Engineering semi-allogeneic whole cell cancer vaccines with enhanced immunogenicity for the treatment of advanced solid tumors.

Miguel Lopez-Lago[1], Vikas Bhardwaj[1], Xiaoyi Zheng[1], Sagarika Pachhal[1] Renee Cortez[1] , Patience Cournoo[1], Charles L. Wiseman[1], William V. Williams[1]

[1]BriaCell Therapeutics Corp. Philadelphia, PA

AACR annual meeting– Orlando
April 14-19, 2023

ABSTRACT

Therapeutic cancer vaccines are designed to program a patient's own immune system to recognize and eliminate tumor cells. We sought to harness gene-modified tumor cells as a vaccine platform and developed cancer vaccines composed of breast cancer cells expressing GM-CSF (SV-BR-1-GM)(Fig.1**). We have recently reported favorable clinical outcomes in patient populations that match SV-BR-1-GM at one or more HLA alleles (poster CT143).** Mechanistically, SV-BR-1-GM cells can directly activate primed CD4+ T-cells in an antigen-specific HLA-restricted manner, as demonstrated by an in vitro antigen presentation assay (1). These observations led us to hypothesize that semi-allogeneic cell-based cancer vaccines (partially matching host HLA genotype) would mount an enhanced immune response by forcing the host immune response to recognize tumor-associated antigens in the context of allogeneic HLA-I or II molecules or in proximity of strong non-self-antigens (Fig.2,3). Therefore, steps were taken to generate semi-allogeneic cancer cell lines expressing an extended repertoire of stimulatory molecules to induce optimal naïve T-cell activation (**Bria-OTS**).

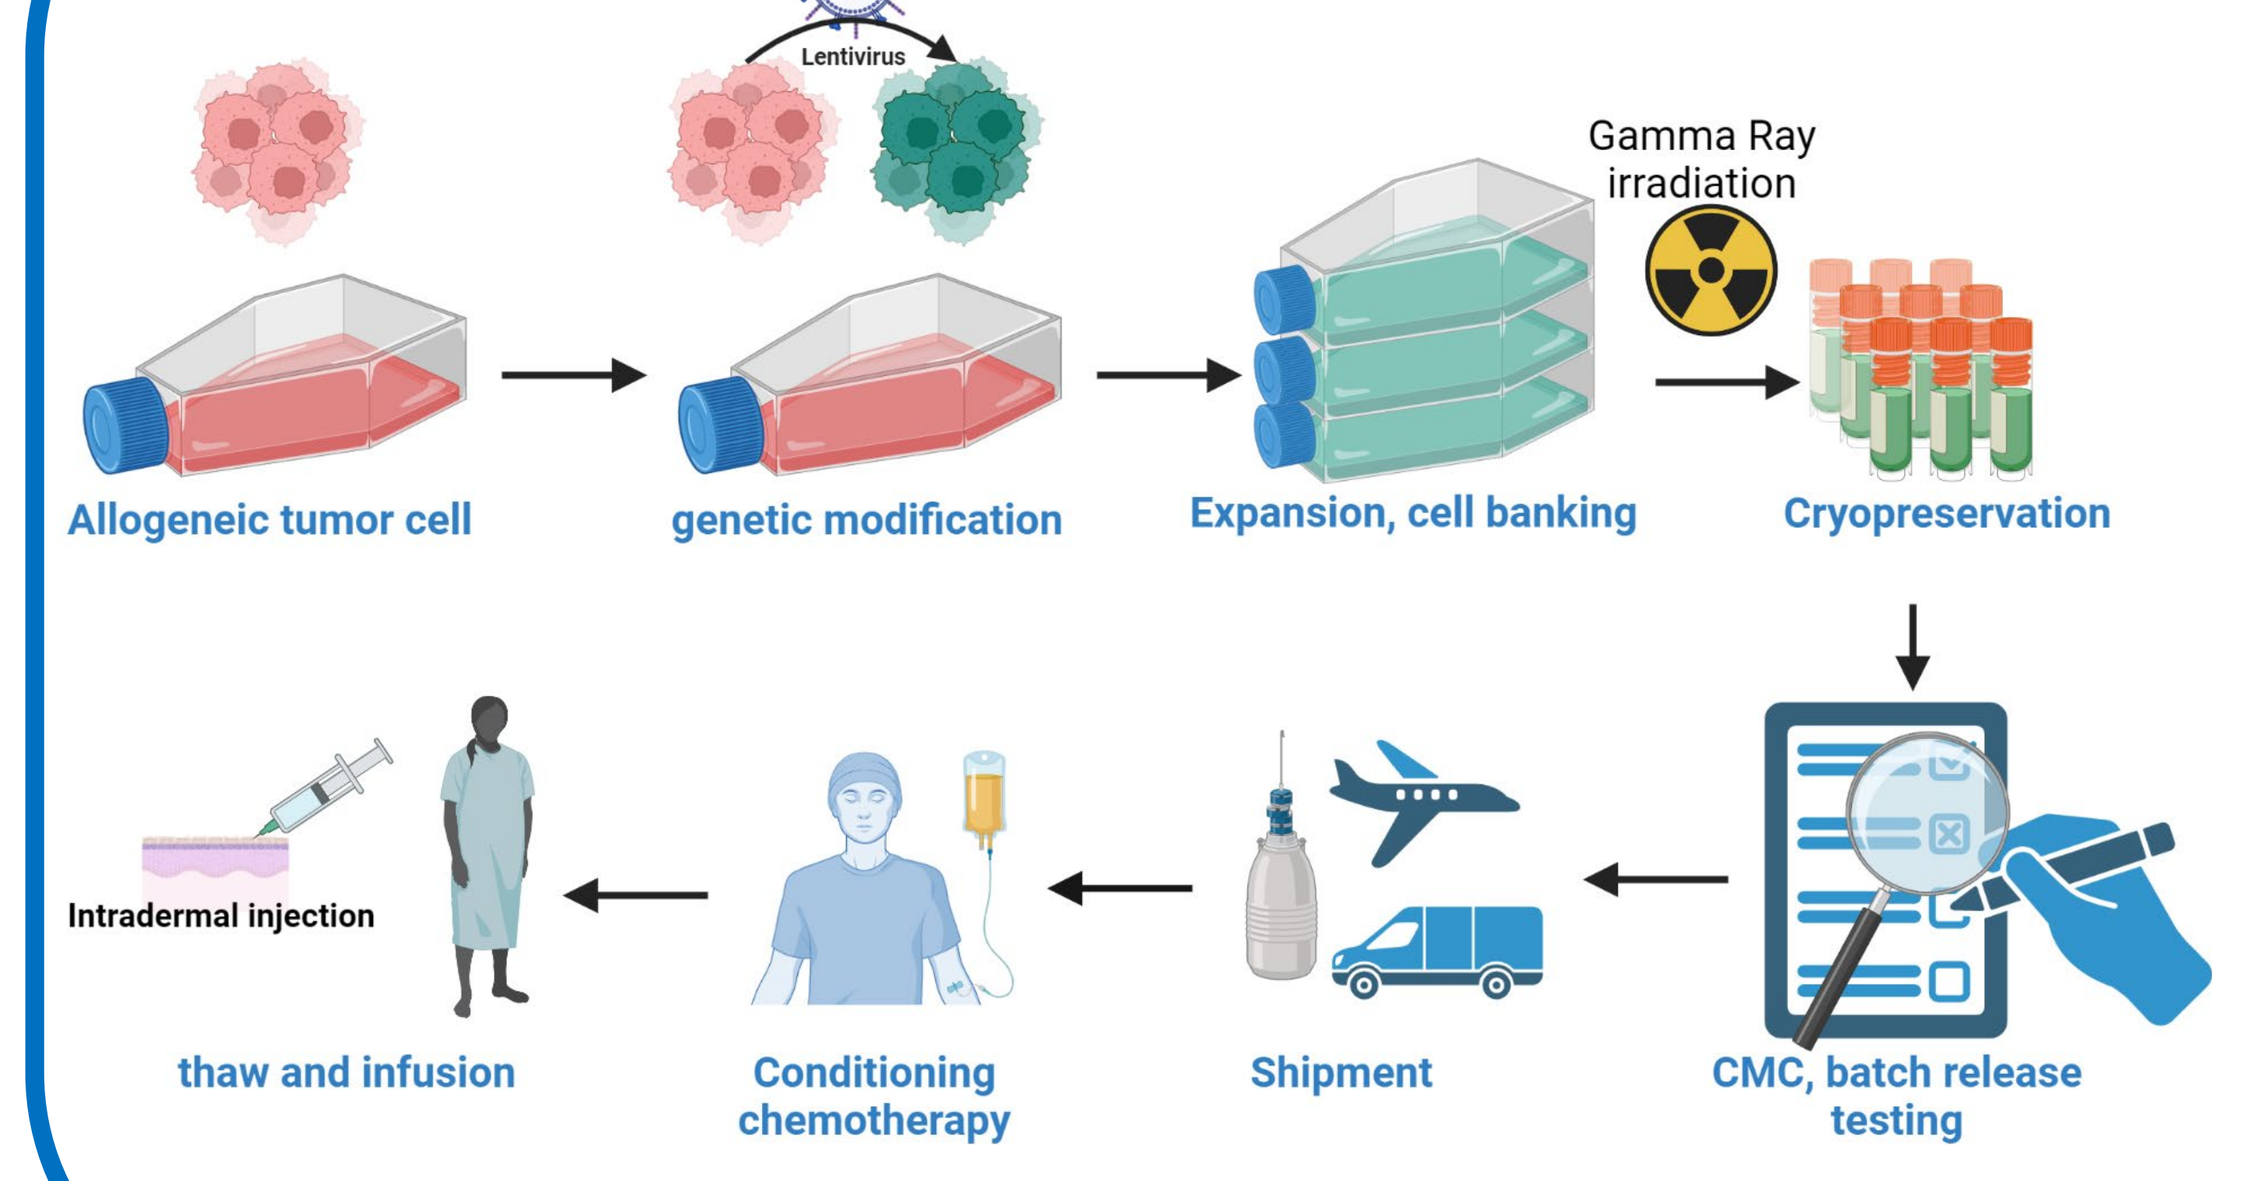
Lentivirus
Gamma Ray irradiation
Allogeneic tumor cell
genetic modification
Expansion, cell banking
Cryopreservation
Intradermal injection
thaw and infusion
Conditioning chemotherapy
Shipment
CMC, batch release testing

Fig.1. BriaCell cell-based cancer immunotherapy workflow. *Allogenic cancer cell lines were generated in house or purchased from ATCC. Previously manufactured lentivirus-based vectors are used to modify the cancer cell lines, conferring new immunomodulatory properties. The modified cells are then expanded to therapeutically relevant numbers, followed by washing, irradiation to render the cells replication incompetent, formulation, bagging, and cryostorage. After that, the cell product is shipped to the clinical site where it is thawed and administered to the patient intradermally. SV-BR-1-GM was used in 3 clinical studies: In two "Monotherapy" studies the SV-BR-1-GM regimen consisted of low dose cyclophosphamide to reduce immune suppression (300 mg/m^2 2-3 days prior to inoculation); 20-40 million irradiated SV-BR-1-GM cells intradermally split into 4 sites; and interferon-α2b (10,000 IU) or peginterferon-alpha-2a (0.1 µg x 4) into the inoculation sites ~2 & ~4 days later with cycles every 2 weeks x3 then monthly. For combination therapy, pembrolizumab (200 mg IV) or retifanlimab (375 mg IV) was given in combination with the SV-BR-1-GM regimen with cycles every 3 weeks.*

BACKGROUND

Despite great advances, current immunotherapies are effective only for a very limited number of patients. Cancer vaccines hold promise as they often lead to strong and durable immune responses. Several approaches have been used to develop cancer vaccines, including peptide and protein tumor-associated antigens (TAAs) or neoantigens, dendritic cells pulsed with TAAs, and whole tumor cells. The advantage of a whole-cell vaccine is that it delivers a range of TAAs. Despite great efforts and promising results in early phase studies, whole cell cancer vaccines have shown low clinical efficacy, likely due to multiple factors including but not limited to poor immunogenicity, lack of co-stimulatory molecules, downregulation of HLA molecules, and immune suppression by the tumor microenvironment. To overcome these obstacles, BriaCell is developing Bria-OTS cell lines that express TAAs, immunostimulatory cytokines, co-stimulatory molecules, and HLA genes, enabling direct presentation of tumor antigens to T cells, leading to an enhanced immune response.

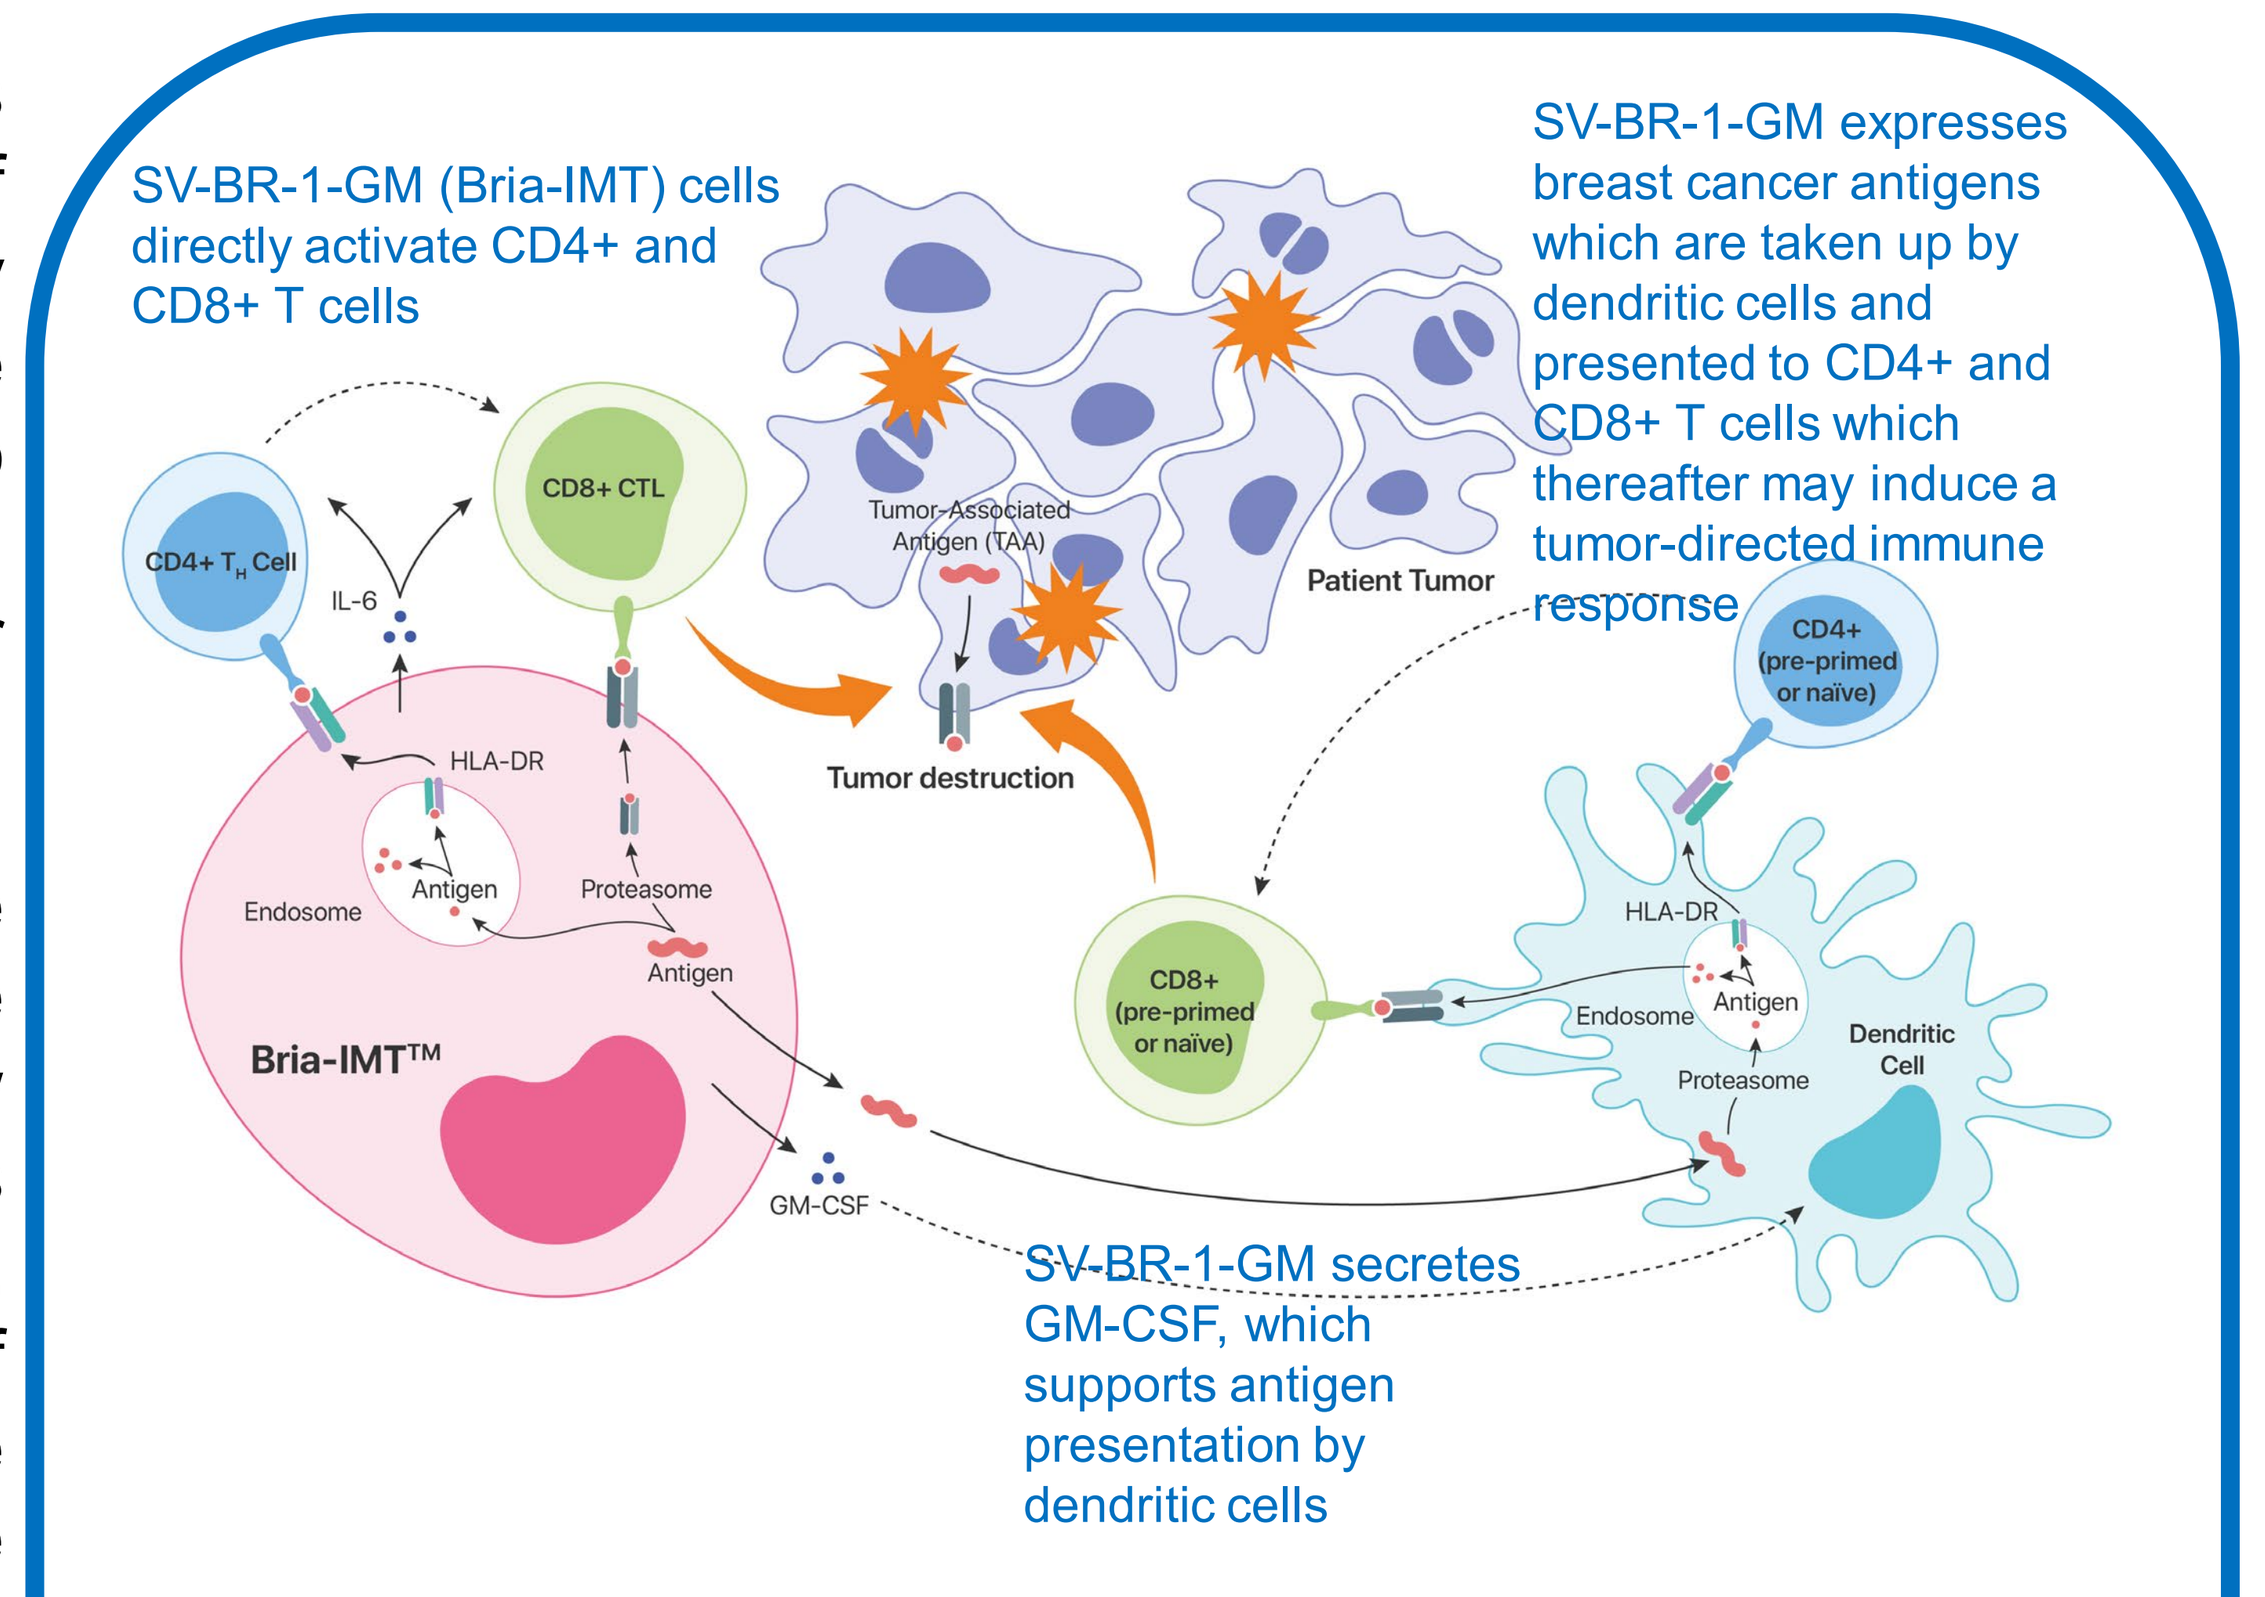
SV-BR-1-GM (Bria-IMT) cells directly activate CD4+ and CD8+ T cells
SV-BR-1-GM expresses breast cancer antigens which are taken up by dendritic cells and presented to CD4+ and CD8+ T cells which thereafter may induce a tumor-directed immune response
SV-BR-1-GM secretes GM-CSF, which supports antigen presentation by dendritic cells
CD8+ CTL
IL-6
HLA-DR
Endosome
Antigen
Proteasome
Bria-IMT™
GM-CSF
Tumor-Associated Antigen (TAA)
Patient Tumor
Tumor destruction
CD8+ (pre-primed or naïve)
CD4+ (pre-primed or naïve)
Dendritic Cell

Fig.2 Mechanism of Action (MoA)
SV-BR-1-GM (Bria-IMT™) acts as an antigen-presenting cell for primed T cells (Lacher *et al.*, Front Immunol. 2018 May 15;9:776).

OBJECTIVES

Based on our previous results we postulated that immunization of patients with an allogeneic whole cell vaccine would be more effective if the tumor cells, in addition to providing tumor antigens, could activate/prime naïve T cells directly. To achieve this goal, tumor cells should express:

- Both HLA alleles that match and don't match host T-cells.
- Co-stimulatory molecules (CD80, CD86. 4-1BBL) and immuno-modulatory cytokines (GM-CSF, IFNα, IL7, IL12): This modification should improve direct presentation of antigens by tumor cells to patient's T cells.

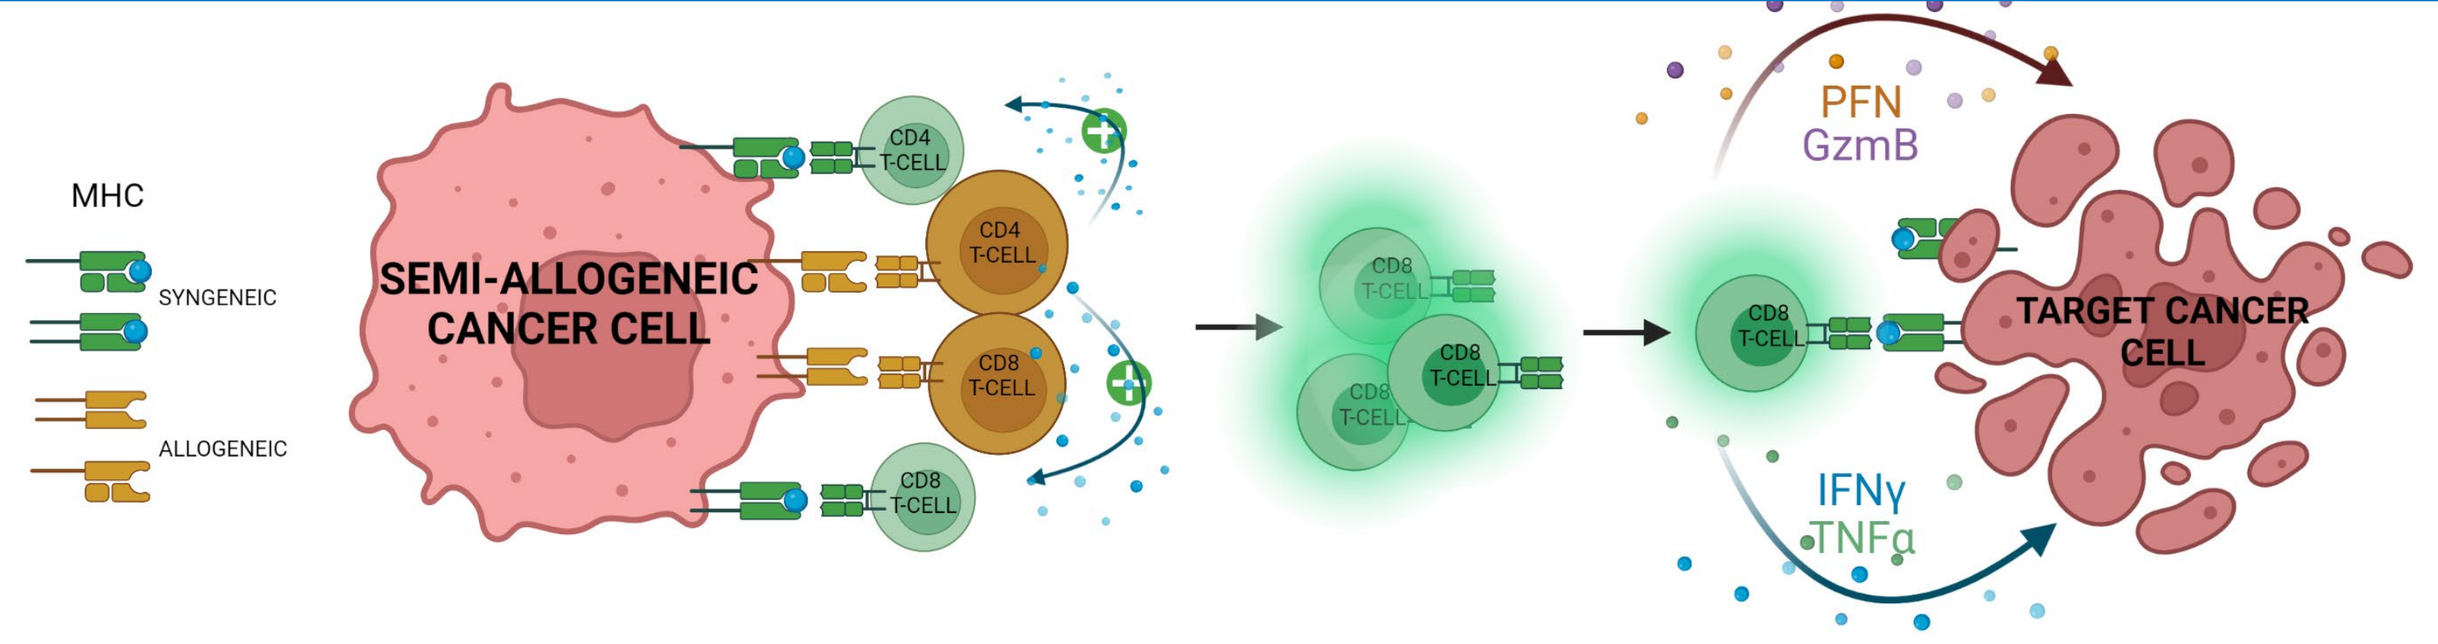
MHC
SYNGENEIC
ALLOGENEIC
SEMI-ALLOGENEIC CANCER CELL
CD4 T-CELL
CD8 T-CELL
PFN
GzmB
IFNγ
TNFα
TARGET CANCER CELL

Fig.3. *Direct CD4+ T-cell allorecognition for provision of T-cell help for the generation of self-MHC restricted T-cell responses to tumor peptides.*

METHODS

- Engineered Bria-OTS cell lines were derived from **breast cancer** (SV-BR-1), **prostate cancer** (PC3), **melanoma** (SK-MEL-24), and **lung cancer** cells (NCI-H2228). These cell lines have been chosen based on the expression of a characteristic 22-gene immune signature, which was originally characterized in SV-BR-1 cells
- **Expression of Co-stimulatory molecules and immuno-modulatory cytokines** Tumor cells were genetically modified to express co-stimulatory molecules and immunomodulatory cytokines by using a lentiviral mediated expression system. Resulting cells were named antigen presenting tumor cells (APTC) (Fig 4, 5B).
- **Expression of allele-specific HLA molecules:** To generate an off-the-shelf semi-allogeneic cell therapy covering most of the population, SV-BR-1 was genetically modified to express an extended repertoire of HLA alleles (Fig.5B). Based on population analysis, four cell lines, each carrying two (2) *HLA-A and two (2) HLA-DRB3/4/5* alleles, should produce at least a single match in 99% of the population, with a 92% match at Class I HLA-A alleles and a 98% match at Class II HLA-DRB3/4/5 alleles (Fig.4, 5A) .
- **Functional Validation:**
 1. **Modified mixed lymphocyte reaction assay**
 2. **T-cell activation assay using NFAT driven luciferase transgenic Jurkat cells**

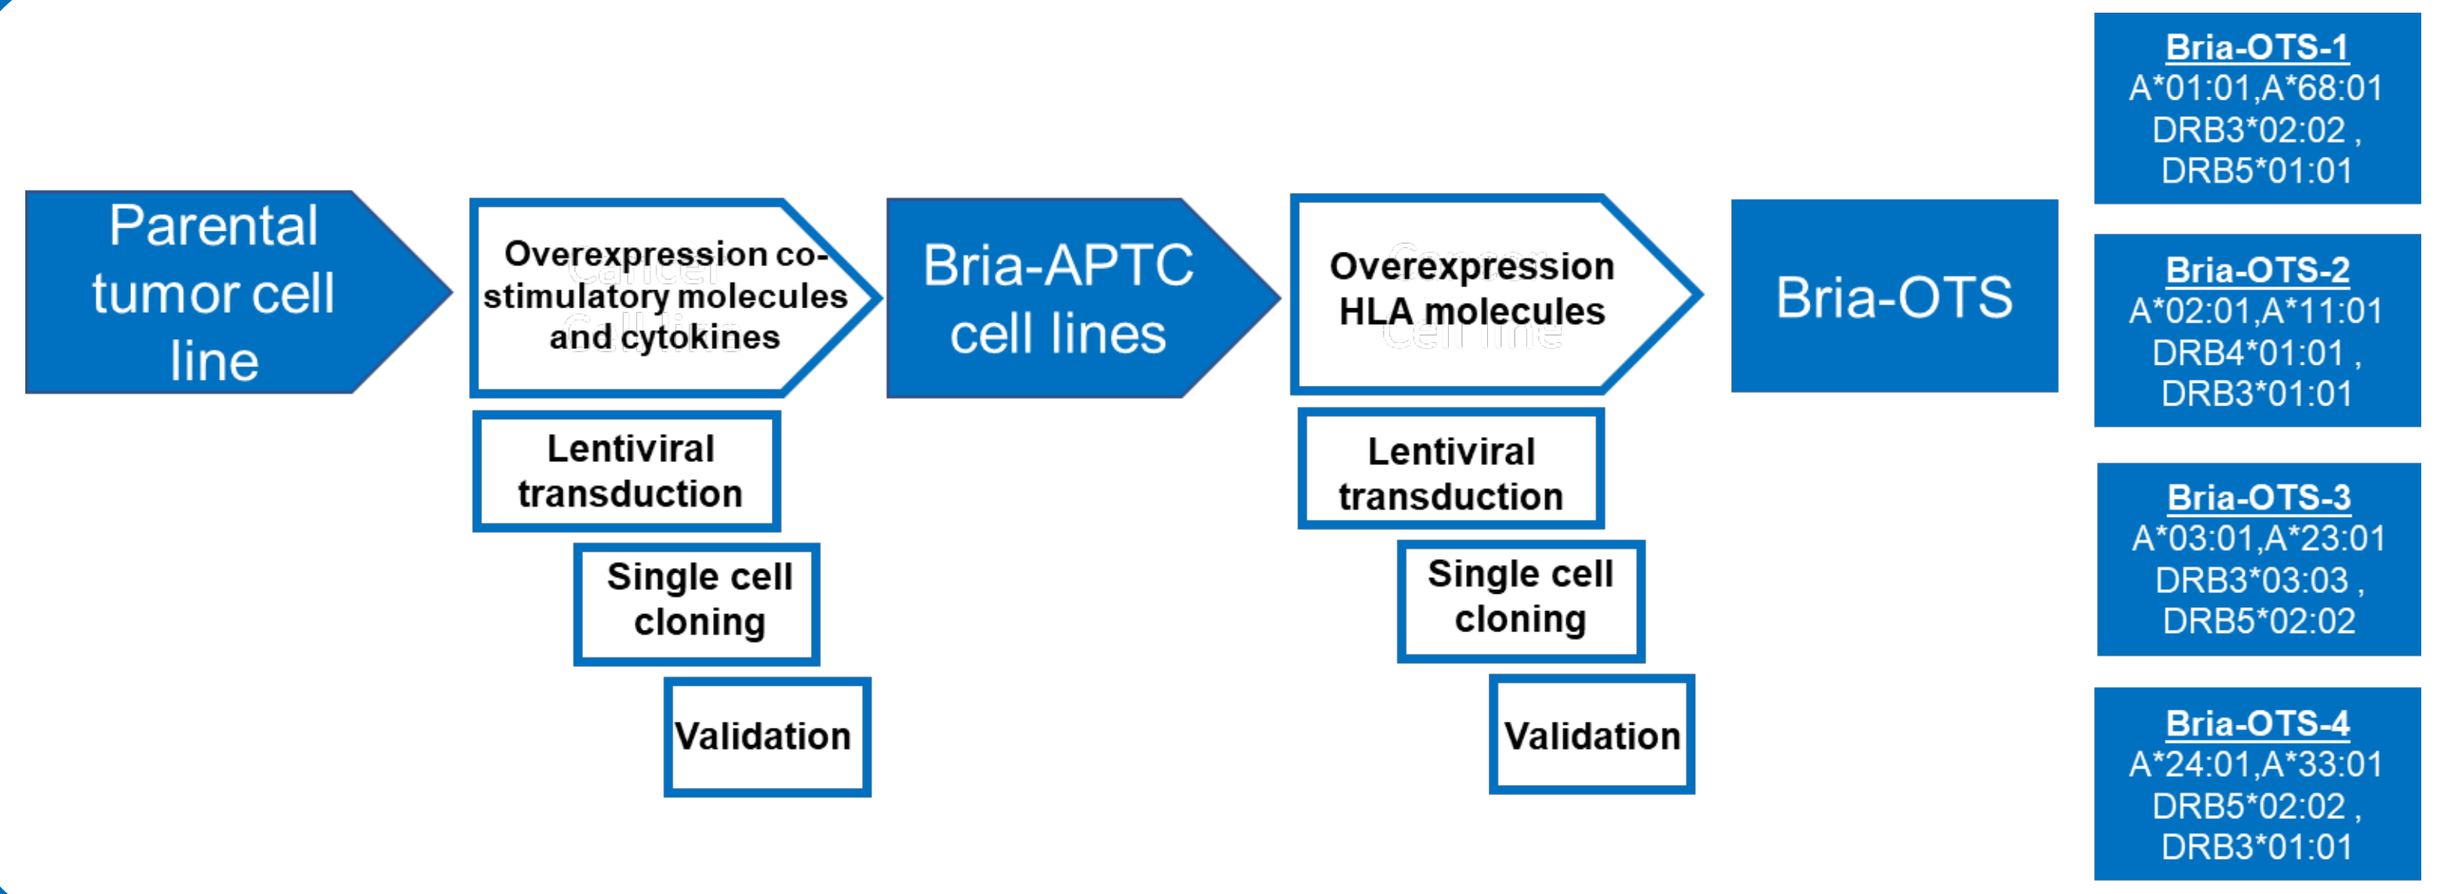
Parental tumor cell line
Overexpression co-stimulatory molecules and cytokines
Lentiviral transduction
Single cell cloning
Validation
Bria-APTC cell lines
Overexpression HLA molecules
Lentiviral transduction
Single cell cloning
Validation
Bria-OTS
Bria-OTS-1
A*01:01,A*68:01
DRB3*02:02 ,
DRB5*01:01
Bria-OTS-2
A*02:01,A*11:01
DRB4*01:01 ,
DRB3*01:01
Bria-OTS-3
A*03:01,A*23:01
DRB3*03:03 ,
DRB5*02:02
Bria-OTS-4
A*24:01,A*33:01
DRB5*02:02 ,
DRB3*01:01

Fig. 4 Experimental strategy to develop Bria- OTS cell lines. *First, GM-CSF and other cytokines and co stimulatory molecules are overexpressed by sequential lentiviral transduction of 4 unique constructs to generate Bria-APTC cells. In addition, two HLA-A and two HLA-DRB alleles are overexpressed by lentiviral transduction, to generate a collection of 4 cell lines (Bria OTS 1 2 3 4)*

A

Co-stimulatory molecules and cytokines		
Lentivirus ID	***EF1a Promoter***	***MNDU3 Promoter***
GM-IFN	CSF2 (GM-CSF)	IFNA2 (IFNα)
CD86-IL12	CD86	IL12
CD80-HLA-DRA	CD80	HLA-DRA
IL7-4-1BBL	IL7	4-1BBL (TNFSF9;CD137L)

B

HLA molecules		
Lentivirus ID	***EF1a Promoter***	***MNDU3 Promoter***
A2-A11	HLA-A*02:01	HLA-A*11:01
A1-A68	HLA-A*01:01	HLA-A*68:01
A3-A23	HLA-A*03:01	HLA-A*23:01
A24-A33	HLA-A*24:02	HLA-A*33:03
DR401-301	DRB4*01:01	DRB3*01:01
DR302-501	DRB3*02:02	DRB5*01:01
DR303-502	DRB3*03:01	DRB5*01:02
DR301-522	DRB5*02:02	DRB3*01:01

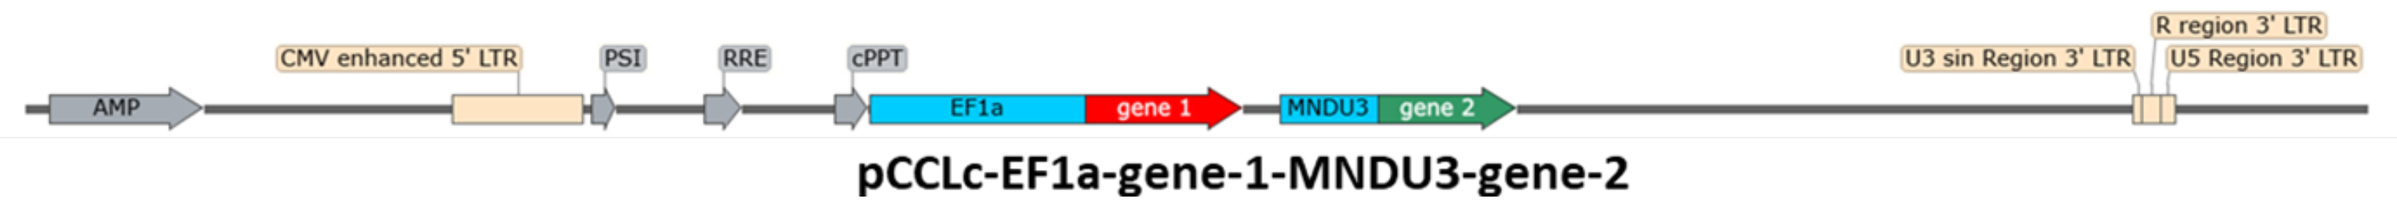
AMP
CMV enhanced 5' LTR
PSI
RRE
cPPT
EF1a
gene 1
MNDU3
gene 2
U3 sin Region 3' LTR
R region 3' LTR
U5 Region 3' LTR
pCCLc-EF1a-gene-1-MNDU3-gene-2

Fig. 5.*Lentiviral vectors used to express 2 different genes from independent promoters. Cytokines and co-stimulatory molecules (**A**) used to generate Bria-APTC cells. List of plasmids expressing HLA-A and HLA-DRB alleles (**B**) for final Bria-OTS products.*

RESULTS

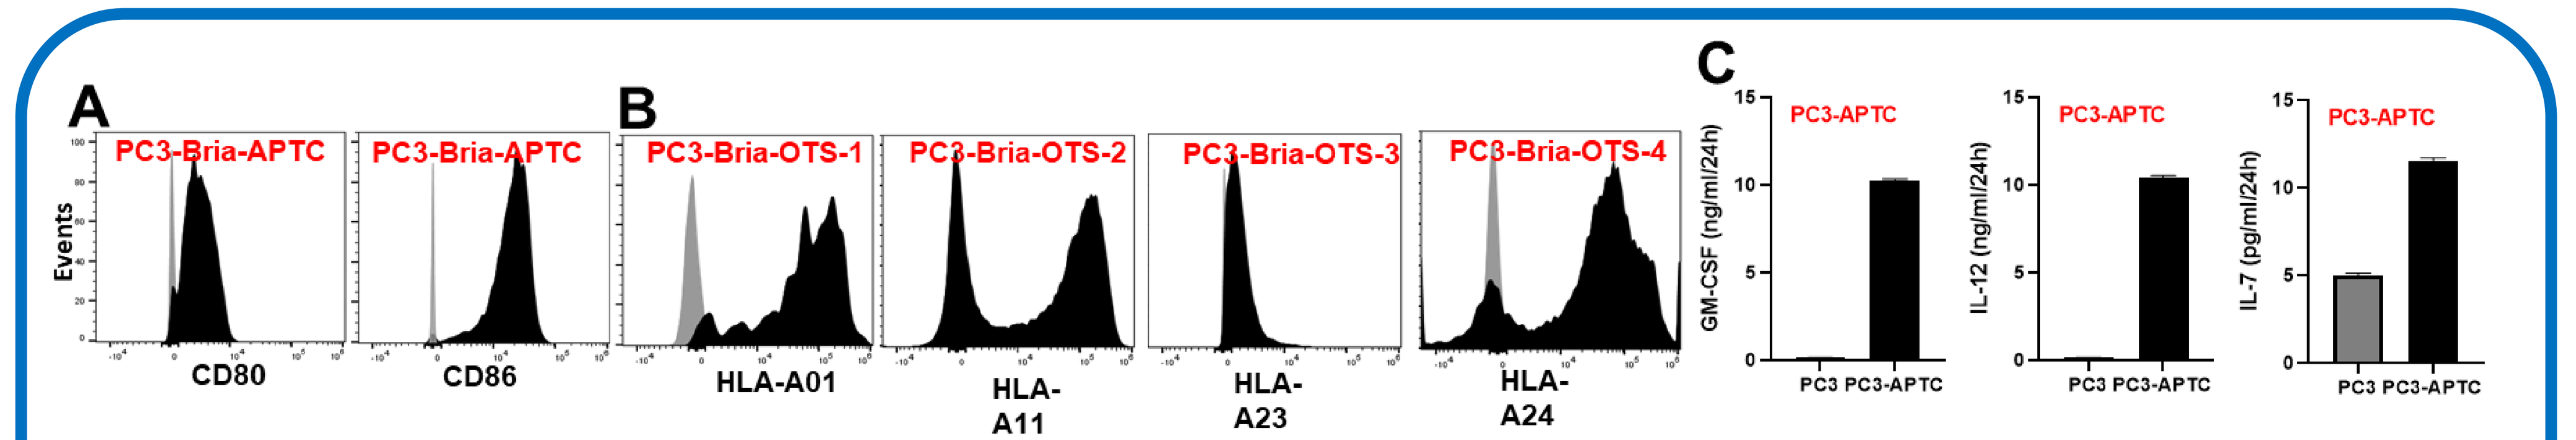
A
B
C
Events
PC3-Bria-APTC
PC3-Bria-APTC
CD80
CD86
PC3-Bria-OTS-1
PC3-Bria-OTS-2
PC3-Bria-OTS-3
PC3-Bria-OTS-4
HLA-A01
HLA-A11
HLA-A23
HLA-A24
PC3-APTC
GM-CSF (ng/ml/24h)
IL-12 (ng/ml/24h)
IL-7 (pg/ml/24h)
PC3 PC3-APTC

Fig.6. Expression of GM-CSF and cytokines/costimulatory molecules in PC3-APTC cells HLA-A alleles on PC3-Bria-OTS. *PC3 were sequentially transduced with lentiviruses expressing 8 different cytokines and co-stimulatory molecules and cloned.* ***A)*** *Expression of surface receptors CD86 and CD80 in SV-BR-1-APTC cells as measured by flow cytometry.* ***B)*** *Expression of HLA-A alleles in SV-BR-1-Bria-OTS cell lines as measured by flow cytometry* ***C)*** *Levels of secreted GM-CSF, IL-12, and IL-7 were measured by ELISA*

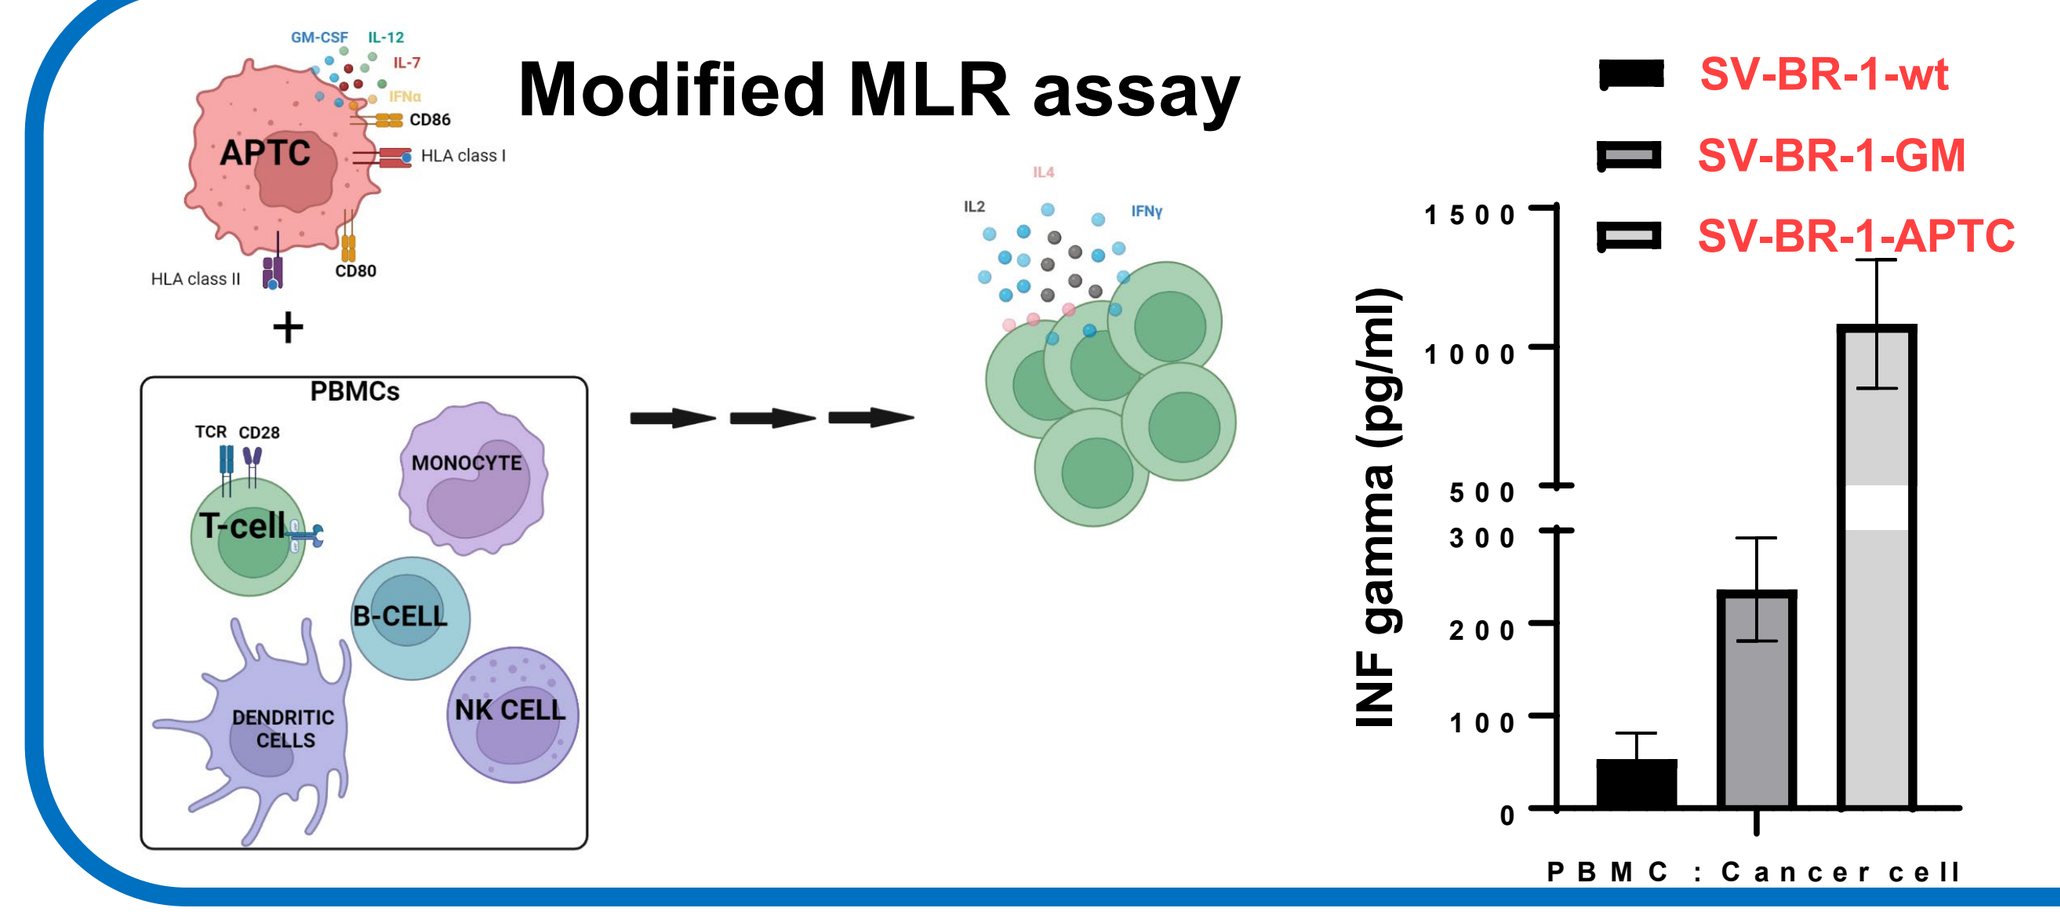
Modified MLR assay
APTC
GM-CSF
IL-12
IL-7
CD86
HLA class I
HLA class II
CD80
PBMCs
T-cell
MONOCYTE
B-CELL
DENDRITIC CELLS
NK CELL
IL2
IL4
IFNγ
SV-BR-1-wt
SV-BR-1-GM
SV-BR-1-APTC
INF gamma (pg/ml)
PBMC : Cancer cell

Fig.7. ***Functional characterization of Bria-OTS breast.*** *Indicated tumor cell lines were co-cultured with PBMC for 24 hours and media was collected and analyzed for IFNɣ production using ELISA. Cancer cell:PBMCs ratio 1:10.*

Conclusion: ***Tumor cell lines that express co-stimulatory molecules and immuno-modulatory cytokines are able to activate naïve T-cells***

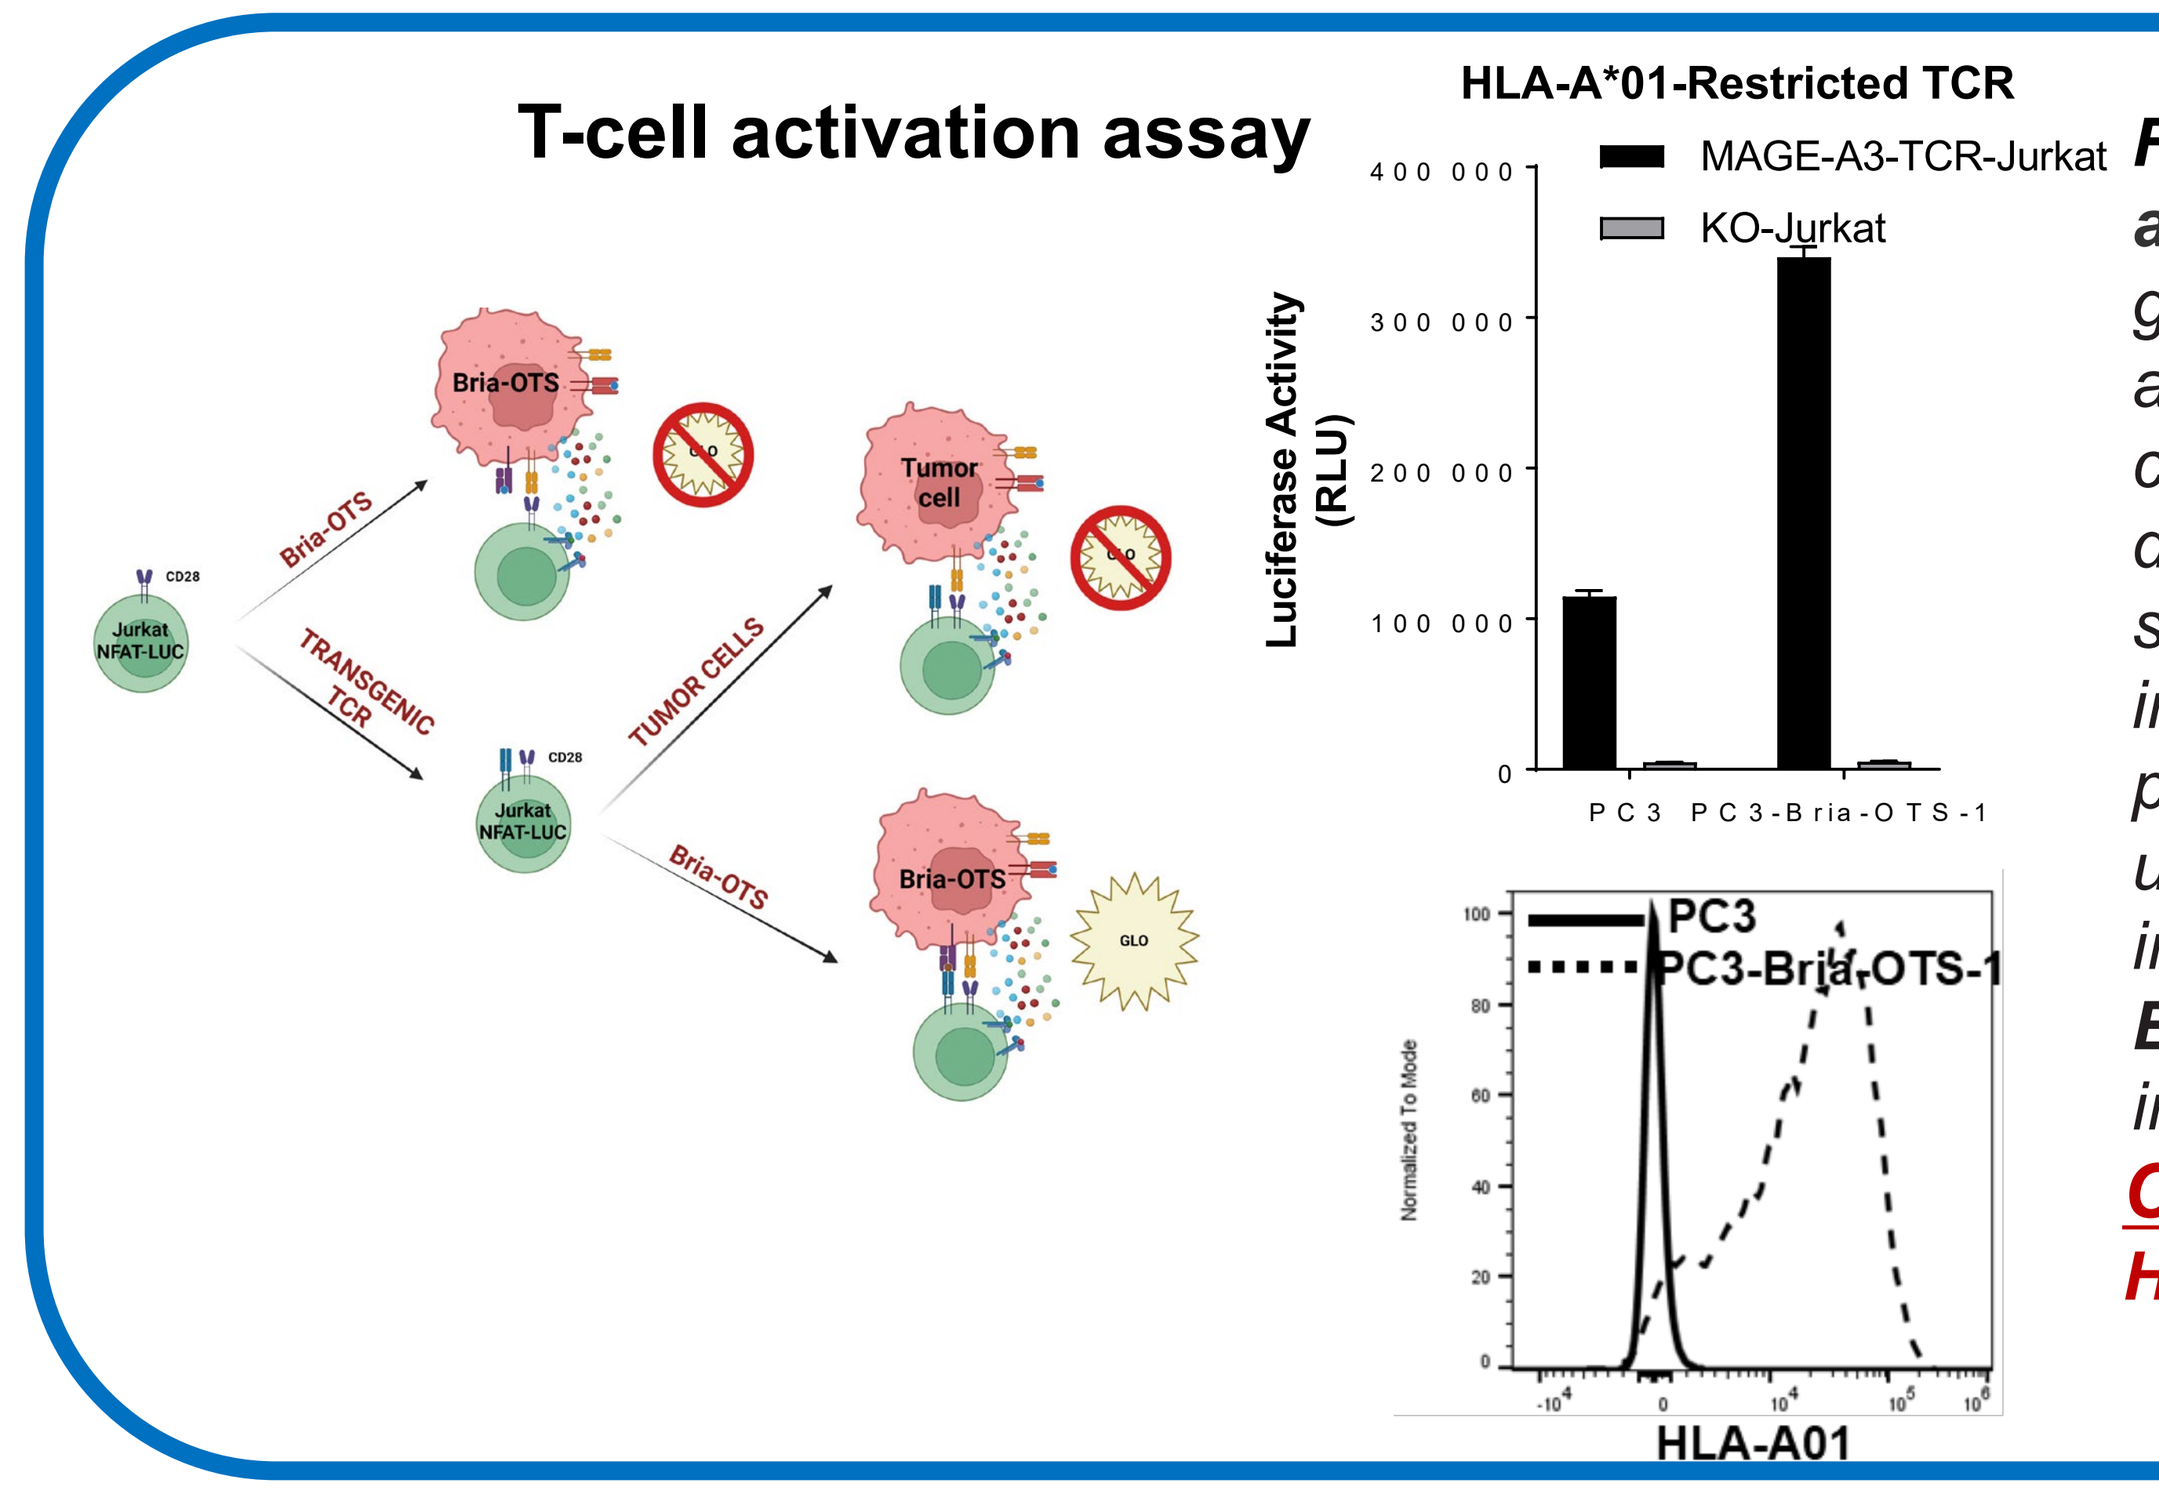
T-cell activation assay
Jurkat NFAT-LUC
Bria-OTS
TRANSGENIC TCR
TUMOR CELLS
Tumor cell
GLO
HLA-A*01-Restricted TCR
MAGE-A3-TCR-Jurkat
KO-Jurkat
Luciferase Activity (RLU)
PC3
PC3-Bria-OTS-1
Normalized To Mode
HLA-A01

Fig.8. Validation of HLA functionality on PC3-Bria-OTS using a T Cell Activation Bioassay (NFAT). *The assay consists of a genetically engineered Jurkat T cell line with endogenous TCR alpha and beta chains knocked out using CRISPR-Cas9. These cells also express a luciferase reporter driven by a TCR pathway-dependent promoter.* ***A.*** *Jurkat were transduced with a TCR specific for MAGE-A3 peptide (HLA-A01 restricted) and incubated with the indicated cell lines and appropriated WT1 peptides. After 6 hours, The bioluminescent signal is quantified using the Bio-Glo-NL™ Luciferase Assay System. Transgenic HLA induces cell activation at higher rates than endogenous HLAs.* ***B.*** *Flow cytometry analysis showing HLA-A01 expression in the indicated cell lines.*

Conclusion: Tumor cell lines that express transgenic HLAs are able to induce TCR signaling.

DISCUSSION AND CONCLUSIONS

We are in the process creating ''off the shelf" personalized cell-based therapeutic cancer vaccines that induce potent T-cell responses for a variety of solid tumors. The Bria-OTS cell line collection will deliver the following features:

- **Dual mechanisms of action to achieve strong immune response and clinical activity**. Bria-OTS cell lines have features of both cancer cells (expressing a myriad of TAAs) and dendritic cells (by presenting TAAs directly to T cells) to enhance the immune response.
- **Precision therapy.** Bria-OTS cell lines will be matched to patients based upon HLA antigens, covering over 99% of the US population .
- **Improved safety.** Relative to current chemotherapeutic and hormone-based therapies that are associated with severe adverse events that can be life threatening.
- **Rapid, cost-effective treatment**. Our "off-the-shelf" cell lines will not require personalized manufacturing and can be administered immediately after patient *HLA* genotyping.

References:

1. Lacher MD et al, Front Immunol. 2018 May 15;9:776

This presentation is the intellectual property of the author/presenter. Contact them at info@briacell.com for permission to reprint and/or distribute.