Bria-OTS+: A Cellular Cancer Vaccine Platform Targeting Innate and Adaptive Immunity

Miguel Lopez-Lago[1], Vikas Bhardwaj[1], Pravin Kesarwani[1], Xiaoyi Zheng[1], Sagarika Pachhal[1], Patience Cournoo[1], Renee Cortez[1], George Woodfield[1], Charles L. Wiseman[1], William V. Williams[1]

[1]BriaCell Therapeutics Corp. Philadelphia, PA

SITC annual meeting– Houston
November 6-10 2024

BACKGROUND

Despite advances, achieving curative cancer therapies remains challenging due to the complex and multi-stage nature of the disease. In recent years, therapeutic cancer vaccines have shown renewed signs of efficacy, thanks to advancements in mRNA technology and improved antigen selection. However, several key challenges persist, including the effective identification of relevant tumor antigens, T-cell immune-escape mechanisms that interfere with antigen presentation, and the development of cost-effective manufacturing processes. To address these limitations, we utilize gene-modified allogeneic tumor cells as an immunotherapeutic platform. This platform aims to provide a broad repertoire of tumor antigens, streamlined manufacturing, and the potential for personalization. Our initial version, SV-BR-1-GM (Bria-IMT), a breast cancer cell line expressing GM-CSF, shows promising clinical data. To enhance the efficacy of this platform we modified tumor cells to not only supply a diverse array of tumor antigens, but also directly stimulate the immune system through the expression of cytokines, co-stimulatory factors, and HLA alleles (Bria-OTS+).

OBJECTIVES

We are introducing our second-generation platform, Bria-OTS+, designed to enhance antigen cross-presentation and stimulate direct immune activation. This is achieved through the expression of immunomodulatory cytokines (IL-12, IL-7, GM-CSF, IFNα), co-stimulatory molecules (CD86, CD80, 4-1BBL), and a broadened array of HLA alleles, further optimizing immune engagement.

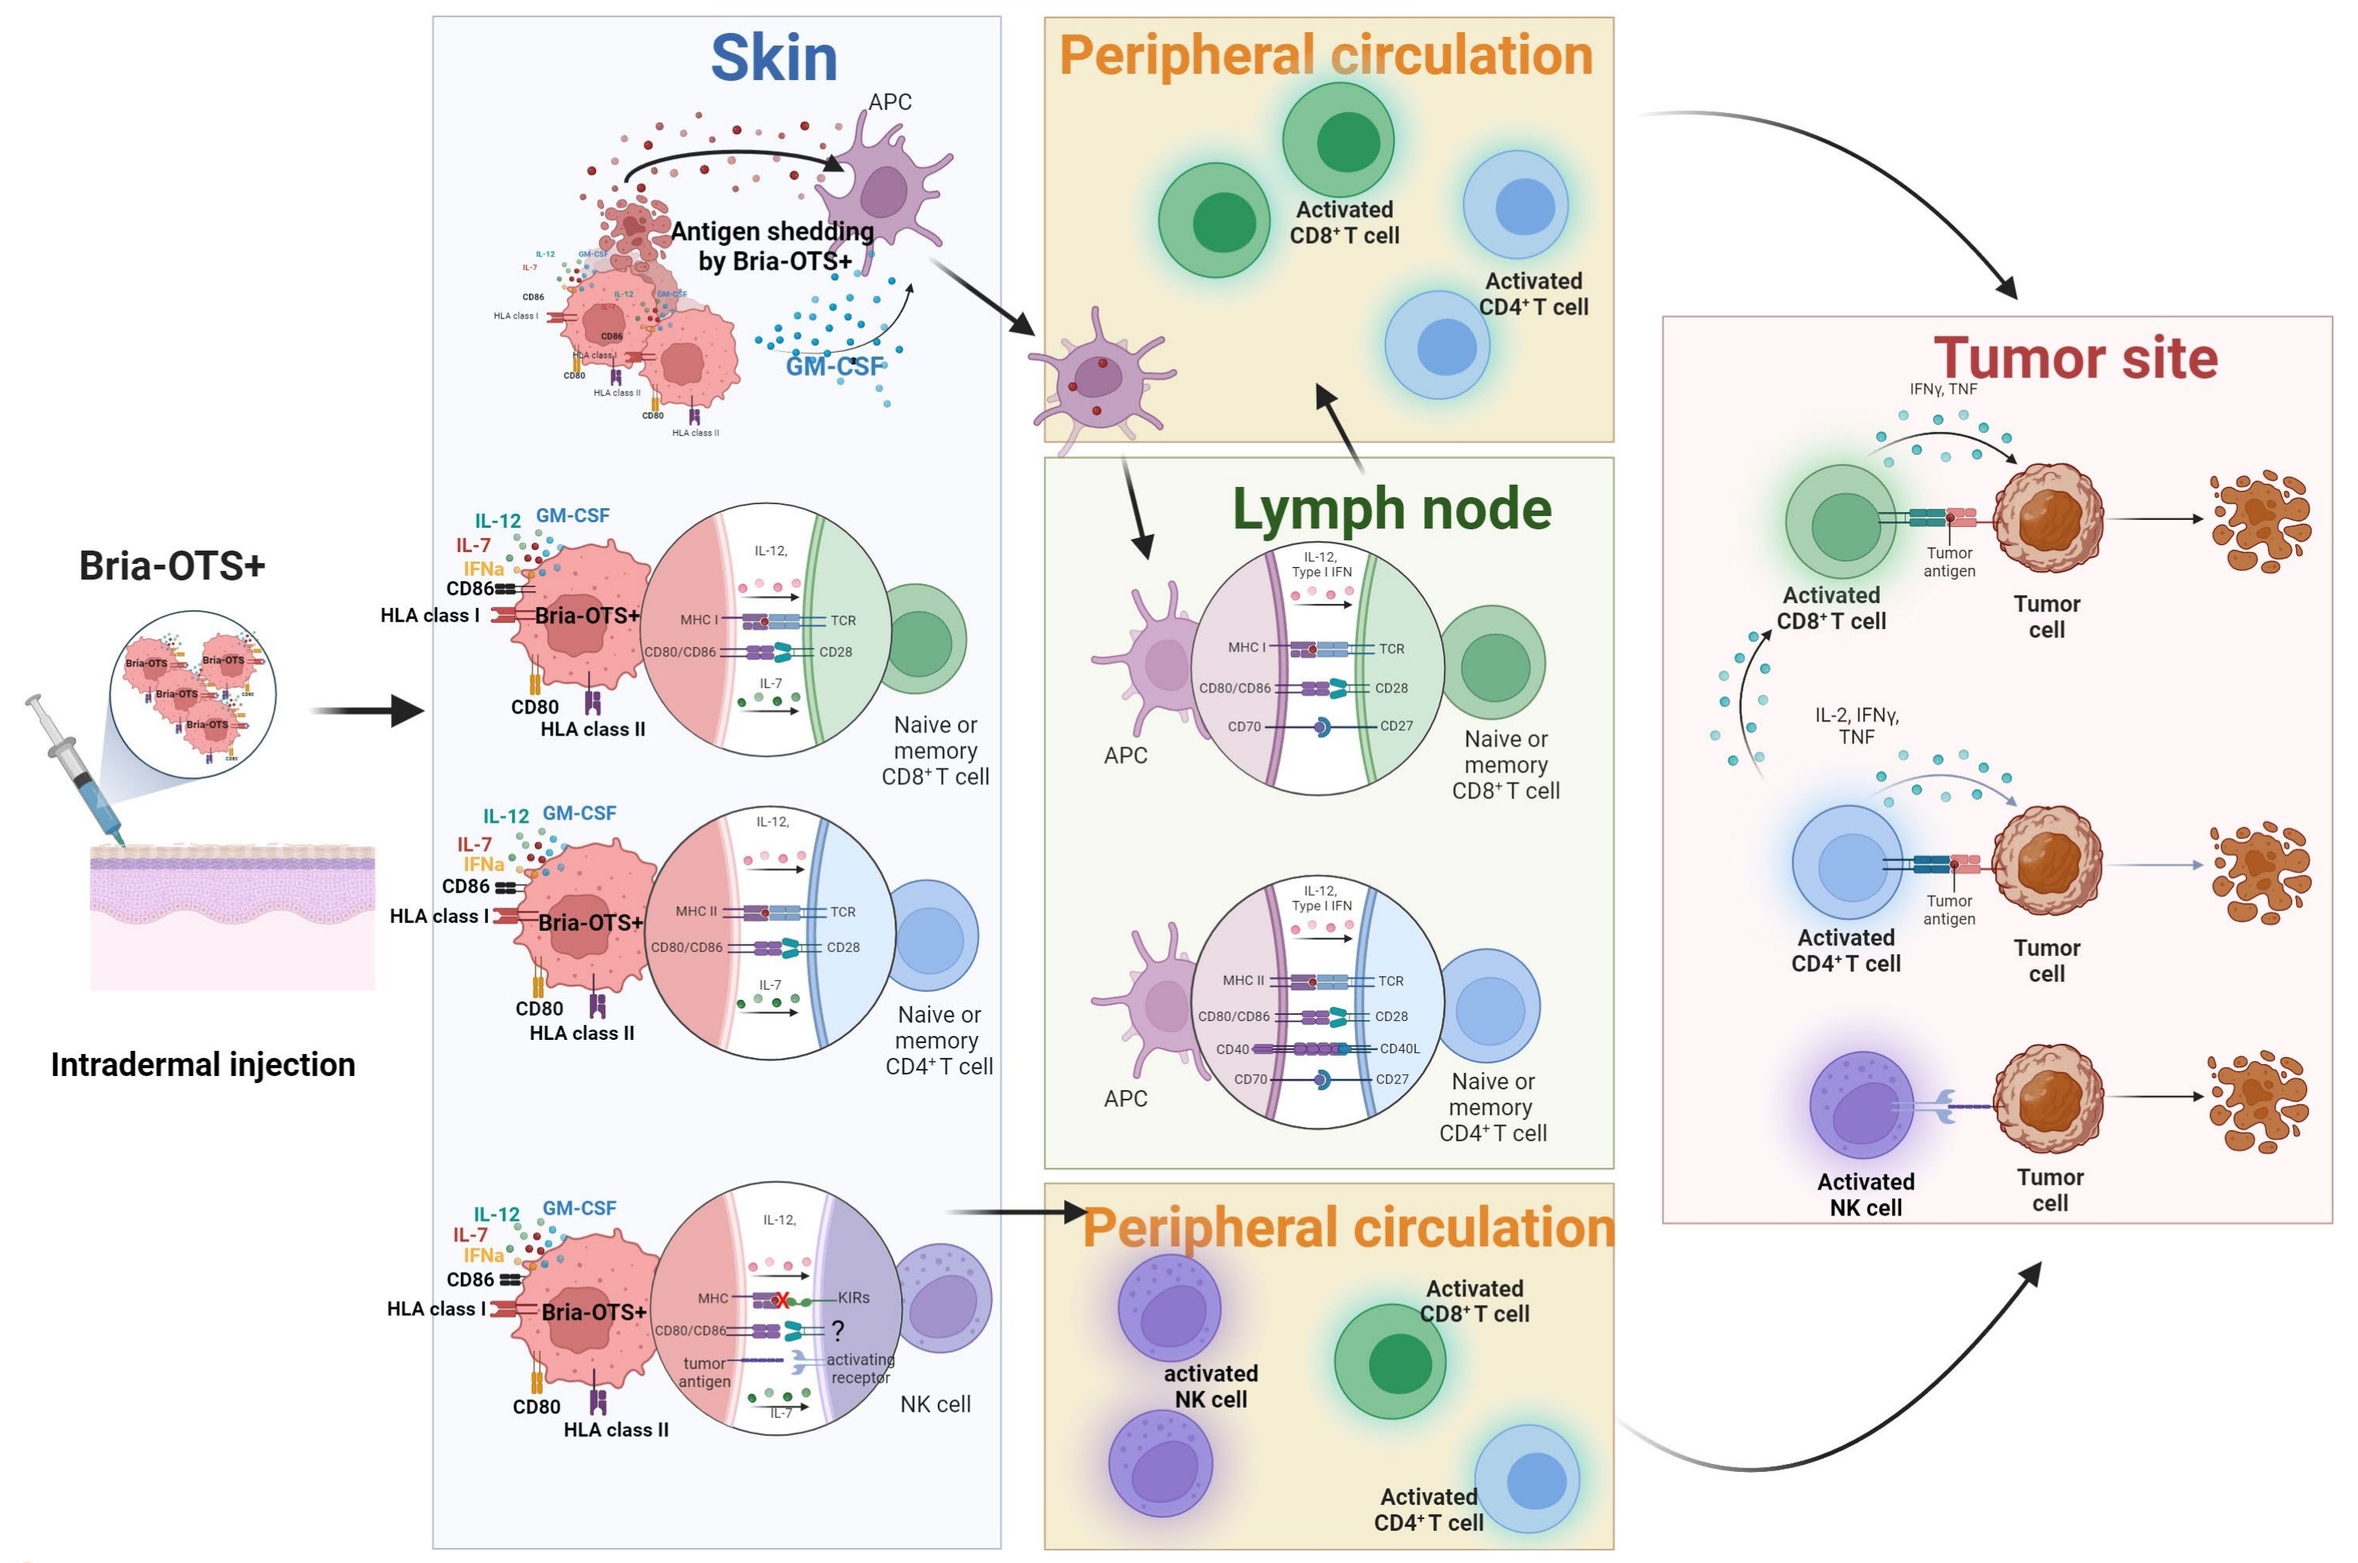
Bria-OTS+
Intradermal injection
Skin
APC
Antigen shedding by Bria-OTS+
GM-CSF
Peripheral circulation
Activated CD8⁺T cell
Activated CD4⁺T cell
Lymph node
Naive or memory CD8⁺T cell
Naive or memory CD4⁺T cell
NK cell
activated NK cell
Tumor site
Tumor cell
Activated NK cell

Bria-OTS+ Mechanism of Action: *Bria-OTS, when injected intradermally, directly activates both naive and previously exposed (memory) T-cells, as well as natural killer (NK) cells. Concurrently, professional antigen-presenting cells (APCs) process the Bria-OTS+ antigens. These APCs then migrate to regional lymph nodes, where they prime T-cells against tumor antigens. The activated T-cells and NK cells subsequently travel to the tumor site, where they trigger a robust anti-tumor immune response.*

Bria-OTS+ cellular cancer vaccine activates the innate and adaptive immune systems

METHODS

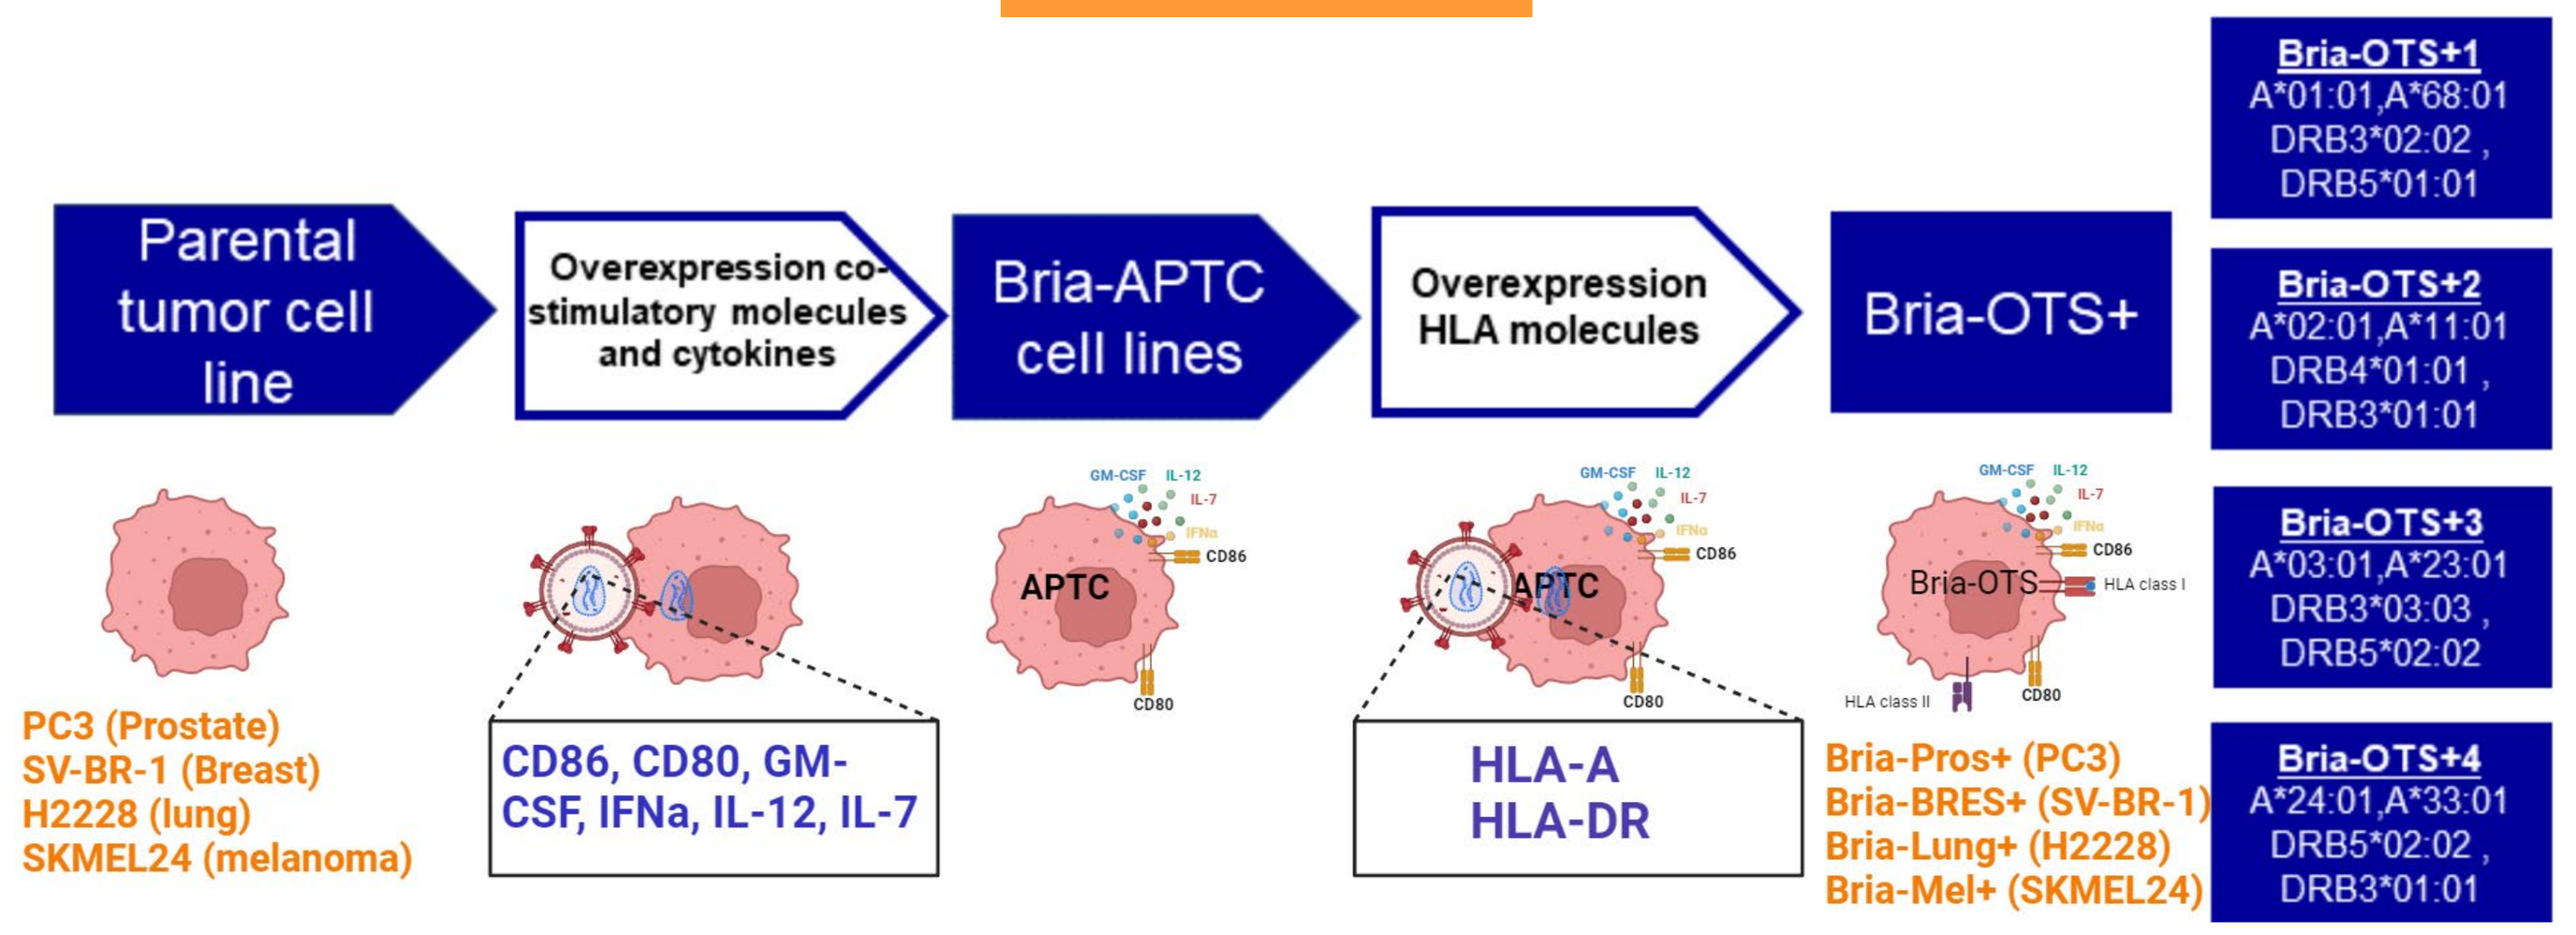
Parental tumor cell line
Overexpression co-stimulatory molecules and cytokines
Bria-APTC cell lines
Overexpression HLA molecules
Bria-OTS+
PC3 (Prostate)
SV-BR-1 (Breast)
H2228 (lung)
SKMEL24 (melanoma)
CD86, CD80, GM-CSF, IFNa, IL-12, IL-7
HLA-A
HLA-DR
Bria-Pros+ (PC3)
Bria-BRES+ (SV-BR-1)
Bria-Lung+ (H2228)
Bria-Mel+ (SKMEL24)
Bria-OTS+1
A*01:01,A*68:01
DRB3*02:02 ,
DRB5*01:01
Bria-OTS+2
A*02:01,A*11:01
DRB4*01:01 ,
DRB3*01:01
Bria-OTS+3
A*03:01,A*23:01
DRB3*03:03 ,
DRB5*02:02
Bria-OTS+4
A*24:01,A*33:01
DRB5*02:02 ,
DRB3*01:01

Engineering of Bria-OTS+ Cell Lines: *The Bria-OTS+ cell lines were developed from various types of cancer, including breast cancer (SV-BR-1), prostate cancer (PC3), melanoma (SK-MEL-24), and lung cancer (NCI-H2228), selected for their unique gene immune signature initially characterized in SV-BR-1 cells. These cell lines were further enhanced to boost their antigen presentation ability and stimulate the immune response by genetically engineering them to express co-stimulatory molecules and immunomodulatory cytokines, resulting in the formation of antigen presenting tumor cells (APTC). Furthermore, to create a semi-allogeneic cell therapy with extensive applicability, the HLA allele expression repertoire of SV-BR-1 was broadened. Population analysis indicated that a configuration of four cell lines, each bearing a combination of two HLA-A and two HLA-DRB3/4/5 alleles, could potentially provide at least one HLA match for 99% of the population. This includes a 92% probability of matching at Class I HLA-A alleles and a 98% likelihood at Class II HLA-DRB3/4/5 alleles.*

RESULTS

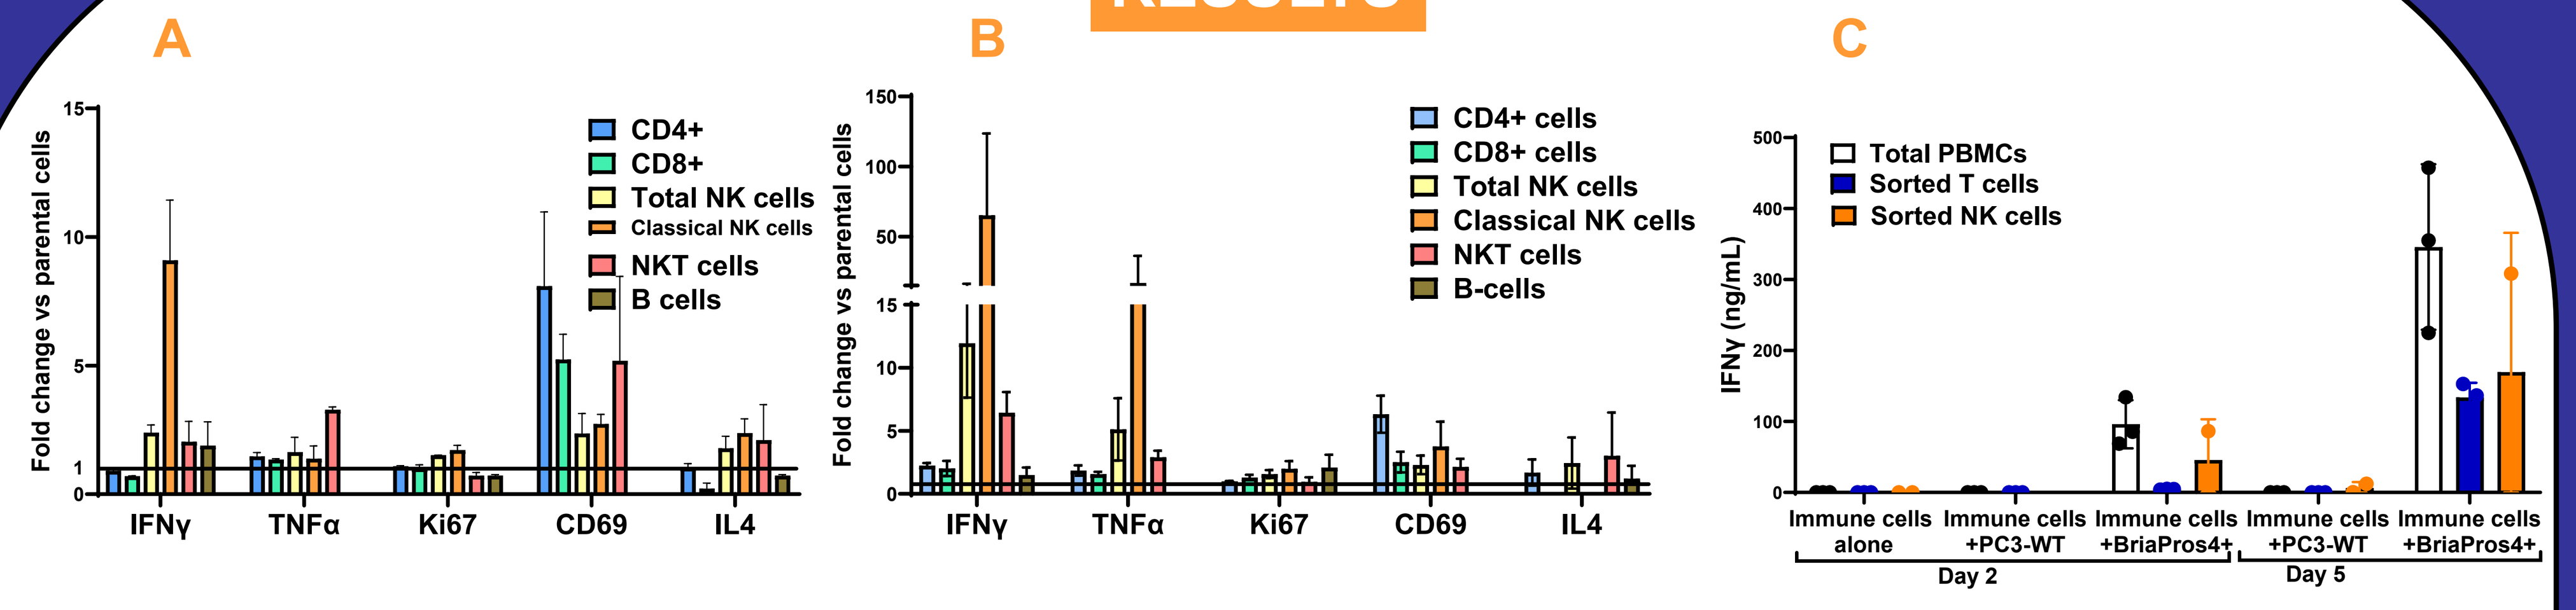
A
Fold change vs parental cells
CD4+
CD8+
Total NK cells
Classical NK cells
NKT cells
B cells
IFNγ
TNFα
Ki67
CD69
IL4
B
CD4+ cells
CD8+ cells
Total NK cells
Classical NK cells
NKT cells
B-cells
C
IFNγ (ng/mL)
Total PBMCs
Sorted T cells
Sorted NK cells
Immune cells alone
Immune cells +PC3-WT
Immune cells +BriaPros4+
Day 2
Day 5

Bria-BRES+ and Bria-PROS+ activates T and NK cells in an in vitro vaccination assay. *SV-BR-1 or PC3, and Bria-BRES+ (A) or Bria-PROS+ (B) (7000 cells) were plated individually and allowed to attach overnight. The next day, 2 different donor PBMCs (E:T =10:1) were added to the cancer cells. After 48 hours of co-culture, immune cells were analyzed by flow cytometry with a variety of activation markers with the % positive shown. (C) PBMCs, isolated NKs or isolated T-cells were incubated alone, with PC3-WT, or with Bria-PROS4+ cells at a E:T ratio of 10:1. IFNγ levels were measured by ELISA in supernatants after 2 or 5 days of incubation. Classical NK cells and T cells were activated.*

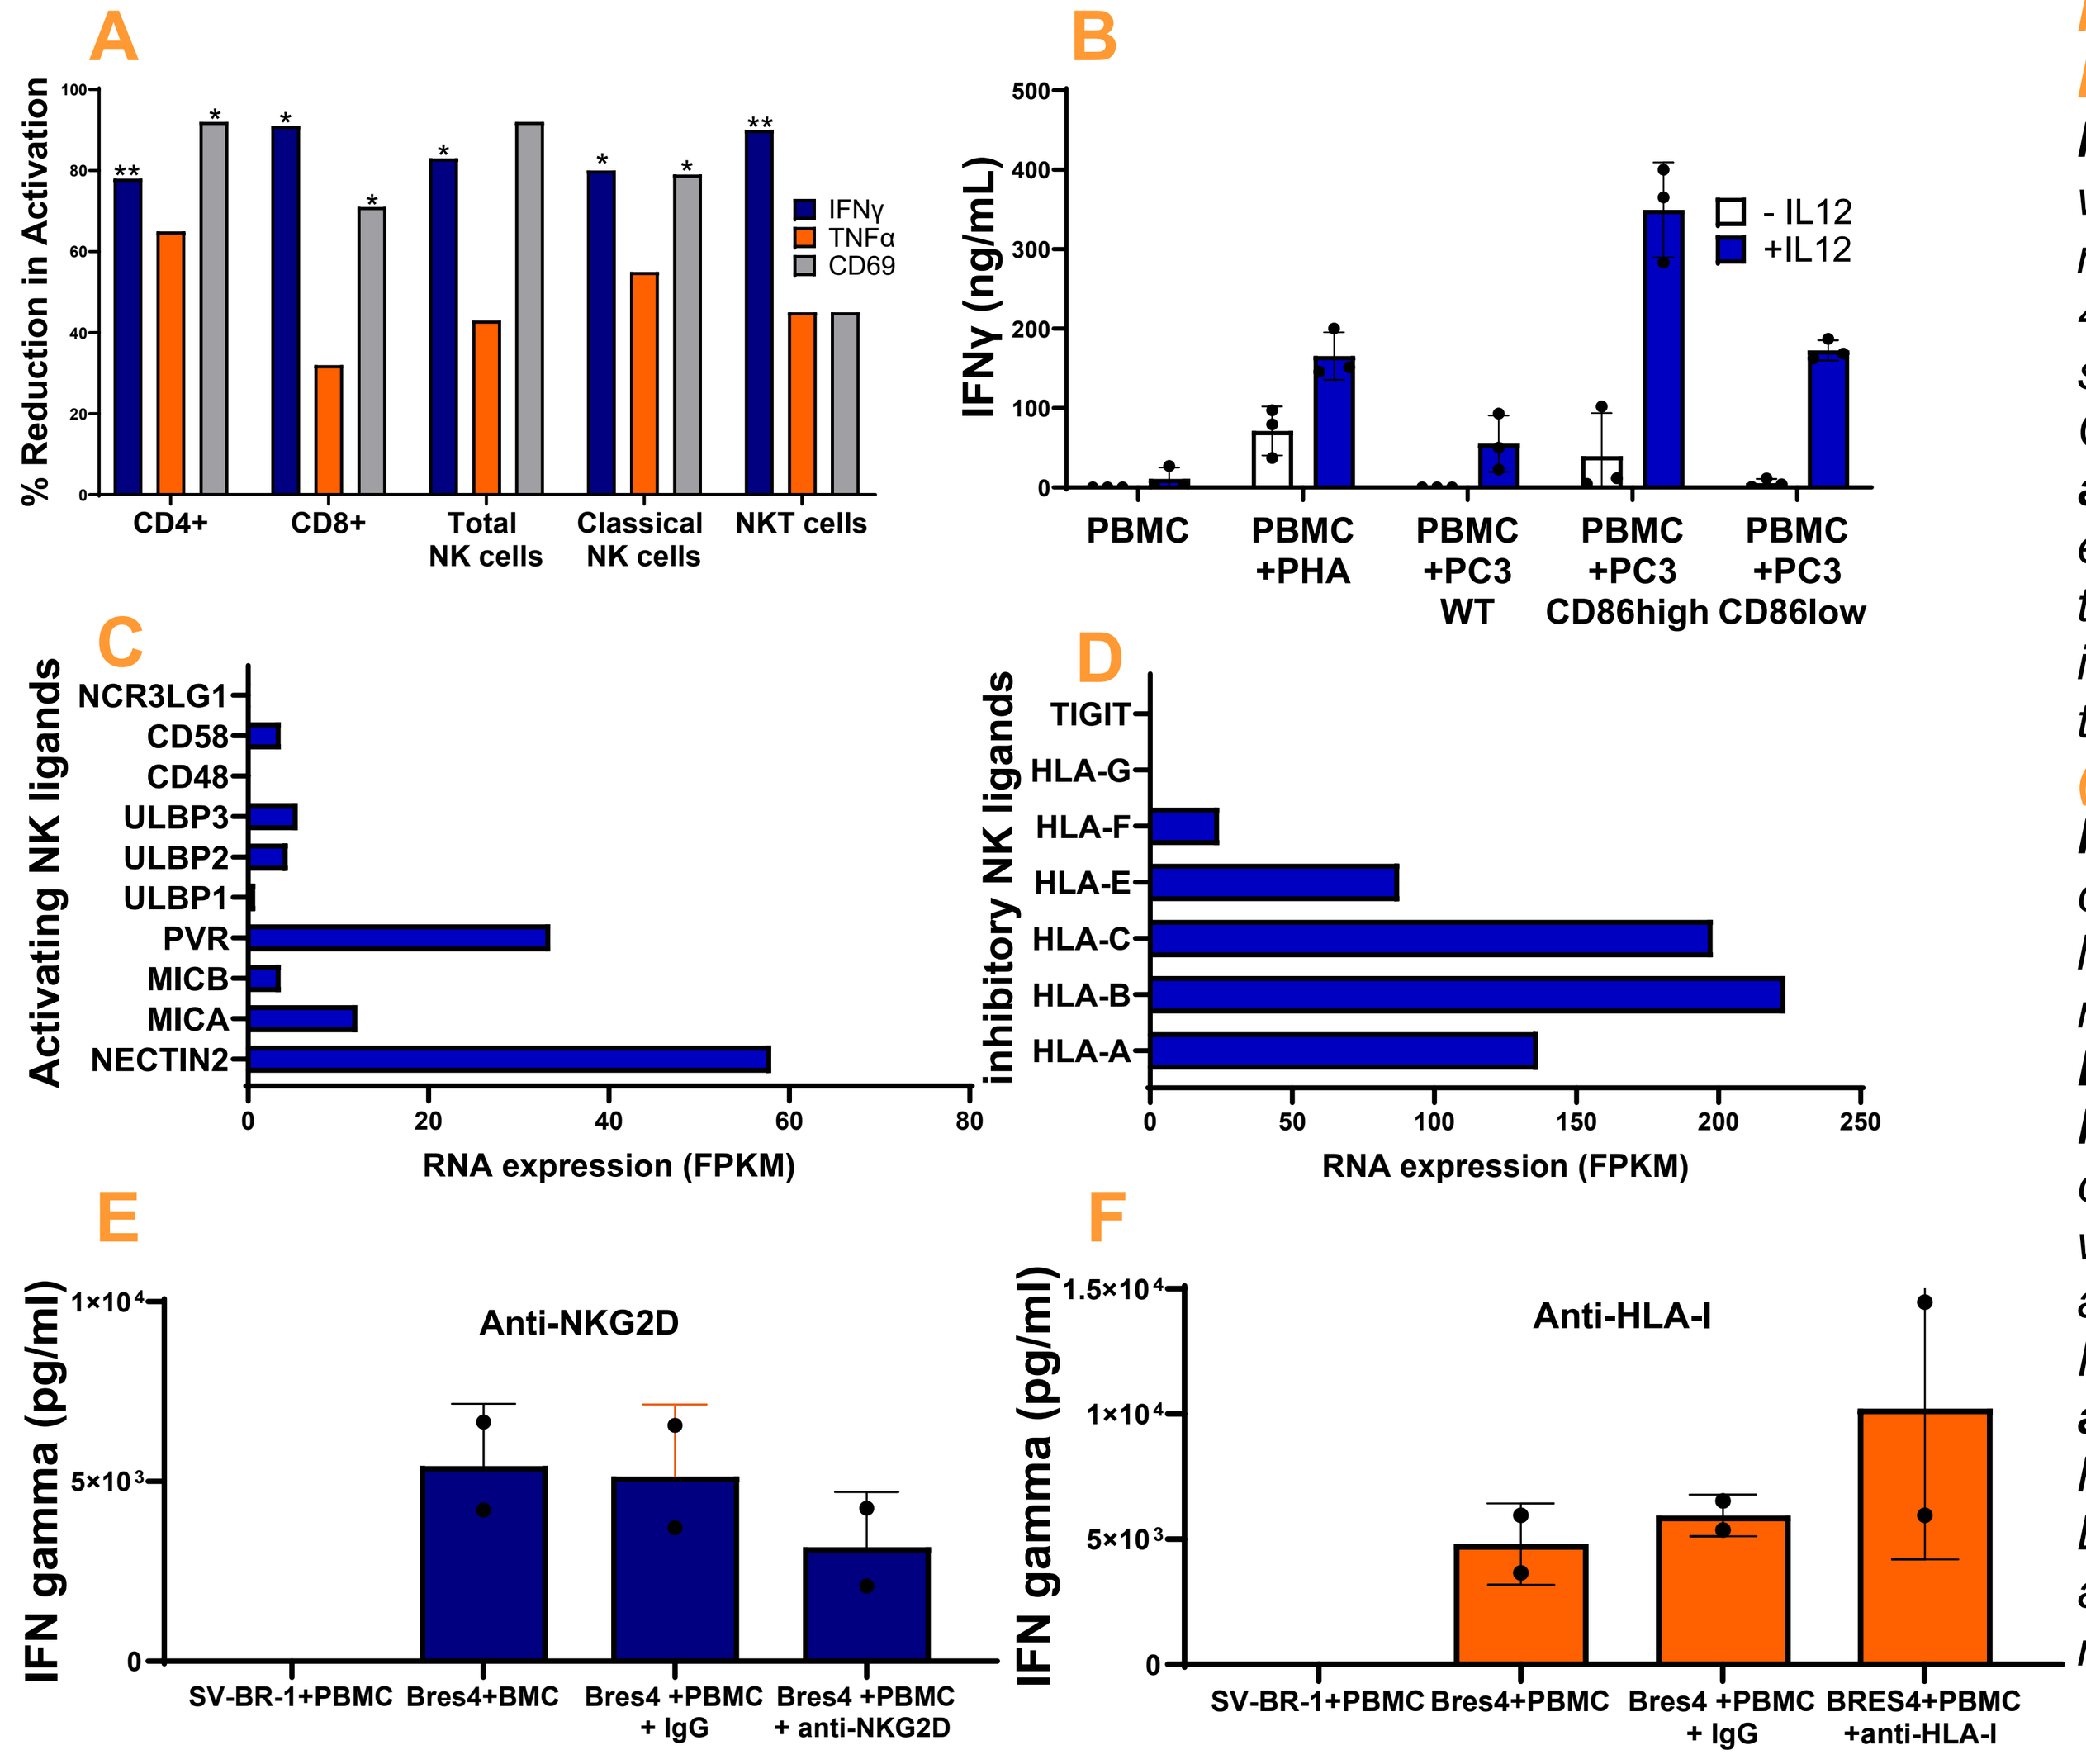
A
% Reduction in Activation
IFNγ
TNFα
CD69
CD4+
CD8+
Total NK cells
Classical NK cells
NKT cells
B
IFNγ (ng/mL)
- IL12
+IL12
PBMC
PBMC +PHA
PBMC +PC3 WT
PBMC +PC3 CD86high
PBMC +PC3 CD86low
C
Activating NK ligands
NCR3LG1
CD58
CD48
ULBP3
ULBP2
ULBP1
PVR
MICB
MICA
NECTIN2
RNA expression (FPKM)
D
inhibitory NK ligands
TIGIT
HLA-G
HLA-F
HLA-E
HLA-C
HLA-B
HLA-A
E
Anti-NKG2D
IFN gamma (pg/ml)
SV-BR-1+PBMC
Bres4+BMC
Bres4 +PBMC + IgG
Bres4 +PBMC + anti-NKG2D
F
Anti-HLA-I
SV-BR-1+PBMC
Bres4+PBMC
Bres4 +PBMC + IgG
BRES4+PBMC +anti-HLA-I

Mechanism of immune Cell Activation by Bria-OTS+. (A) Bria-PROS+ cells activate T and NK cells partly through the CD86 pathway*. PBMCs were co-cultured with PC3 or Bria-PROS+ cells (E:T ratio 10:1), with or without anti-CD86 antibody. After 48 hours, activation was assessed by flow cytometry, showing % change in activation with and without anti-CD86. N=2; *p < 0.05; **p < 0.005.* ***(B) Role of CD86 and IL-12.*** *PC3 cells with low and high CD86 expression were co-cultured with PBMCs and either treated with IL-12 or left untreated. The results indicate that both high CD86 expression and IL-12 treatment are critical for enhanced immune activation.* ***(C) (D). Expression of activating and inhibitory ligands in SV-BR-1 cells.*** *SV-BR-1 breast cancer cells (the parental line of Bria-BRES+) express various ligands for both activating and inhibitory NK cell receptors, as assessed by RNAseq data.* ***(E) Bria-BRES+ cells activate NK cells partly through the MICA/B/ULBPs-NKG2D pathway.*** *PBMCs were co-cultured with SV-BR-1 or Bria-BRES+ (E:T ratio 10:1), with or without anti-NKG2D antibody. After 48 hours, activation was assessed by measuring the release of IFNγ by ELISA.* ***(F) Bria-BRES+ cells inhibit NK cell activation partly through the HLA-I-KIRs pathway.*** *PBMCs were co-cultured with SV-BR-1 or Bria-BRES+ (E:T ratio 10:1), with or without anti-HLA-I antibody. After 48 hours, activation was assessed by measuring the release of IFNg by ELISA.*

Activation of the innate immune system by Bria-OTS+ is mediated by CD86, IL-12, NKG2D and inhibited by HLA class I

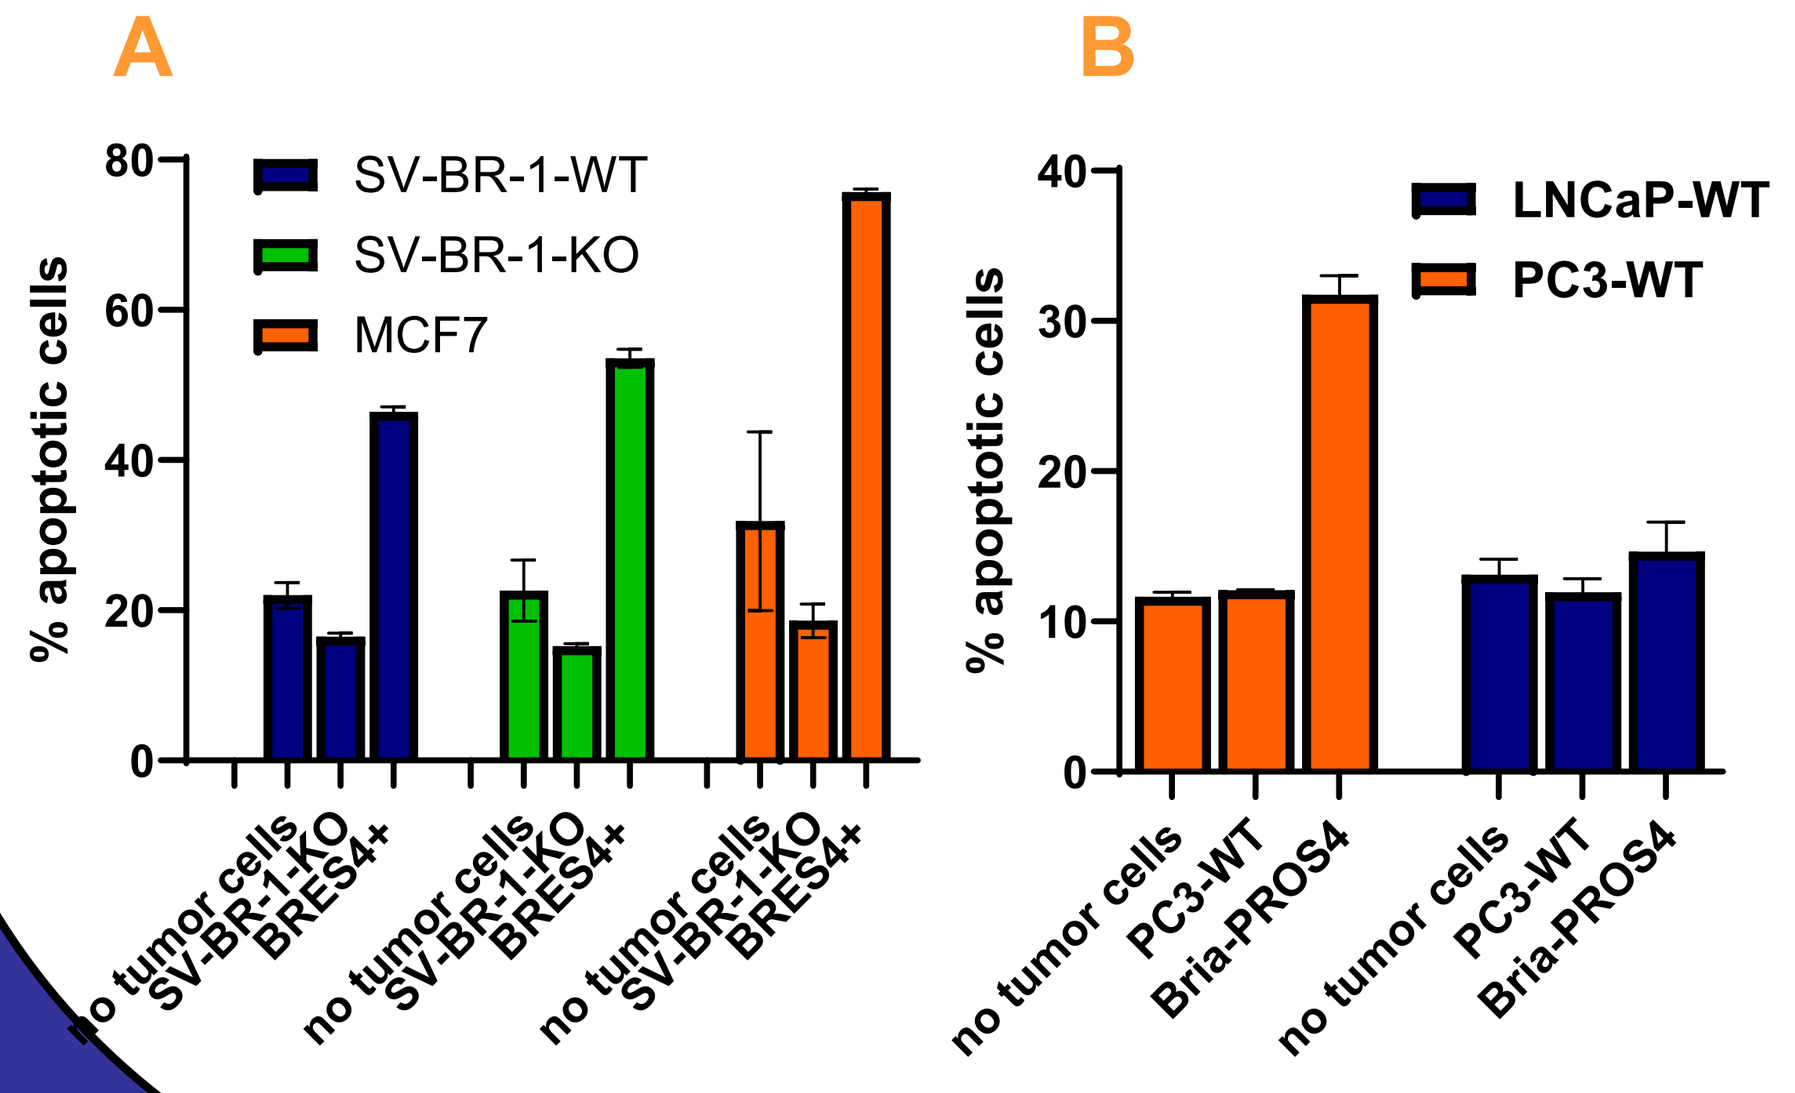
A
% apoptotic cells
SV-BR-1-WT
SV-BR-1-KO
MCF7
no tumor cells
SV-BR-1-KO
BRES4+
B
LNCaP-WT
PC3-WT
no tumor cells
PC3-WT
Bria-PROS4

BRIA-BRES4+ and Bria-PROS+ Primed PBMCs Effectively Target Breast Cancer and prostate cancer Cell Lines in an in vitro vaccination assay: *On Day 0, (A) SV-BR-1-KO and BRIA-Bres4+ or (B) PC3 and Bria-PROS4+ cells (300,000 cells/well) were plated in 6-well plates for the initial priming phase, while PBMCs were thawed and stabilized overnight. The priming phase began on Day 1, with PBMCs co-cultured with indicated cancer cell lines at an E:T ratio of 10:1 for 48 hours. On Day 2, fresh SV-BR-1-wt, SV-BR-1-KO, and MCF-7 or PC3 and LNCaP cells were stained with CellTrace Violet and plated in 12-well plates (100,000 cells/well) as target cells for the effector phase. After the 48-hour sensitization period, PBMCs were gently collected and transferred to the stained target cells for a 72-hour effector phase at an E:T ratio of 10:1. On Day 6, stained target cells were harvested and analyzed for cell death using PI/Annexin V staining.*

RESULTS

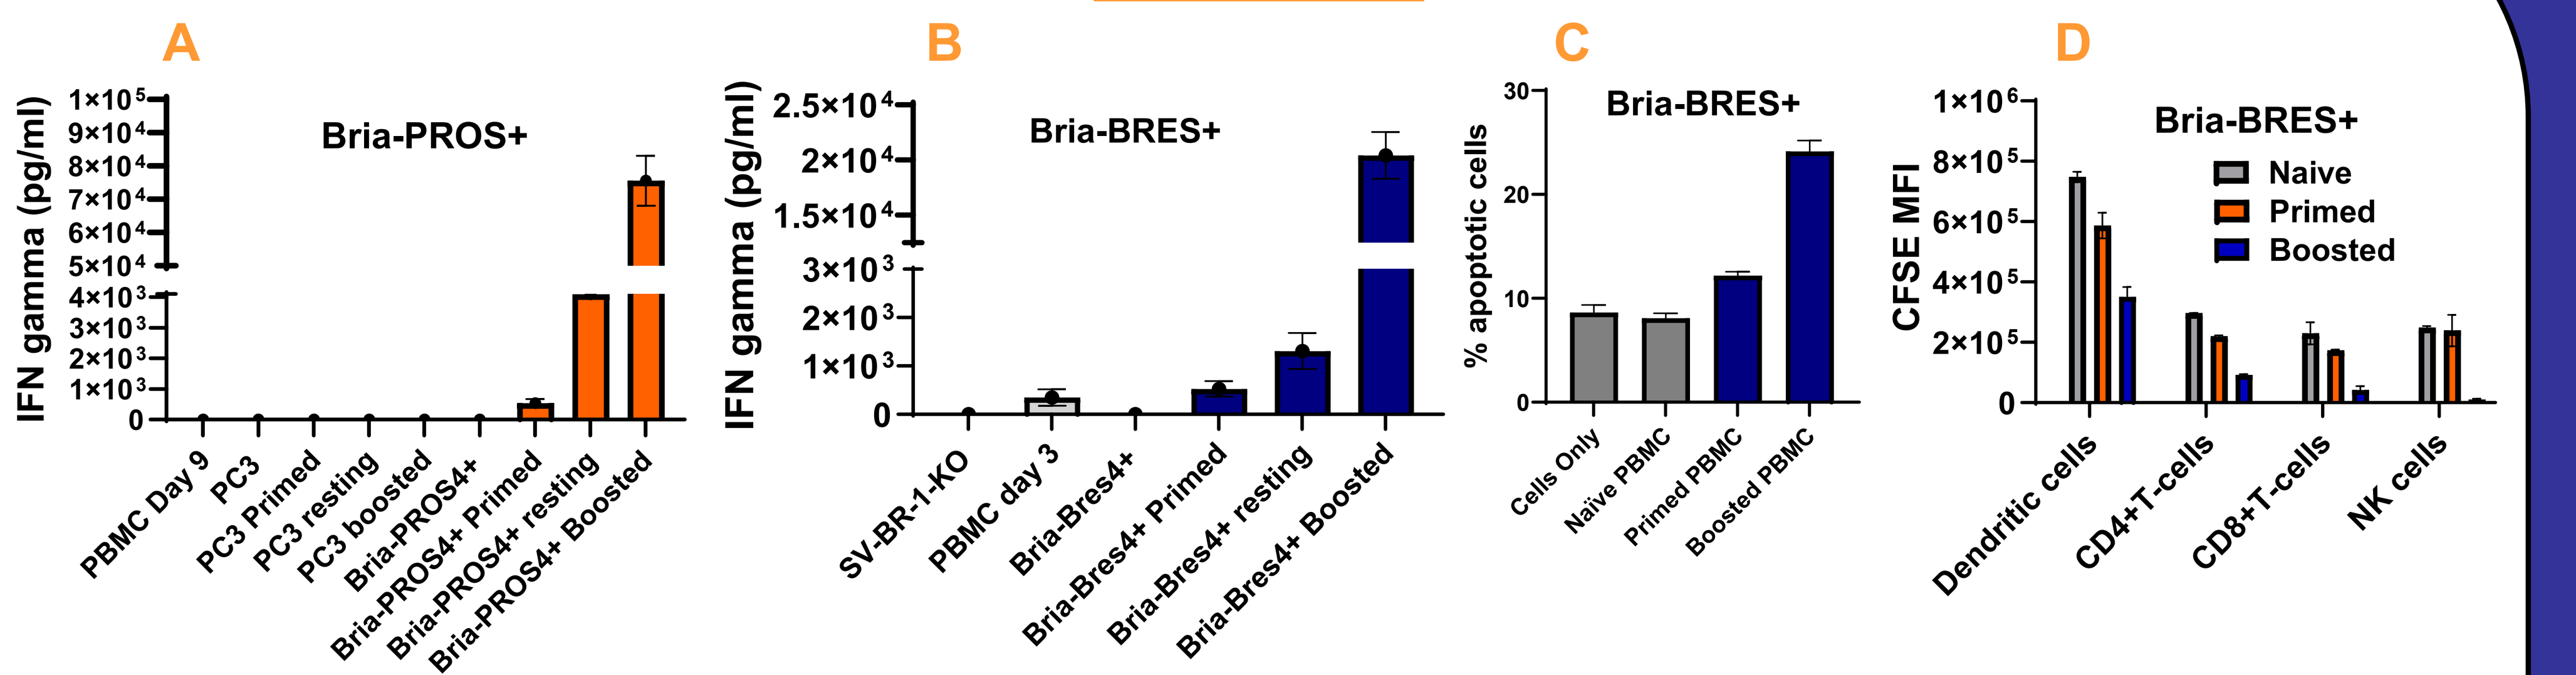
A
Bria-PROS+
IFN gamma (pg/ml)
PBMC Day 9
PC3
PC3 Primed
PC3 resting
PC3 boosted
Bria-PROS4+
Bria-PROS4+ Primed
Bria-PROS4+ resting
Bria-PROS4+ Boosted
B
Bria-BRES+
SV-BR-1-KO
PBMC day 3
Bria-Bres4+
Bria-Bres4+ Primed
Bria-Bres4+ resting
Bria-Bres4+ Boosted
C
Bria-BRES+
% apoptotic cells
Cells Only
Naïve PBMC
Primed PBMC
Boosted PBMC
D
Bria-BRES+
CFSE MFI
Naive
Primed
Boosted
Dendritic cells
CD4+T-cells
CD8+T-cells
NK cells

Bria-OTS+ activated Immune cells exhibit features of immunological memory. (A) *One million irradiated PC3 or Bria-PROS4+ or (B)SV-BR-1-KO or Bria-Bres4+ cells were cultured with PBMCs at an effector-to-target (E:T) ratio of 10:1 for 48 hours (priming phase). After this, the PBMCs were allowed to rest without any stimulus for four days (resting phase). The PBMCs were then re-stimulated (boosted) with 250,000 irradiated Bria-Bres4+ cells at an E:T ratio of 7:1 for 48 hours. Culture media was collected at the indicated time points and analyzed for IFN-γ release using the R&D Systems Quantikine kit. The results demonstrate a significant increase in IFN-γ production upon boosting, indicating the activation and memory formation of PBMCs in response to Bria-Bres4+ cells.(C) Similar to (A), one million irradiated SV-BR-1-KO or Bria-Bres4+ cells were used for the priming phase with PBMCs at an E:T ratio of 10:1 for 48 hours, followed by a four-day resting period. Subsequently, the PBMCs were boosted with one million irradiated Bria-Bres4+ cells at an E:T ratio of 10:1 for 24 hours, followed by an additional 96 hours of resting. The cytotoxic activity of the activated PBMCs was assessed using an Annexin-V-PI kit after 16 hours of co-culture with SV-BR-1-KO cells at a 4:1 E:T ratio. The percentage of apoptotic cells was significantly higher in the boosted PBMCs, indicating enhanced cytotoxicity and the development of immunological memory. * = p value< 0.05 *** = p value< 0.0005. (D) Proliferation of various cell populations is shown by CFSE dye dilution. One million irradiated Bria-Bres4+ cells were cultured with PBMCs at an effector-to-target (E:T) ratio of 10:1 for 24 hours (priming phase). The PBMCs were then re-stimulated (boosted) with 500,000 irradiated Bria-Bres4+ cells at an E:T ratio of 10:1 for 24 hours, stained with cell trace Violet and allowed to proliferate for four days. The proliferation of indicated population was measured using flow cytometry.*

PATH TO THE CLINIC

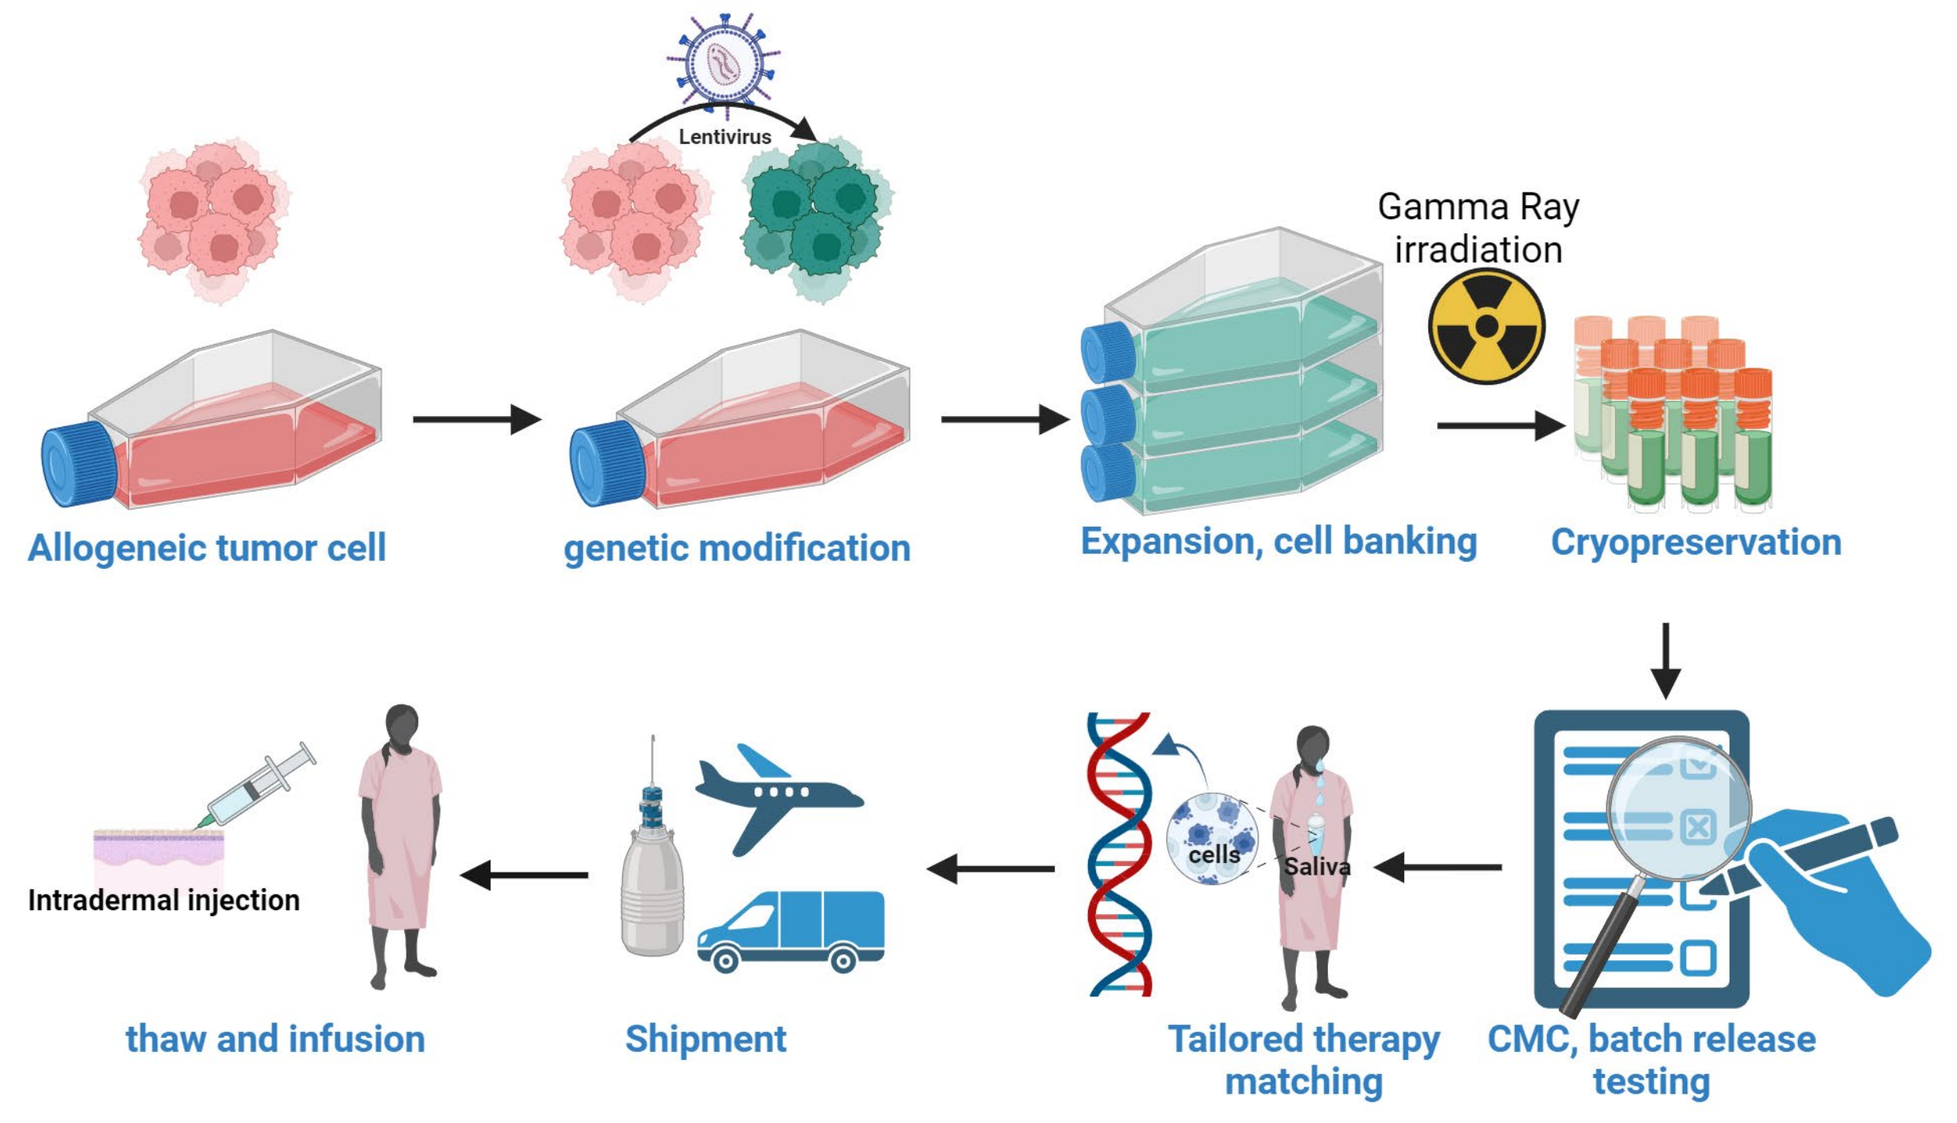
Lentivirus
Gamma Ray irradiation
Allogeneic tumor cell
genetic modification
Expansion, cell banking
Cryopreservation
Intradermal injection
cells
Saliva
thaw and infusion
Shipment
Tailored therapy matching
CMC, batch release testing

1. ***Allogeneic Tumor Cell****: The process begins with the selection of allogeneic tumor cells*
2. ***Genetic Modification:*** *Tumor cells are genetically modified*
3. ***Expansion and Cell Banking****: The modified cells are expanded to create a sufficient quantity, followed by banking them to maintain a consistent supply.*
4. ***Gamma Ray Irradiation:*** *Cells undergo gamma irradiation to ensure they cannot proliferate while retaining their immunogenic properties.*
5. ***Cryopreservation:*** *The irradiated cells are cryopreserved, allowing them to be stored until needed for patient treatment.*
6. ***CMC and Batch Release Testing****: Each batch undergoes rigorous testing for quality control (CMC: Chemistry, Manufacturing, and Controls) to ensure safety and efficacy.*
7. ***Tailored Therapy Matching****: Patients' genetic information (e.g., from saliva samples) is used to match them with the most suitable cell-based therapy.*
8. ***Shipment:*** *The tailored vaccine product is shipped under controlled conditions to maintain integrity until it reaches the clinical site.*
9. ***Thaw and Inoculation:*** *The cryopreserved cells are thawed and prepared for inoculation, typically administered via intradermal injection.*
10. ***Intradermal Injection****: The vaccine is delivered to the patient, where it is designed to stimulate an immune response against cancer cells*

CONCLUSIONS

Bria-OTS+ exhibits the following characteristics:

- **Expresses a variety of cancer-related antigens, including Tumor-Associated Antigens (TAAs) and Post-Translational Modifications (PTMs) (data presented previously).**
- **Engages multiple facets of the adaptive immune response.**
- **Activates components of the innate immune system.**
- **Counteracts immune escape mechanisms, specifically activating Natural Killer (NK) cells in the case of HLA deletions.**
- **Designed for both personalized and ready-to-use applications, ensuring robust long-term stability.**
- **Simplified administration process.**
- **Anticipated to have a favorable side effect profile, indicating good tolerance.**