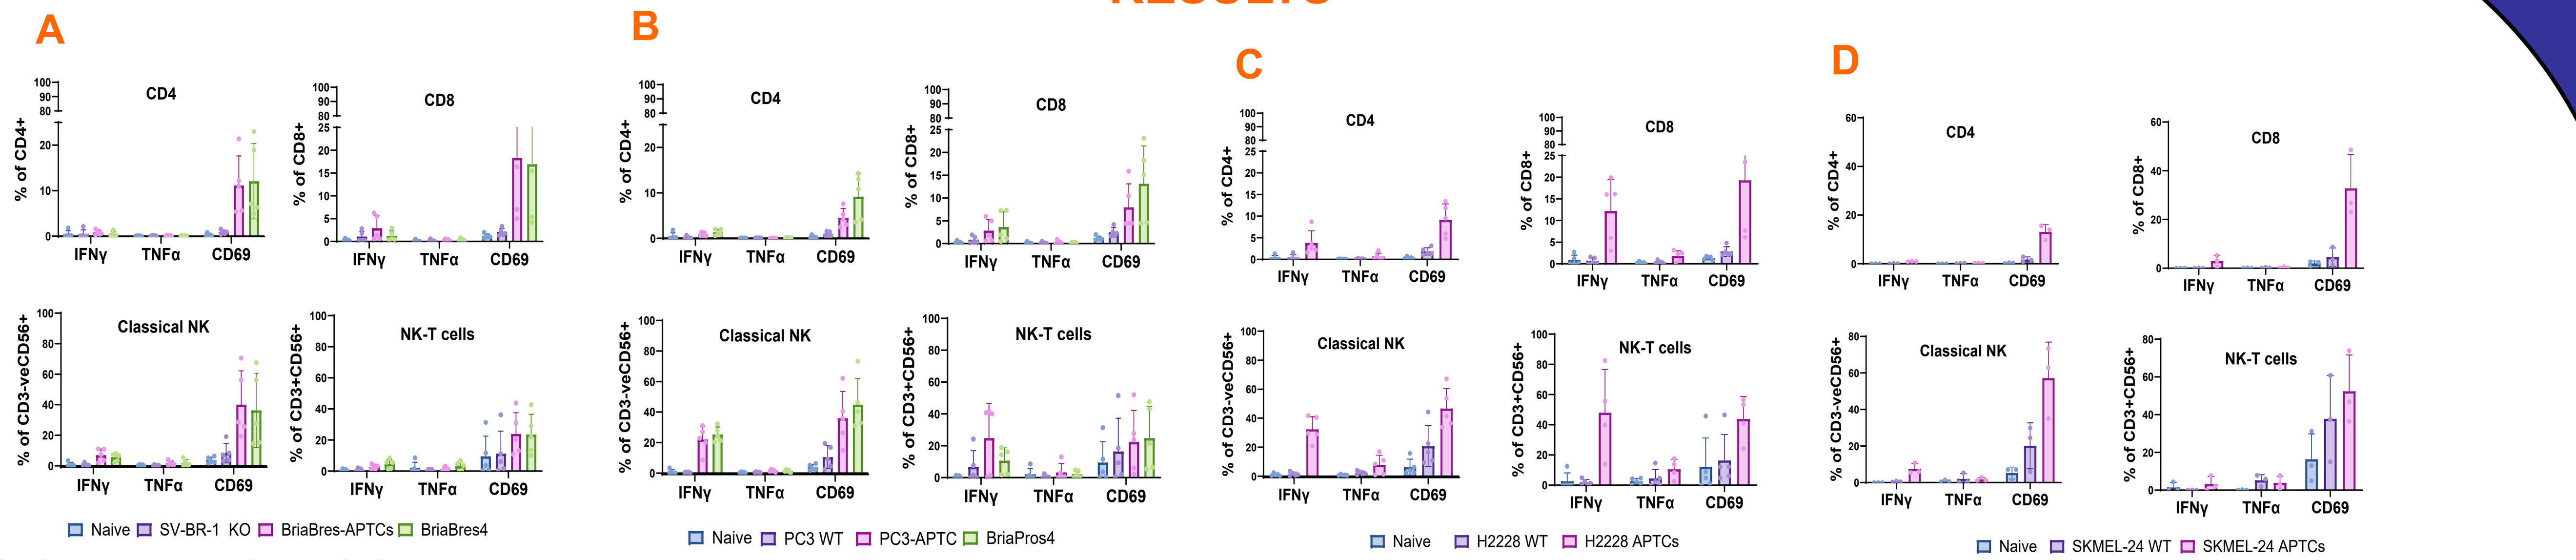
A
B
C
D
CD4
CD8
Classical NK
NK-T cells
IFNγ TNFα CD69
Naive SV-BR-1 KO BriaBres-APTCs BriaBres4
Naive PC3 WT PC3-APTC BriaPros4
Naive H2228 WT H2228 APTCs
Naive SKMEL-24 WT SKMEL-24 APTCs

Bria-OTS+ activates CD4+ & CD8+ T, NK and NKT cells in an in vitro vaccination assay *Irradiated SV-BR-1-KO (HLA-A and DR alleles knocked out), Bria-BRES4+ (A) PC3-WT, Bria-PROS4+ (B) H2228-WT, H2228 APTCs (C) or SK-MEL24 WT and SK-MEL24 APTCs (D) (10000 cells) were cocultured with 3-5 different donor PBMCs (E:T =10:1). After 48 hours of co-culture, immune cells were analyzed by flow cytometry with a variety of activation markers with the % positive shown.*

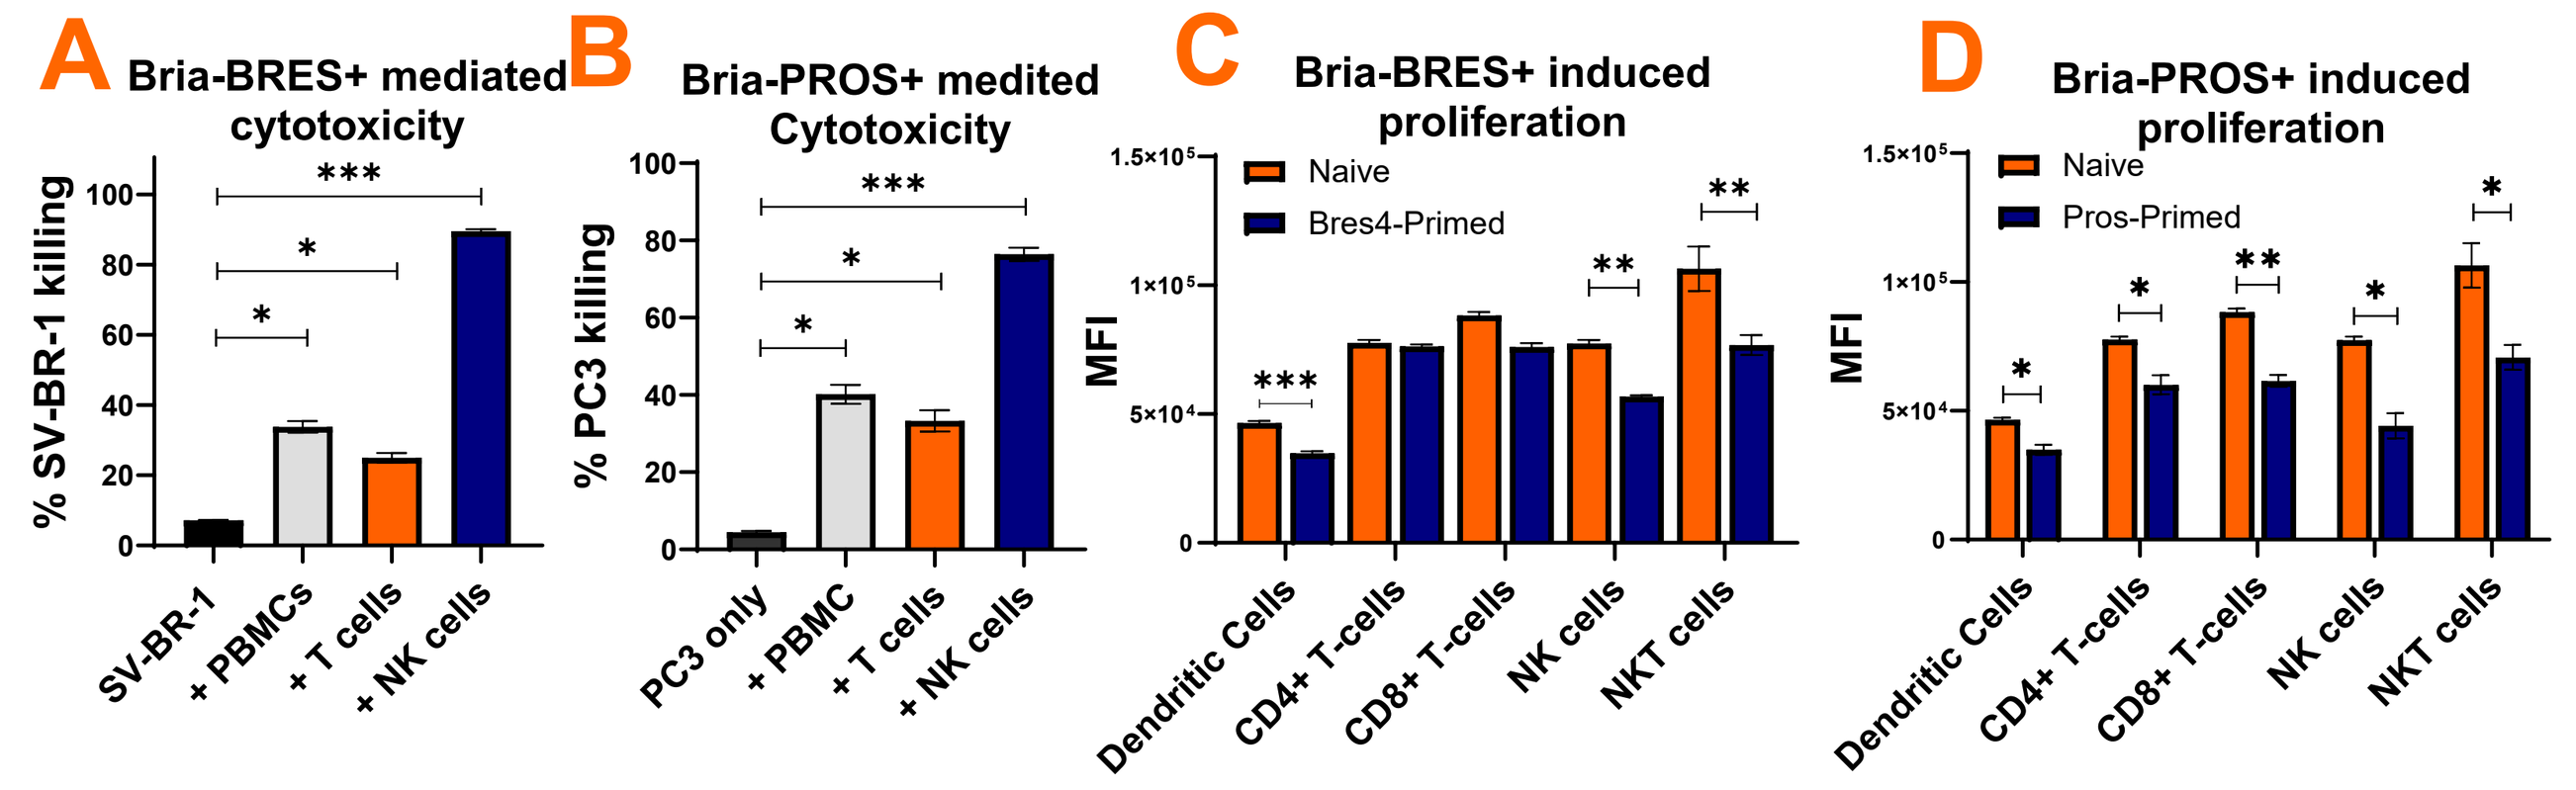
A Bria-BRES+ mediated cytotoxicity
% SV-BR-1 killing
SV-BR-1
+ PBMCs
+ T cells
+ NK cells
B Bria-PROS+ medited Cytotoxicity
% PC3 killing
PC3 only
+ PBMC
+ T cells
+ NK cells
C Bria-BRES+ induced proliferation
Naive
Bres4-Primed
D Bria-PROS+ induced proliferation
Naive
Pros-Primed
MFI
Dendritic Cells
CD4+ T-cells
CD8+ T-cells
NK cells
NKT cells

Bria-OTS+ Primes Immune Cells for Enhanced Proliferation and Tumor Cell Killing. (A-B) *Cytotoxic activity of PBMCs (3 donors) , isolated T cells, or NK cells primed with Bria-BRES4+ cells (A) or Bria-PROS4+ cells (B).One million irradiated Bria-PROS4+ or Bria-BRES4+ cells were used for the priming phase with PBMCs and/or isolated T or NK cells at an effector-to-target (E:T) ratio of 10:1 for 48 hours. The cytotoxic activity of the activated cells was assessed using an Annexin-V-PI kit after 16 hours of co-culture with PC3 or SV-BR-1 cells at a 4:1 E:T ratio. NK cell-mediated killing was significantly stronger than T-cell-mediated killing, suggesting a preferential activation of innate immune cytotoxic responses. Statistical significance: *p < 0.05, **p < 0.005, ***p < 0.0005. (C-D) Proliferation of Dendritic cells, CD8+ T cells, CD4+ T cells, NK and NKT cells following priming with Bria-BRES4+ cells (C) or Bria-PROS4+ cells (D). One million irradiated Bria-PROS4+ or Bria-BRES4+ cells were cultured with PBMCs (3 donors) at an E:T ratio of 10:1 for 24 hours. The PBMCs were stained with CellTrace Violet and allowed to proliferate for four days. Proliferation of dendritic cells, CD8+ T cells, CD4+ T cells, NK cells and NKT cells was measured using flow cytometry, revealing differential expansion among these immune subsets.*

Mechanism of immune Cell Activation by Bria-OTS+. *(A) Bria-PROS+ cells activate T and NK cells partly through the CD86 pathway. PBMCs (1-3 donors) were co-cultured with PC3 or Bria-PROS4+ cells (E:T ratio 10:1), with or without anti-CD86 antibody. After 48 hours, activation was assessed by flow cytometry, showing % change in activation with and without anti-CD86. N=2; *p < 0.05; **p < 0.005. (B) Role of IL-12. PC3 or Bria-PROS+ were co-cultured with PBMCs and either treated with IL-12 or left untreated. The results indicate that IL-12 treatment are critical for enhanced immune activation. (C) (D). Expression of activating and inhibitory ligands in SV-BR-1 cells. SV-BR-1 breast cancer cells (the parental line of Bria-BRES4+) express various ligands for both activating and inhibitory NK cell receptors, as assessed by RNAseq data. (E-H) PBMCs (3 donors) were co-cultured with SV-BR-1 or Bria-BRES4+ cells at a 1:10 effector-to-target (E:T) ratio for 48 hours in the presence or absence of blocking antibodies, and IFNγ release was measured by ELISA. (E) Blocking NKG2D did not significantly reduce IFNγ secretion, suggesting a limited role for the NKG2D pathway in Bria-BRES4+mediated NK cell activation. (F) Blocking HLA-I increased IFNγ secretion, indicating that HLA-I exerts an inhibitory effect via KIRs, while HLA-II blockade had no effect. (G) Blocking NKp46 reduced IFNγ levels, implicating this receptor in Bria-BRES4+-driven NK cell activation. (H) Blocking DNAM-1 partially reduced IFNγ release, suggesting its contribution to the activation process*

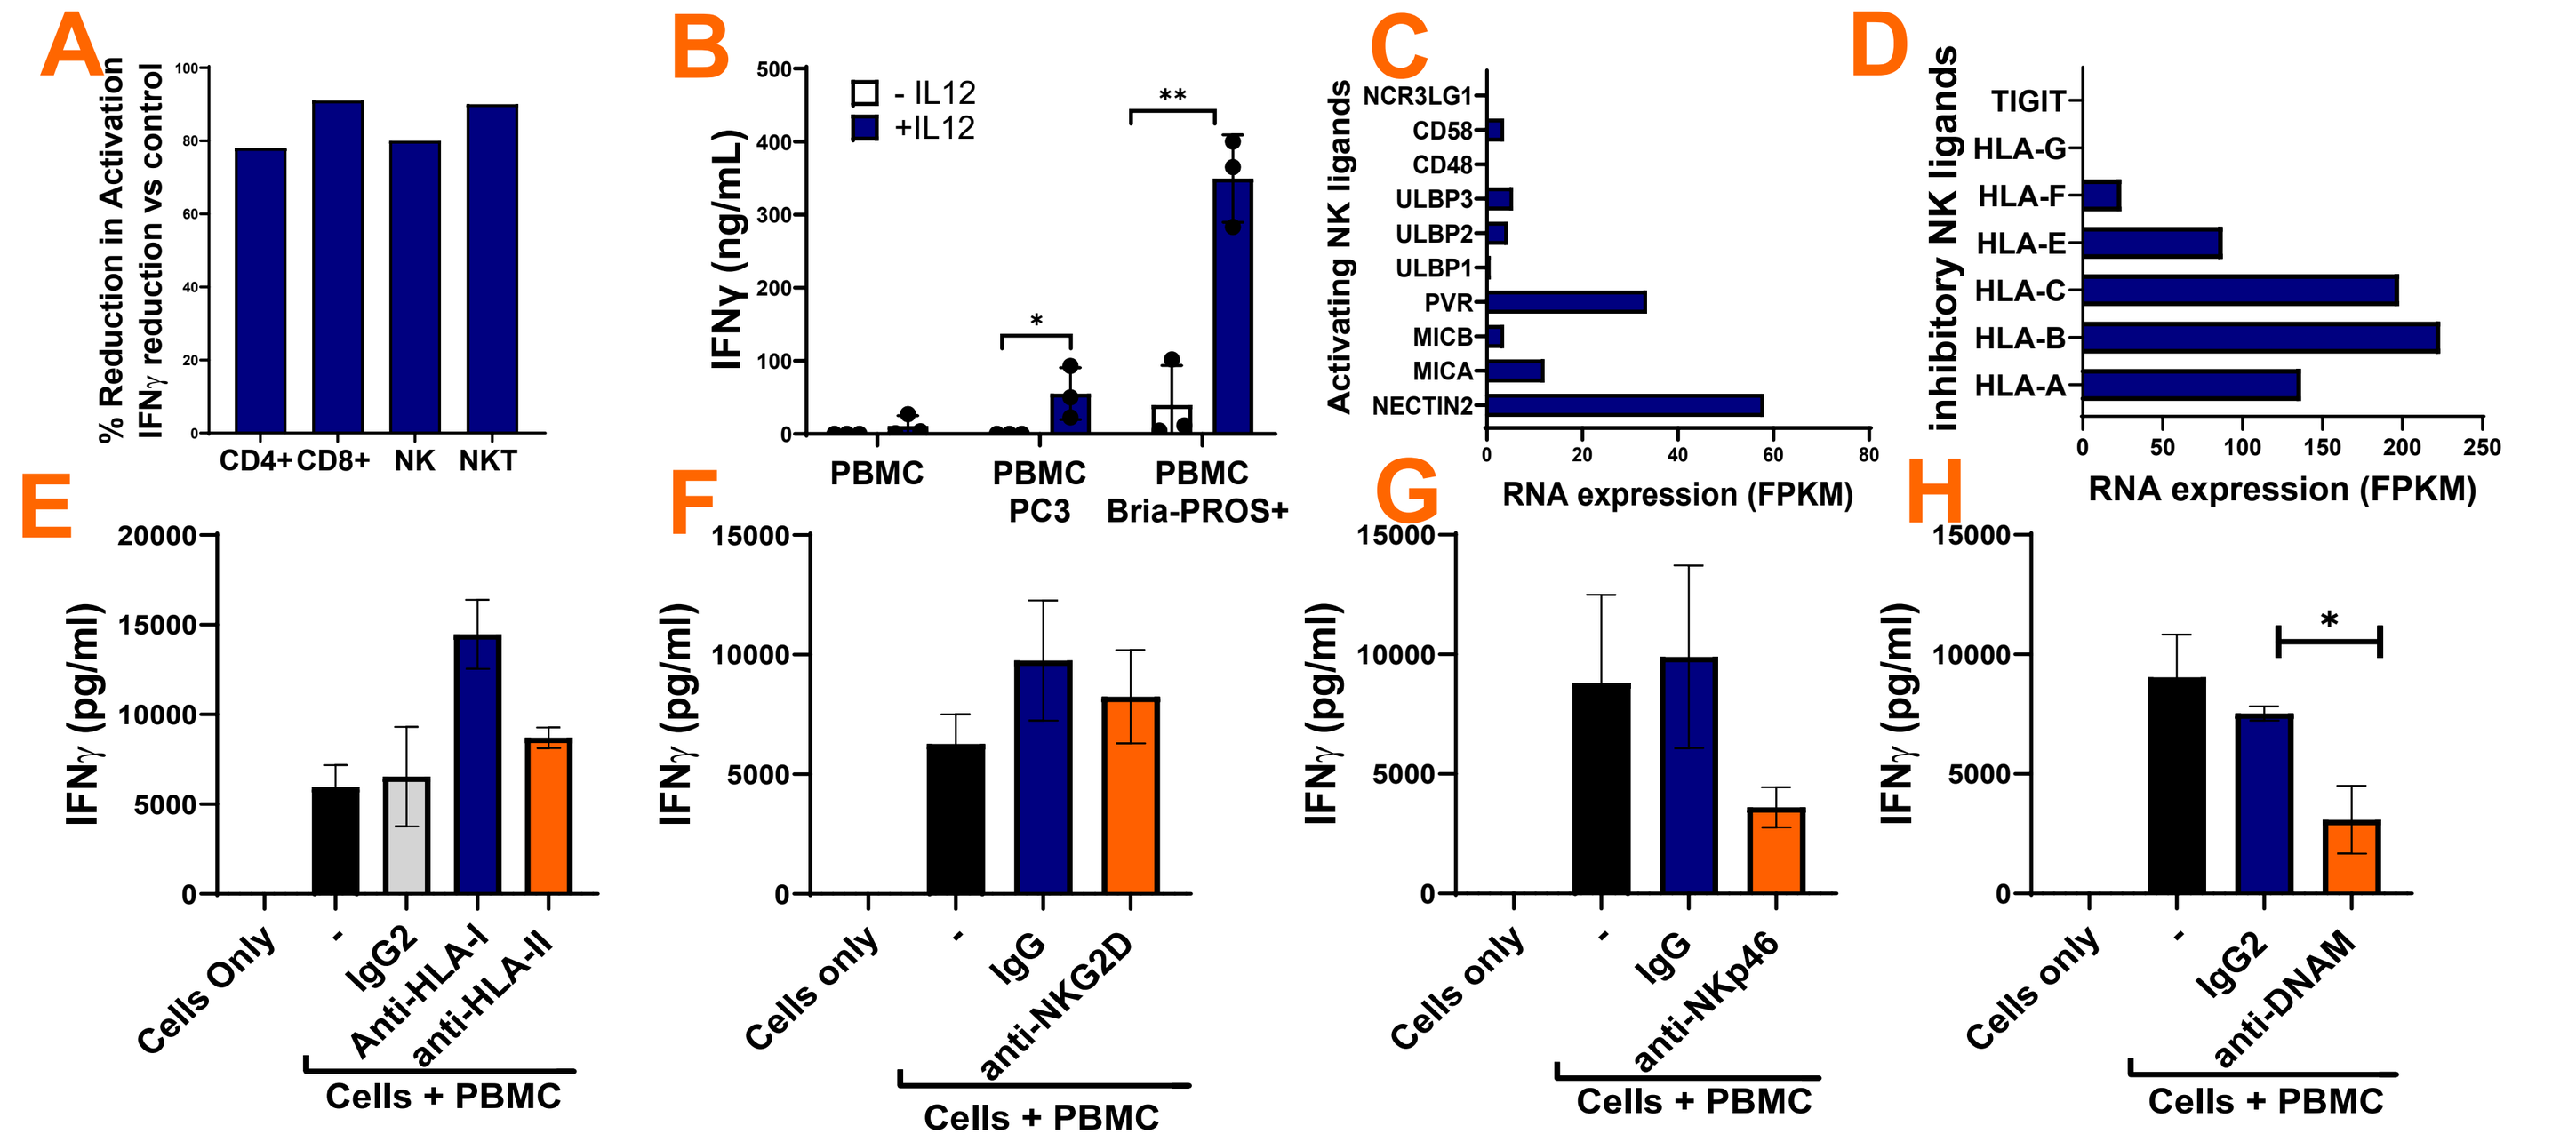
A
% Reduction in Activation IFNγ reduction vs control
CD4+ CD8+ NK NKT
B
IFNγ (ng/mL)
- IL12
+IL12
PBMC
PBMC PC3
PBMC Bria-PROS+
C
Activating NK ligands
NCR3LG1
CD58
CD48
ULBP3
ULBP2
ULBP1
PVR
MICB
MICA
NECTIN2
RNA expression (FPKM)
D
inhibitory NK ligands
TIGIT
HLA-G
HLA-F
HLA-E
HLA-C
HLA-B
HLA-A
RNA expression (FPKM)
E
F
G
H
IFNγ (pg/ml)
Cells Only
IgG2
Anti-HLA-I
anti-HLA-II
IgG
anti-NKG2D
anti-NKp46
anti-DNAM
Cells + PBMC

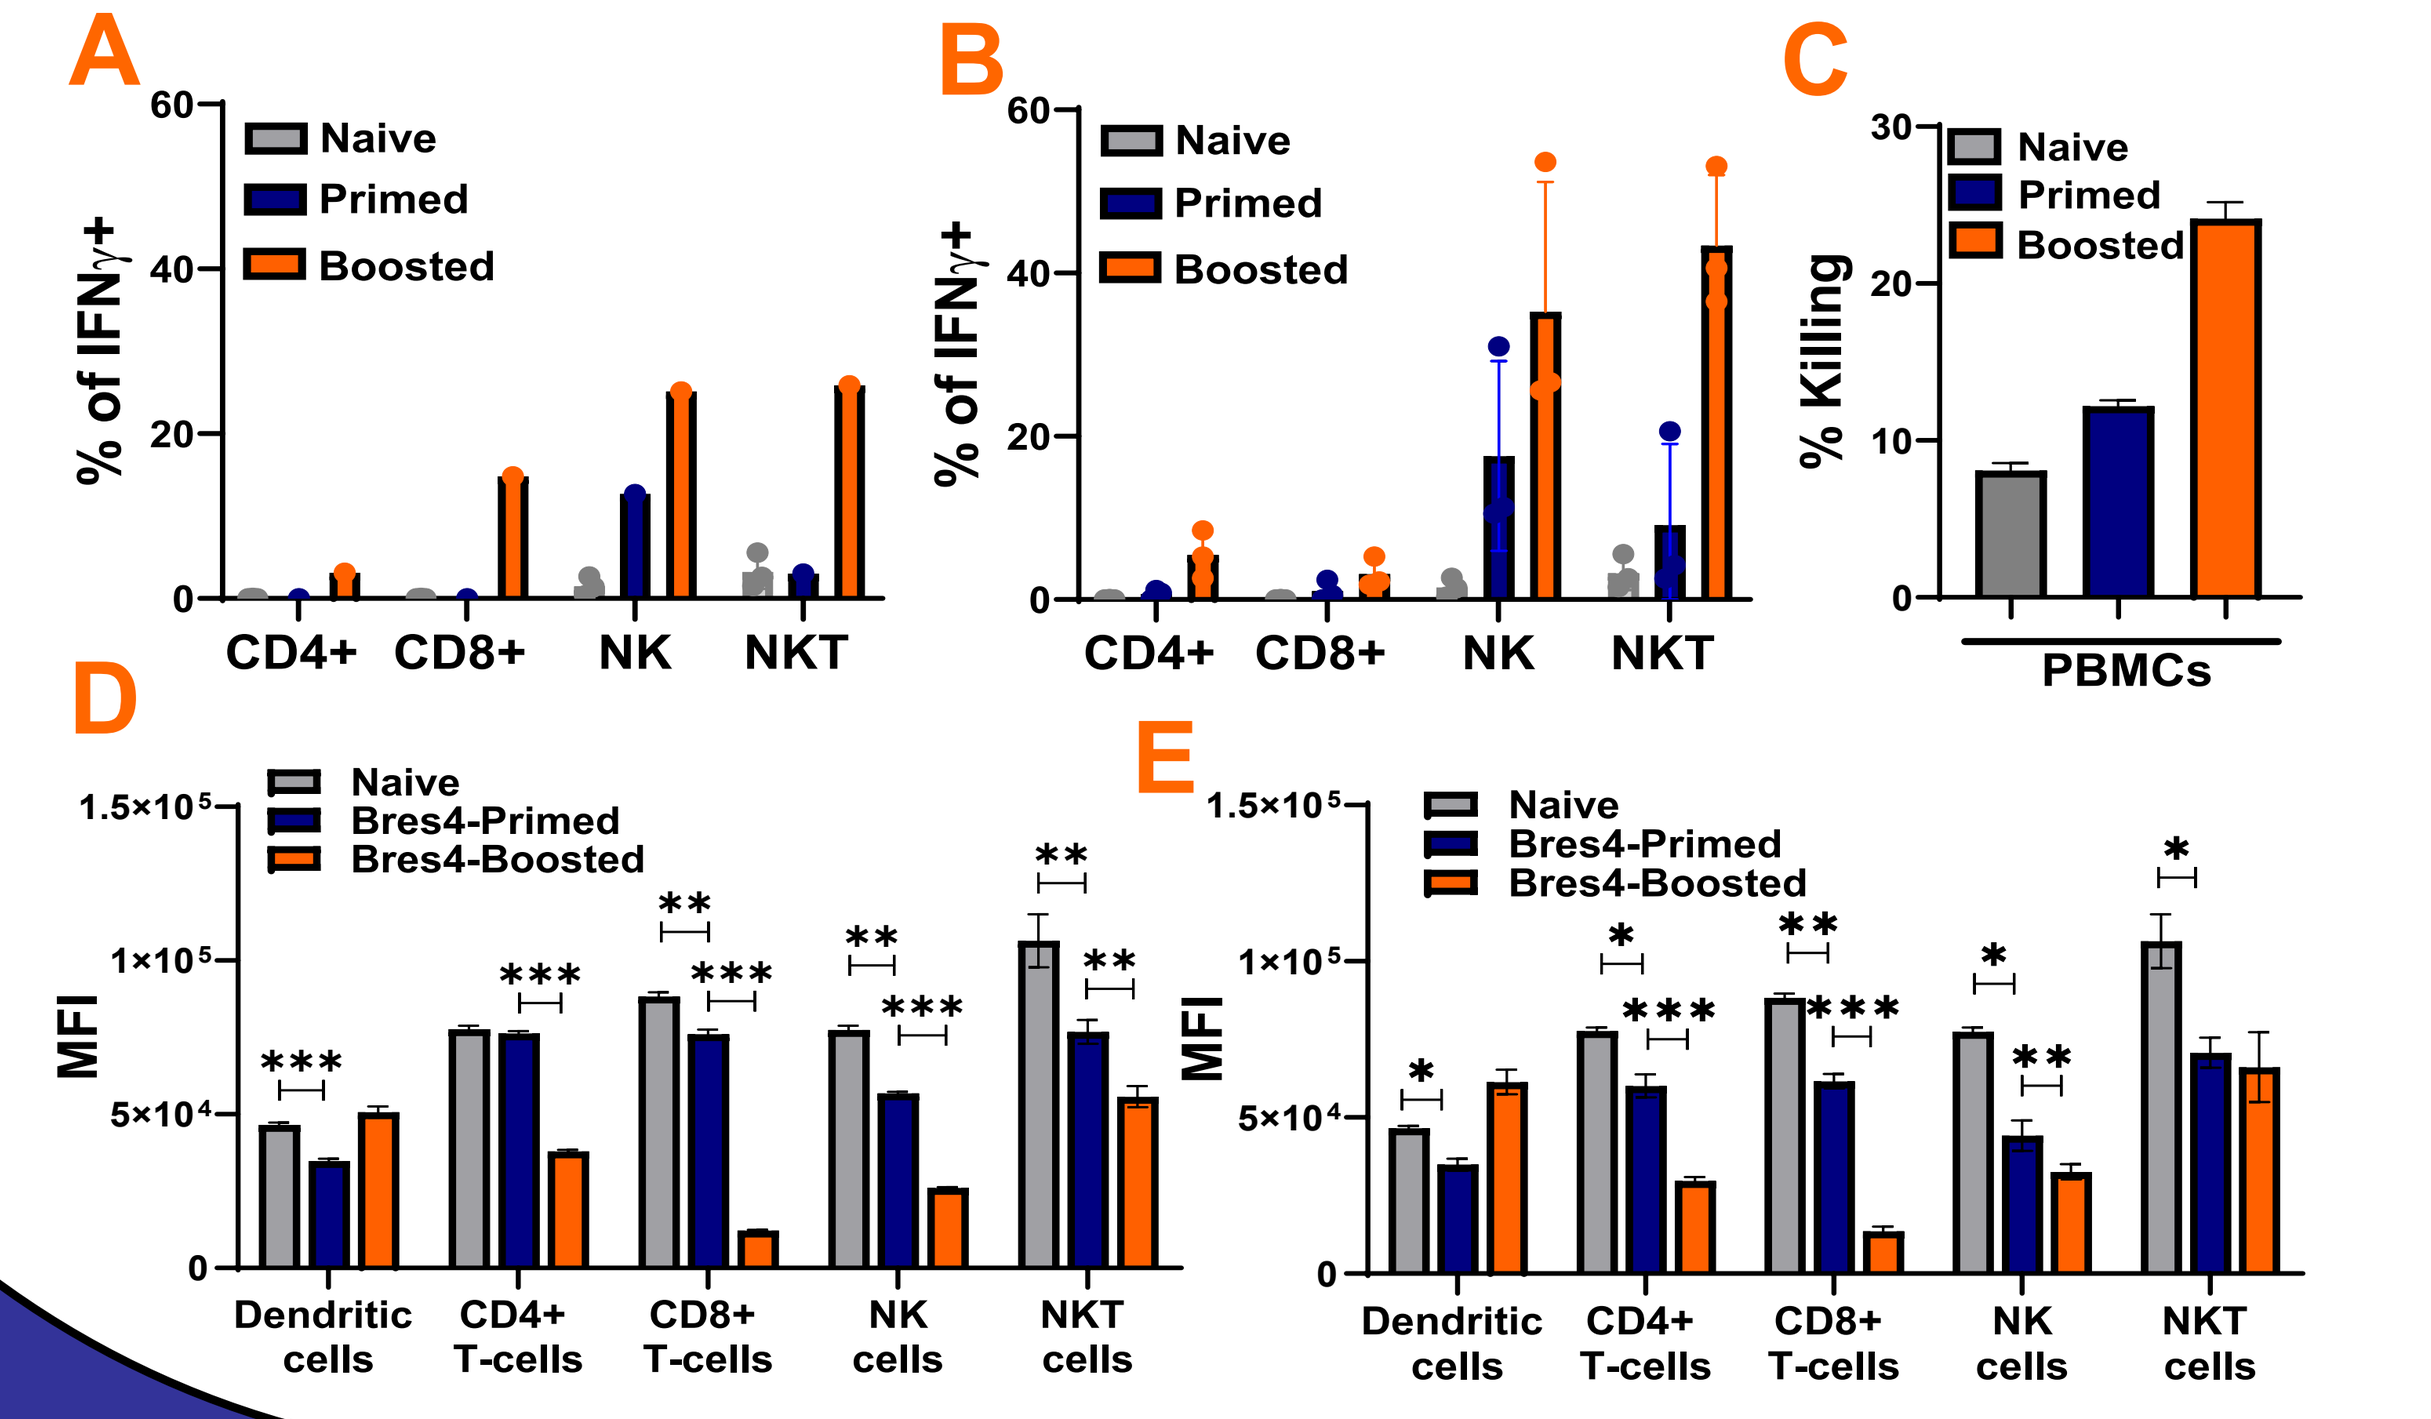
A
B
% of IFNγ+
Naive
Primed
Boosted
CD4+ CD8+ NK NKT
C
% Killing
PBMCs
D
E
MFI
Naive
Bres4-Primed
Bres4-Boosted
Dendritic cells
CD4+ T-cells
CD8+ T-cells
NK cells
NKT cells

Bria-OTS+ activated Immune cells exhibit features of immunological memory. *(A) One million irradiated SV-BR-1 or Bria-BRES4+ or (B) PC3 or Bria-PROS4+ cells were cultured with PBMCs at an effector-to-target (E:T) ratio of 10:1 for 48 hours (priming phase). After this, the PBMCs were allowed to rest without any stimulus for four days (resting phase). The PBMCs were then re-stimulated (boosted) with 250,000 irradiated Bria-Bres4+ cells (A) or Bria-PROS4+ cells (B) at an E:T ratio of 7:1 for 48 hours.. After this, immune cells were analyzed by flow cytometry for intracellular IFN γ with the % positive shown. The results demonstrate a significant increase in IFNγ production upon boosting, indicating the activation and memory formation of PBMCs in response to Bria-PROS4+ cells.(C) Similar to (A), one million irradiated SV-BR-1 or Bria-Bres4+ cells were used for the priming phase with PBMCs (3 donors) at an E:T ratio of 10:1 for 48 hours, followed by a four-day resting period. Subsequently, the PBMCs were boosted with one million irradiated Bria-Bres4+ cells at an E:T ratio of 10:1 for 24 hours, followed by an additional 96 hours of resting. The cytotoxic activity of the activated PBMCs was assessed using an Annexin-V-PI kit after 16 hours of co-culture with SV-BR-1-KO cells at a 4:1 E:T ratio. The percentage of apoptotic cells was significantly higher in the boosted PBMCs, indicating enhanced cytotoxicity and the development of immunological memory. * = p value< 0.05 *** = p value< 0.0005. (D-E) Proliferation of various cell populations is shown by CFSE dye dilution. One million irradiated Bria-BRES4+ (D) or Bria-PROS4+ (E) cells were cultured with PBMCs at an effector-to-target (E:T) ratio of 10:1 for 24 hours (priming phase). The PBMCs were then re-stimulated (boosted) with 500,000 irradiated Bria-Bres4+ cells (D) or Bria-PROS4+ cells (E) cells at an E:T ratio of 10:1 for 24 hours, stained with cell trace Violet and allowed to proliferate for four days. The proliferation of indicated population was measured using flow cytometry.*

PROPOSED MECHANISM OF ACTION

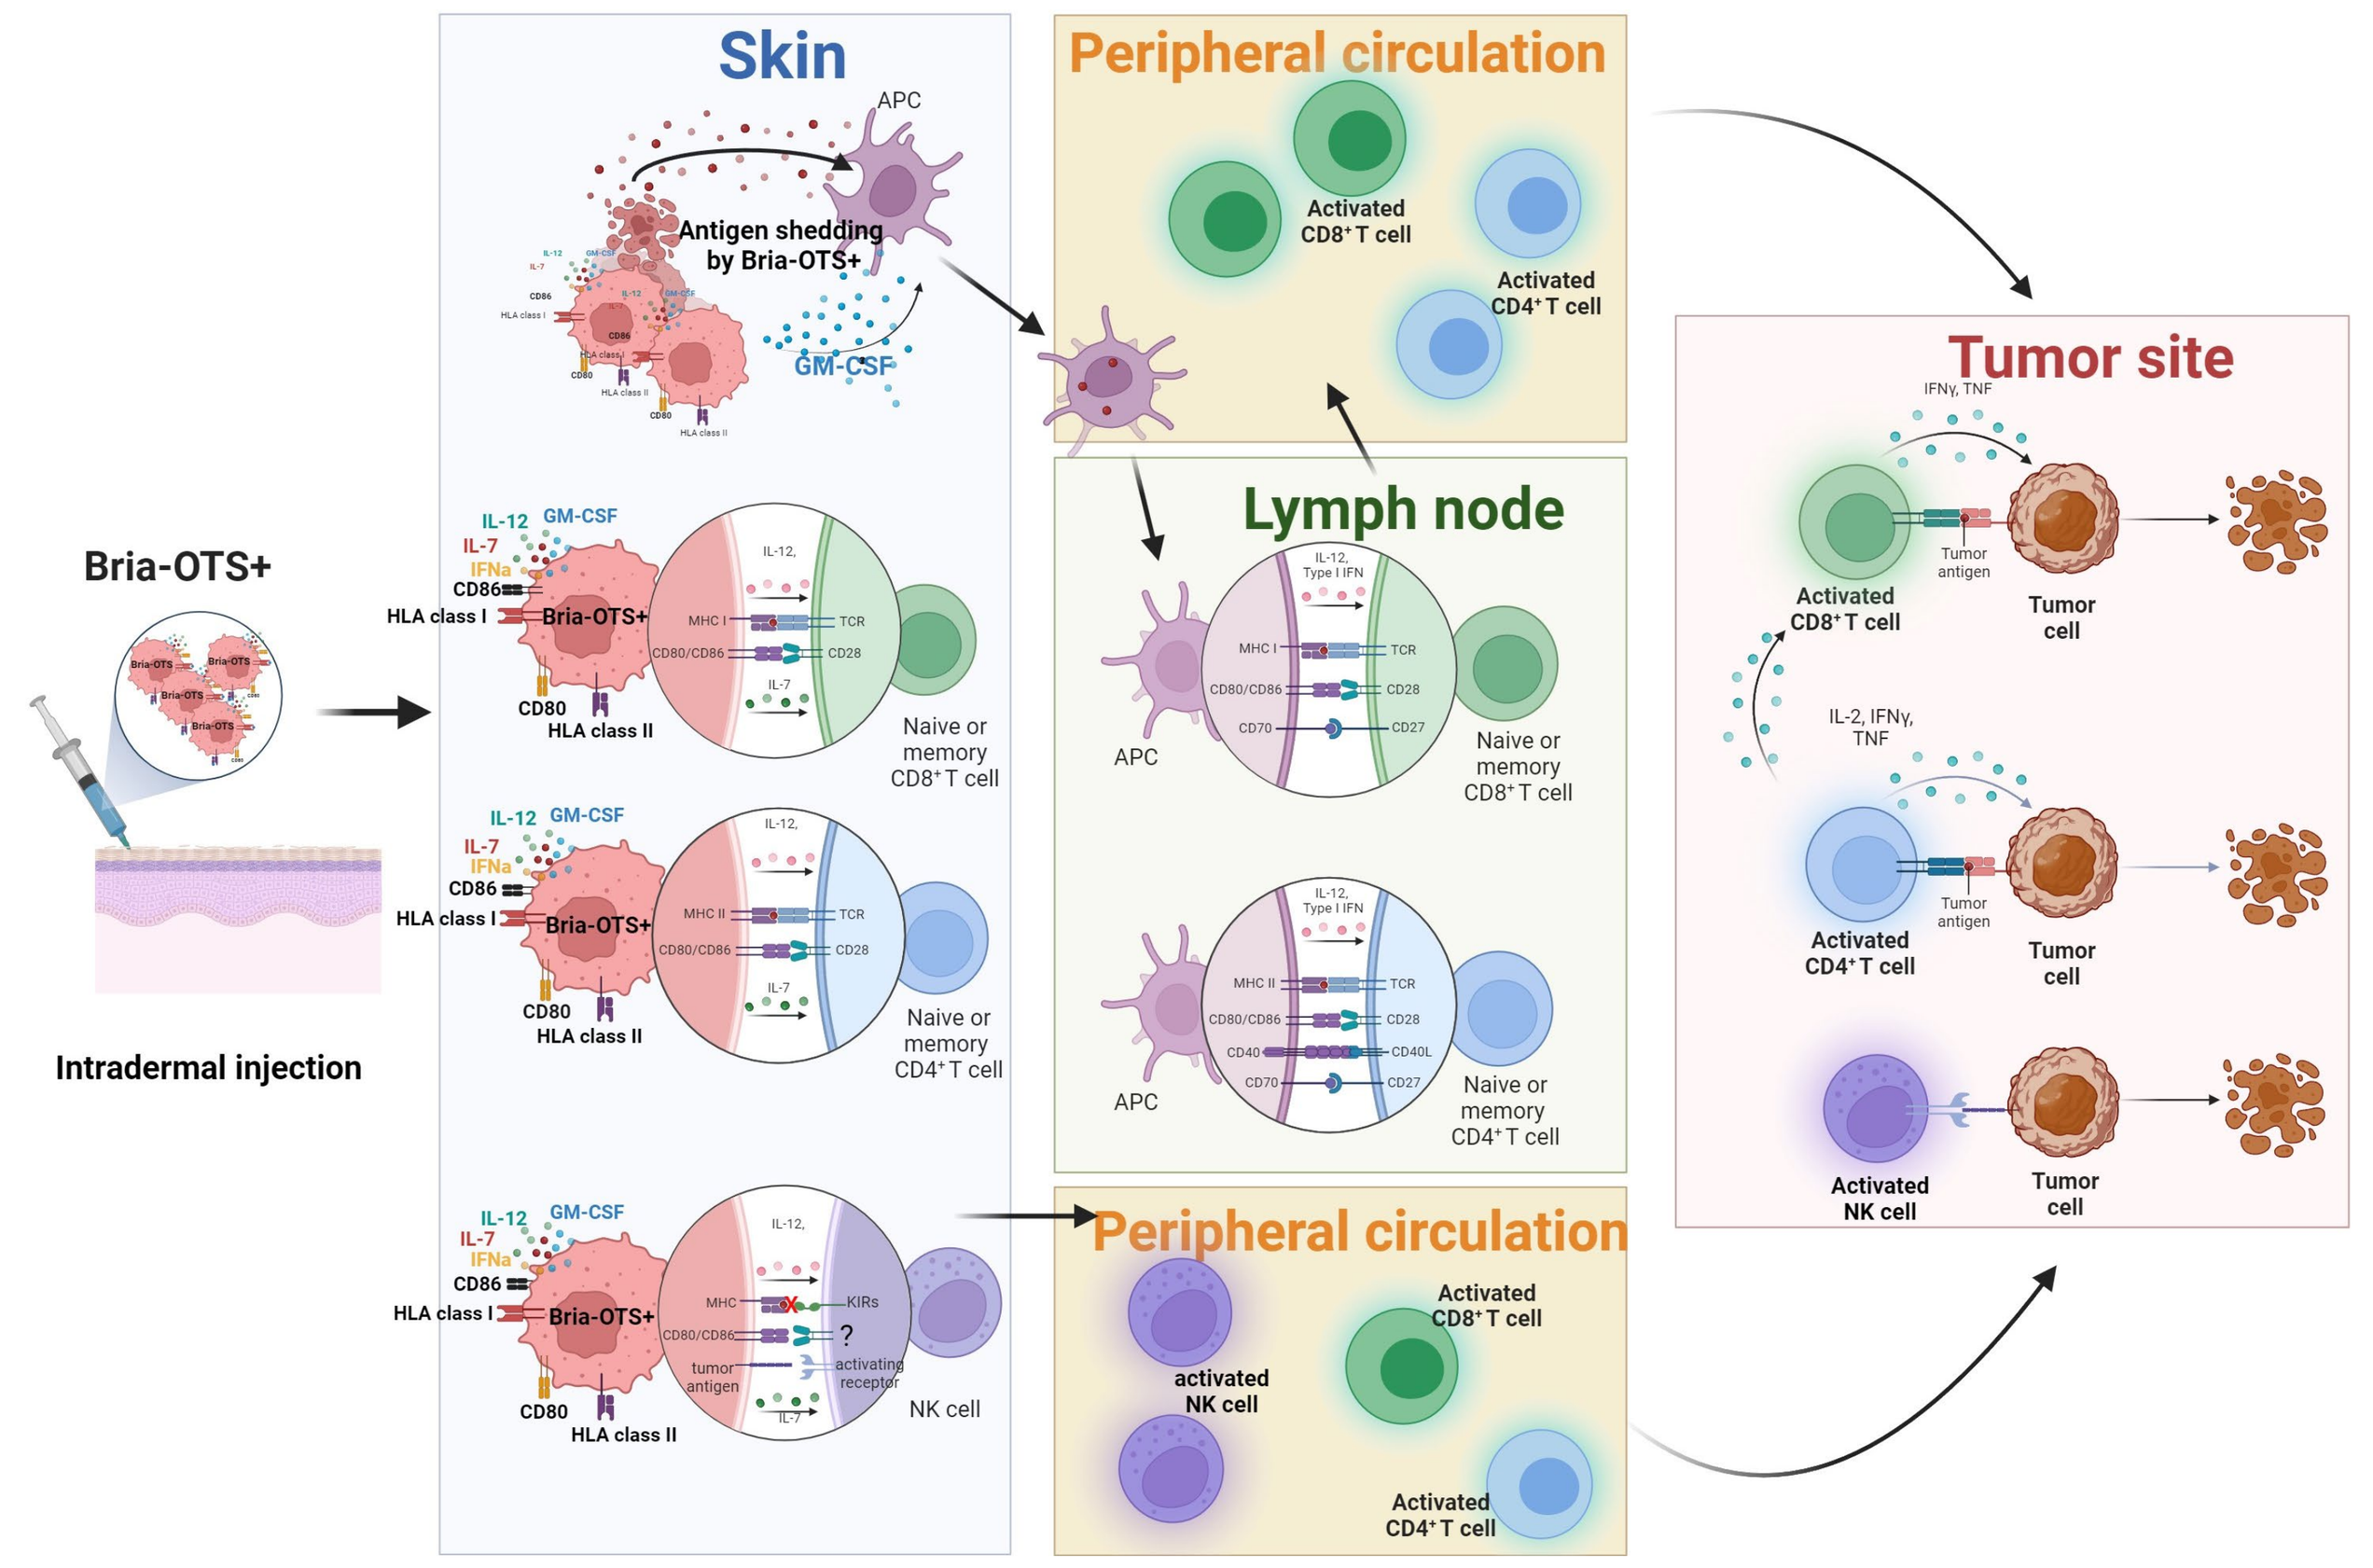
Skin
Peripheral circulation
Lymph node
Tumor site
Peripheral circulation
Bria-OTS+
Intradermal injection
Antigen shedding by Bria-OTS+
APC
GM-CSF
Activated CD8+ T cell
Activated CD4+ T cell
Naive or memory CD8+ T cell
Naive or memory CD4+ T cell
NK cell
activated NK cell
Tumor cell
Tumor antigen
Activated NK cell

Bria-OTS+ Proposed Mechanism of Action: *Bria-OTS, when injected intradermally, directly activates both naive and previously exposed (memory) T-cells, as well as natural killer (NK) cells. Concurrently, professional antigen-presenting cells (APCs) process the Bria-OTS+ antigens. These APCs then migrate to regional lymph nodes, where they prime T-cells against tumor antigens. The activated T-cells and NK cells subsequently travel to the tumor site, where they trigger a robust anti-tumor immune response.*

PATH TO CLINICAL APPLICATION

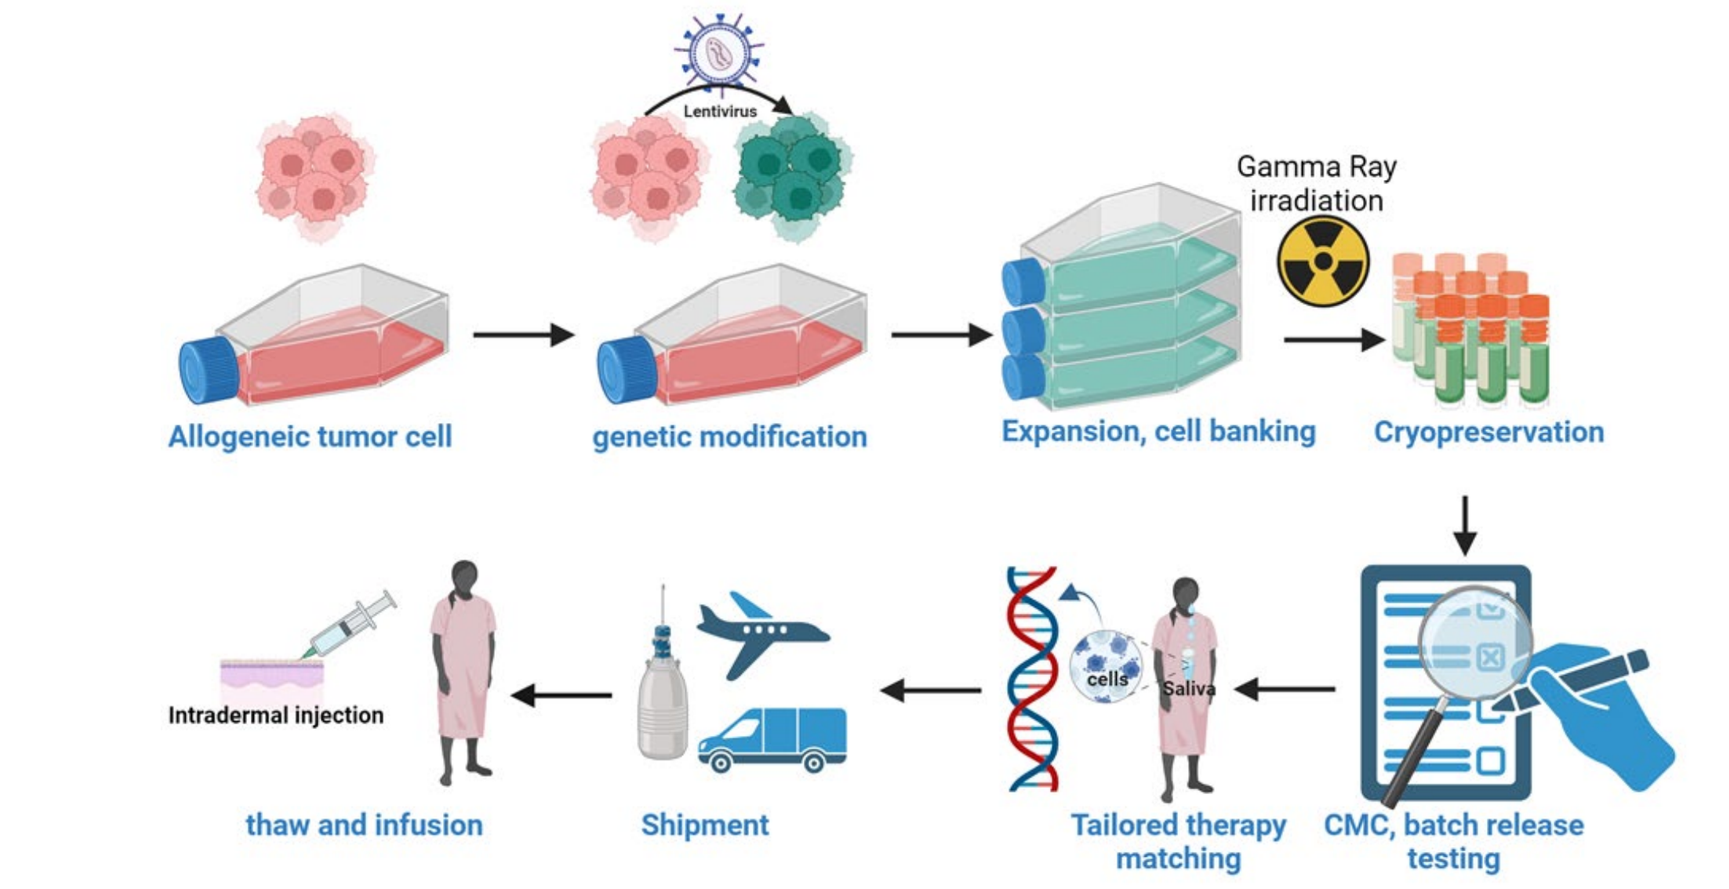
Allogeneic tumor cell
Lentivirus
genetic modification
Gamma Ray irradiation
Expansion, cell banking
Cryopreservation
CMC, batch release testing
Tailored therapy matching
Shipment
Intradermal injection
thaw and infusion

CONCLUSIONS

Bria-OTS+ exhibits the following characteristics:

- **Expresses a variety of cancer-related antigens, including Tumor-Associated Antigens (TAAs) and Post-Translational Modifications (PTMs).**
- **Engages multiple facets of the adaptive immune response.**
- **Activates the innate immune system components.**
- **Counteracts immune escape mechanisms, specifically activating Natural Killer (NK) cells in the case of HLA deletions.**
- **Bria-OTS+ induces a durable, long-lasting anti-tumor immune response by promoting the formation of immunological memory**
- **Designed for both personalized and ready-to-use applications, ensuring robust long-term stability.**
- **Simplified administration process.**
- **Anticipated to have a favorable side effect profile, indicating good tolerance.**