Turning tumor cells into antigen-presenting cells for cancer immunotherapy

Miguel Lopez-Lago[1] , Sagarika Pachhal[1], Vikas Bhardwaj[1], Charles L. Wiseman[1], William V. Williams[1]
[1]BriaCell Therapeutics Corp., Philadelphia, PA

San Antonio Breast Cancer Symposium – December 7-10, 2022

BACKGROUND

Therapeutic cancer vaccines are based on specific stimulation of the immune system using tumor antigens to elicit an antitumor response. We have been conducting clinical trials using the breast cancer cell line SV-BR-1-GM as a therapeutic vaccine (**See poster # P3-07-12**). SV-BR-1-GM is a GM-CSF expressing breast cancer cell line with features of an antigen presenting cell (APC) owning to the expression of several immunomodulatory molecules, including MHC-I (HLA-A, B & C) and MHC-II (HLA-DRB3 & -DRA). Initial results in patients treated with irradiated SV-BR-1-GM cells, low dose cyclophosphamide and local IFNα suggest that patients that match SV-BR-1-GM at least at 1 HLA allele are more likely to derive clinical benefit. This clinical observation, together with the fact that SV-BR-1-GM cells can directly activate CD4+ T-cells in an antigen-specific HLA-restricted manner, as demonstrated by an in vitro antigen presentation assay (1), lead us to hypothesize that SV-BR-1 (the parent cell line) **can function as APC** (Fig.1). To further enhance direct antigen presentation to T-cells, SV-BR-1 cells have been genetically modified to express co-stimulatory molecules, immunomodulatory cytokines, and an extended repertoire of HLA alleles.

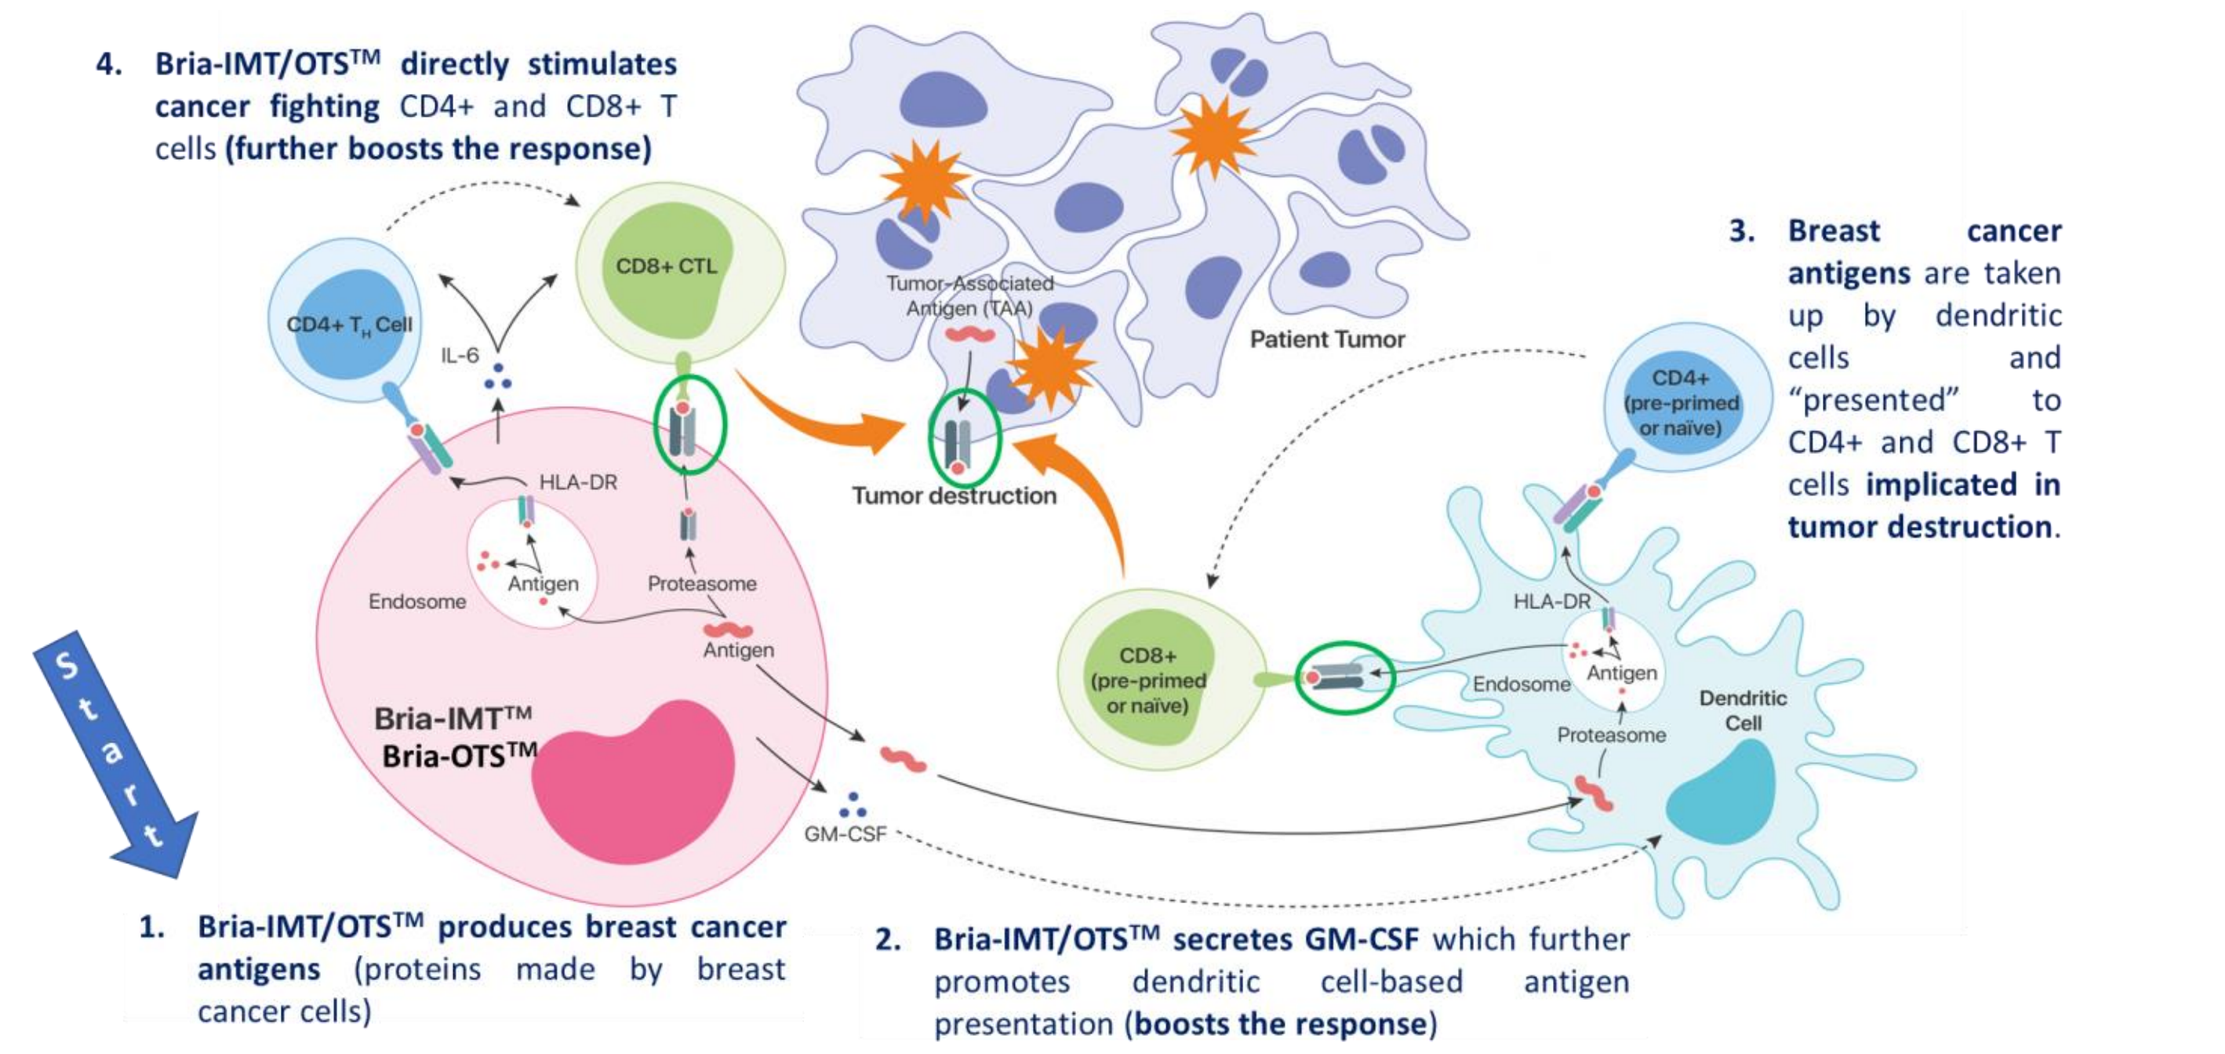
4. Bria-IMT/OTS™ directly stimulates cancer fighting CD4+ and CD8+ T cells (further boosts the response)
CD4+ T_H Cell
IL-6
CD8+ CTL
Tumor-Associated Antigen (TAA)
Patient Tumor
Tumor destruction
3. Breast cancer antigens are taken up by dendritic cells and "presented" to CD4+ and CD8+ T cells implicated in tumor destruction.
CD4+ (pre-primed or naïve)
HLA-DR
Endosome
Antigen
Proteasome
Bria-IMT™
Bria-OTS™
CD8+ (pre-primed or naïve)
Dendritic Cell
GM-CSF
Start
1. Bria-IMT/OTS™ produces breast cancer antigens (proteins made by breast cancer cells)
2. Bria-IMT/OTS™ secretes GM-CSF which further promotes dendritic cell-based antigen presentation (boosts the response)

Figure 1. Dual mechanism of action of Bria-IMT and Bria-OTS therapeutics. *SV-BR-1-GM cells secrete GM-CSF that supports antigen presentation by DCs. Cancer cell antigens, following degradation of cells, are taken up by DCs and presented to CD4⁺ and CD8⁺ T cells, which induce a tumor-directed immune response. SV-BR-1-GM cells can also directly activate T cells in an antigen-specific HLA-restricted manner, as an additional boost to the immune response.*

RATIONALE AND OBJECTIVES

Based on our previous results we postulated that immunization of patients with an allogeneic whole cell vaccine would be more effective if the tumor cells, in addition to provide tumor antigens, could activate/prime naïve T-cells directly. To achieve this goal, we will generate genetically modified breast cancer cell lines expressing an extended repertoire of stimulatory molecules required for optimal T-cell activation/priming:

- **Allele-specific HLA molecules**. This modification will allow that multiple antigens shared by both the immunizing cell line and the patient's tumor are presented by shared HLA molecules, leading to a specific anti-tumor immune response.
- **Co-stimulatory molecules (CD80, CD86. 41BB-L) and immuno-modulatory cytokines (GM-CSF, IFNα, IL7, IL12):** This modification will improve direct presentation of antigens by tumor cells to patient's T-cells.

METHODS

- **Crispr/Cas9 HLA-A and HLA-DRB3:** SV-BR-1 was genetically modified using CRISPR/cas9 deletion of the endogenous HLA-A and HLA-DRB3 alleles and subsequent lentiviral mediated expression of alternative HLA-A and DRB3 alleles (Fig.2).
- **Expression of Co-stimulatory molecules and immuno-modulatory cytokines** SV-BR-1 cells were genetically modified to express co-stimulatory molecules and immunomodulatory cytokines by using a lentiviral mediated expression system. Resulting cells were named antigen presenting tumor cells (APTC) (Fig 2,3A).
- **Expression of allele-specific HLA molecules:** To generate an off-the-shelf semi-allogeneic cell therapy covering most of the population, SV-BR-1 was genetically modified to express an extended repertoire of HLA alleles (Fig.3B). Based on population analysis, four cell lines, each carrying two (2) *HLA-A and two (2) HLA-DRB3/4/5* alleles, should produce at least a single match in 99% of the population, with a 92% match at Class I HLA-A alleles and a 98% match at Class II HLA-DRB3/4/5 alleles (Table 1) .
- **Validation:** Expression and functionality of the stimulatory molecules was established using flow cytometry and cell-based assays (Fig.4).

METHODS

	Alleles in Bria-OTS	African American	White	Asian
US Census 2010 (Frequencies)		12.60%	72.4	4.8
Sub-Population		AAFA	EURCAU	JAPI
		Frequency	Frequency	Frequency
HLA-A	A*02:01	12.30%	27.60%	14.80%
HLA-A	A*01:01	4.70%	16.50%	1%
HLA-A	A*03:01	8.40%	14%	0.90%
HLA-A	A*24:02	2.50%	8.50%	35.30%
HLA-A	A*11:01	1.40%	6.10%	8.70%
HLA-A	A*68:01	4%	3.20%	0.20%
HLA-A	A*23:01	11%	2%	0.10%
HLA-A	A*33:03	5.20%	0.3	6.50%
At least 1 HLA-A match	Sum of allele frequencies:	49.40%	78	67.50%
At least 1 HLA-A match	Per individual (2n):	74.40%	95%	89.40%

	Alleles in Bria-OTS	African American	White	Asian
US Census 2010 (Frequencies)		12.6	72.4	4.8
Sub-Population		AAFA	EURCAU	JAPI
		Frequency*	Frequency*	Frequency*
DRB3/4/5	DRB4*01:01	18.30%	31.20%	38.40%
DRB3/4/5	DRB3*02:02	27.20%	18.20%	10.40%
DRB3/4/5	DRB3*01:01	13.40%	14.90%	6.30%
DRB3/4/5	DRB5*01:01	14.40%	13.50%	8.70%
DRB3/4/5	DRB3*03:01	9.60%	4.90%	7.50%
DRB3/4/5	DRB5*01:02	0.20%	0.70%	9.70%
DRB3/4/5	DRB5*02:02	1.50%	1.60%	0.70%
At least 1 HLA-DRB345 match	Sum of allele frequencies:	84.60%	85%	81.60%
At least 1 HLA-DRB345 match	Per individual (2n):	97.60%	97.80%	96.60%

Table 1 **HLA allele frequencies by Gragert et al. (2) AAFA, African American; EURCAU, European Caucasian, JAPI, Japanese. Percentages of "At least 1 HLA-A match" are higher per individual than the sum (ΣAFHLA-A) of the allele frequencies (AF) since allele frequencies refer to one chromosome set (1n), with each individual having two chromosome sets (2n). The per-individual (2n) "phenotype frequencies" (PF) indicating the percentage of individuals with at least one HLA-A match with the exogenous HLA-A alleles from the Bria-OTS cell lines were calculated as follows: PFHLA-A = 1 - (1 - ΣAFHLA-A)2, whereby (1-ΣAFHLA-A)2 is the probability that an individual does **not** carry at least 1 of the HLA-A alleles. Example: for African American, PFHLA-A = 1 - (1 - ΣAFHLA-A)2 = 1 – (1 – 49.4%)2 = 74.4%.*

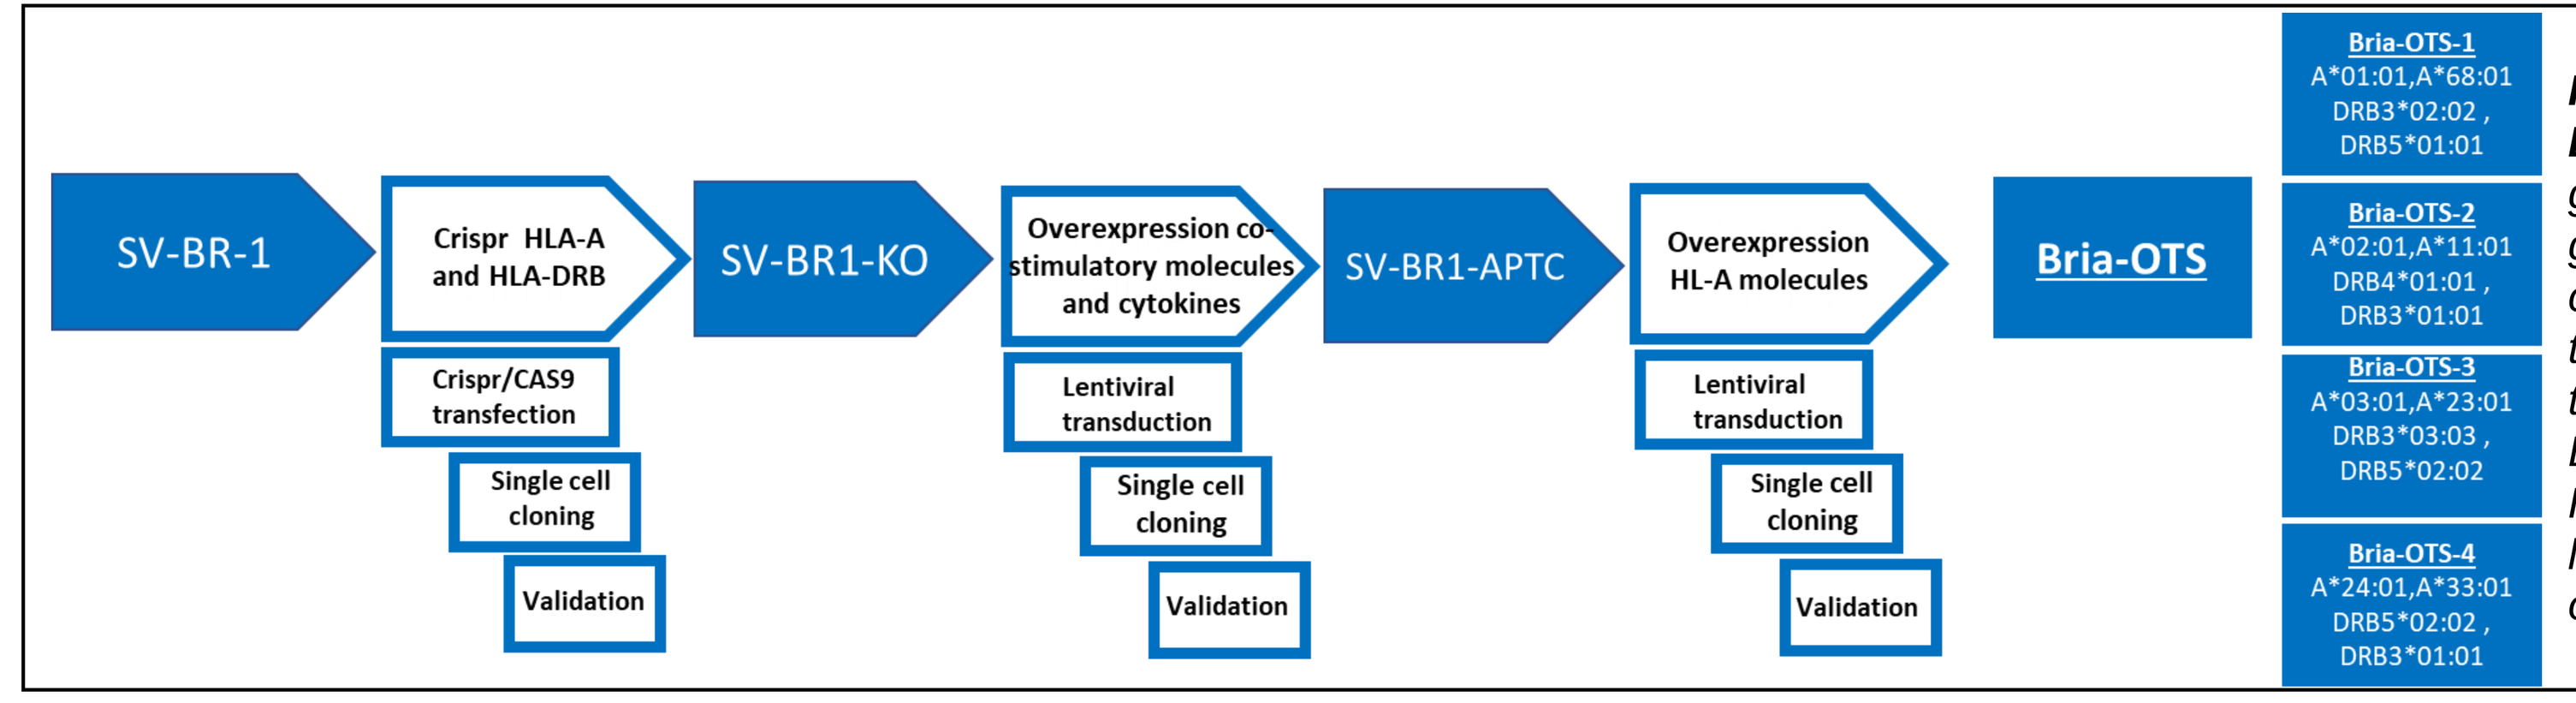
SV-BR-1
Crispr HLA-A and HLA-DRB
Crispr/CAS9 transfection
Single cell cloning
Validation
SV-BR1-KO
Overexpression co-stimulatory molecules and cytokines
Lentiviral transduction
Single cell cloning
Validation
SV-BR1-APTC
Overexpression HL-A molecules
Lentiviral transduction
Single cell cloning
Validation
Bria-OTS
Bria-OTS-1 A*01:01,A*68:01 DRB3*02:02 , DRB5*01:01
Bria-OTS-2 A*02:01,A*11:01 DRB4*01:01 , DRB3*01:01
Bria-OTS-3 A*03:01,A*23:01 DRB3*03:03 , DRB5*02:02
Bria-OTS-4 A*24:01,A*33:01 DRB5*02:02 , DRB3*01:01

Figure 2. Experimental strategy to develop Bria-OTS cell lines. *Endogenous HLA-A/DRB genes are first KO by CRISPR/Cas9 to generate Bria-KO cells. GM-CSF and other cytokines and co-stimulatory molecules are then overexpressed by sequential lentiviral transduction of 4 unique constructs to generate Bria-APTC cells. Lastly, two HLA-A and two HLA-DRB alleles are overexpressed by lentiviral transduction, to generate a collection of 4 cell lines (Bria-OTS-1/2/3/4).*

A

Co-stimulatory molecules and cytokines		
Lentivirus ID	***EF1a Promoter***	***MNDU3 Promoter***
GM-IFN	CSF2 (GM-CSF)	IFNA2 (IFNα)
CD86-IL12	CD86	IL12
CD80-HLA-DRA	CD80	HLA-DRA
IL7-4-1BBL	IL7	4-1BBL (TNFSF9;CD137L)

B

HLA molecules		
Lentivirus ID	***EF1a Promoter***	***MNDU3 Promoter***
A2-A11	HLA-A*02:01	HLA-A*11:01
A1-A68	HLA-A*01:01	HLA-A*68:01
A3-A23	HLA-A*03:01	HLA-A*23:01
A24-A33	HLA-A*24:02	HLA-A*33:03
DR401-301	DRB4*01:01	DRB3*01:01
DR302-501	DRB3*02:02	DRB5*01:01
DR303-502	DRB3*03:01	DRB5*01:02
DR301-522	DRB5*02:02	DRB3*01:01

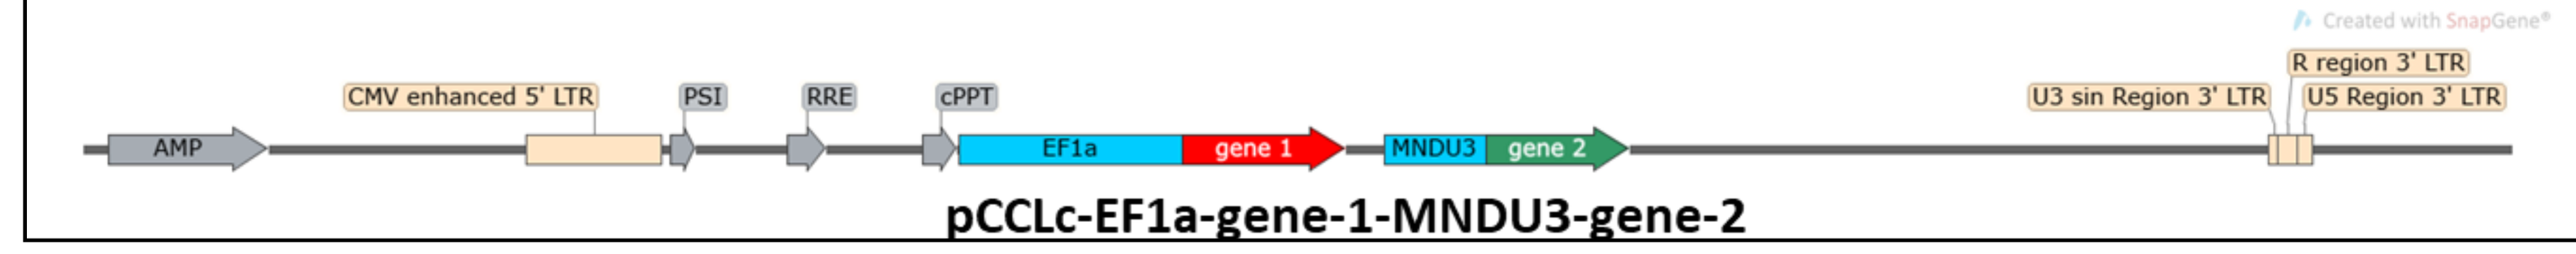
Created with SnapGene®
AMP
CMV enhanced 5' LTR
PSI
RRE
cPPT
EF1a
gene 1
MNDU3
gene 2
U3 sin Region 3' LTR
R region 3' LTR
U5 Region 3' LTR
pCCLc-EF1a-gene-1-MNDU3-gene-2

Fig.3. *Lentiviral vectors used to express two different genes from independent promoters. Cytokines and co-stimulatory molecules (Left Table) used to generate Bria-APTC cells. HLA-A and HLA-DRB alleles (Right Table) used to generate final Bria-OTS+ products.*

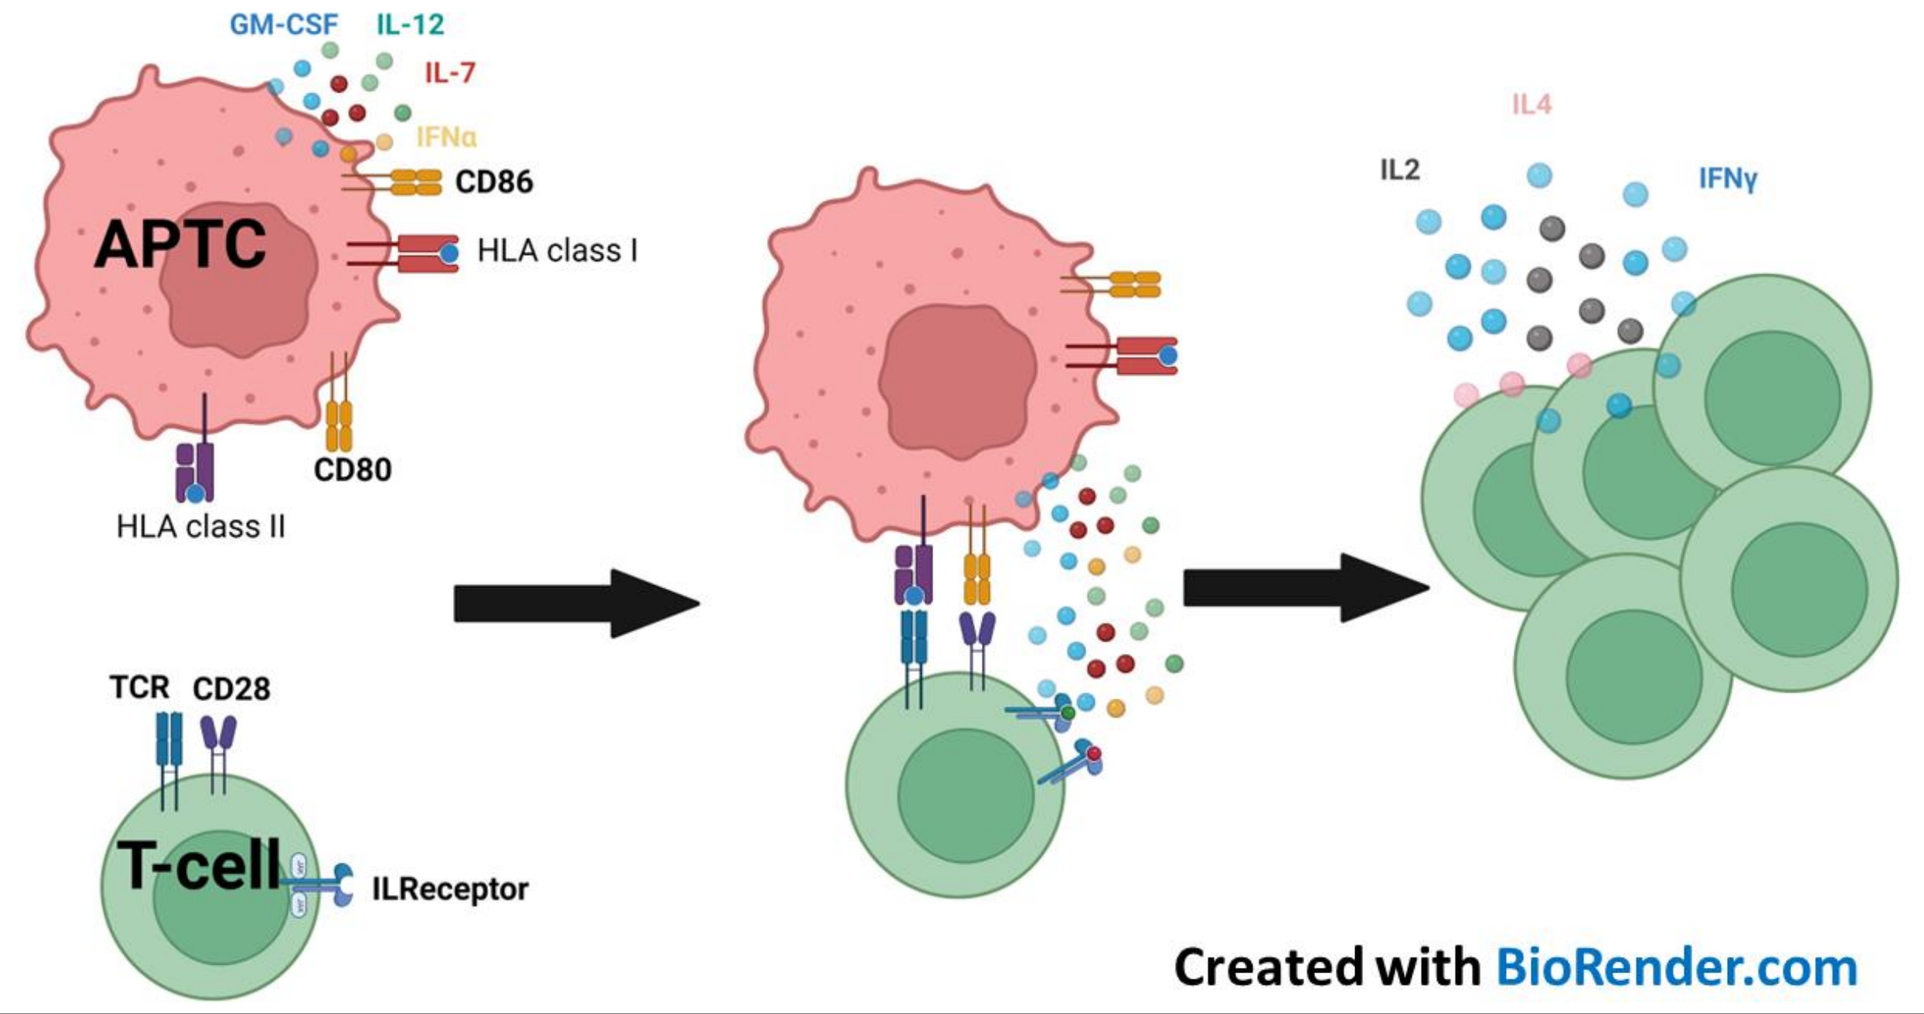
GM-CSF
IL-12
IL-7
IFNα
CD86
APTC
HLA class I
CD80
HLA class II
TCR
CD28
T-cell
ILReceptor
IL4
IL2
IFNγ
Created with BioRender.com

Fig. 4. Modified mixed lymphocyte reaction assay (MLR). *Bria-APTC cells or Bria-KO cells are incubated with PMBCs at different ratios. At days1, 2, 3, and 7, secretion of INFα was measured to assess T cell activation.*

RESULTS

Following sequential CRISPR/Cas9 editing, the SV-BR-1 cells were cloned, and one clone selected for further engineering (SV-BR-1-KO). SV-BR-1-KO was subsequently transduced with 6 lentiviral vectors each expressing 2 genes under the control of separate promoters:CD86-IL12, CD80-HLA-DRA, 4-1BBL-IL7, GM-CSF-IFNa, HLA-allele-1-HLA-allele-2 and HLA-DR-allele-1-HLA-DR-allele-2. Four cell lines were generated with different combinations of HLA alleles. Following selection, cells were evaluated by ELISA, flow cytometry to confirm gene expression (Fig. 5). Using mixed lymphocyte reaction assays we demonstrated that the generated cells stimulate naïve T-cells (Fig. 6).

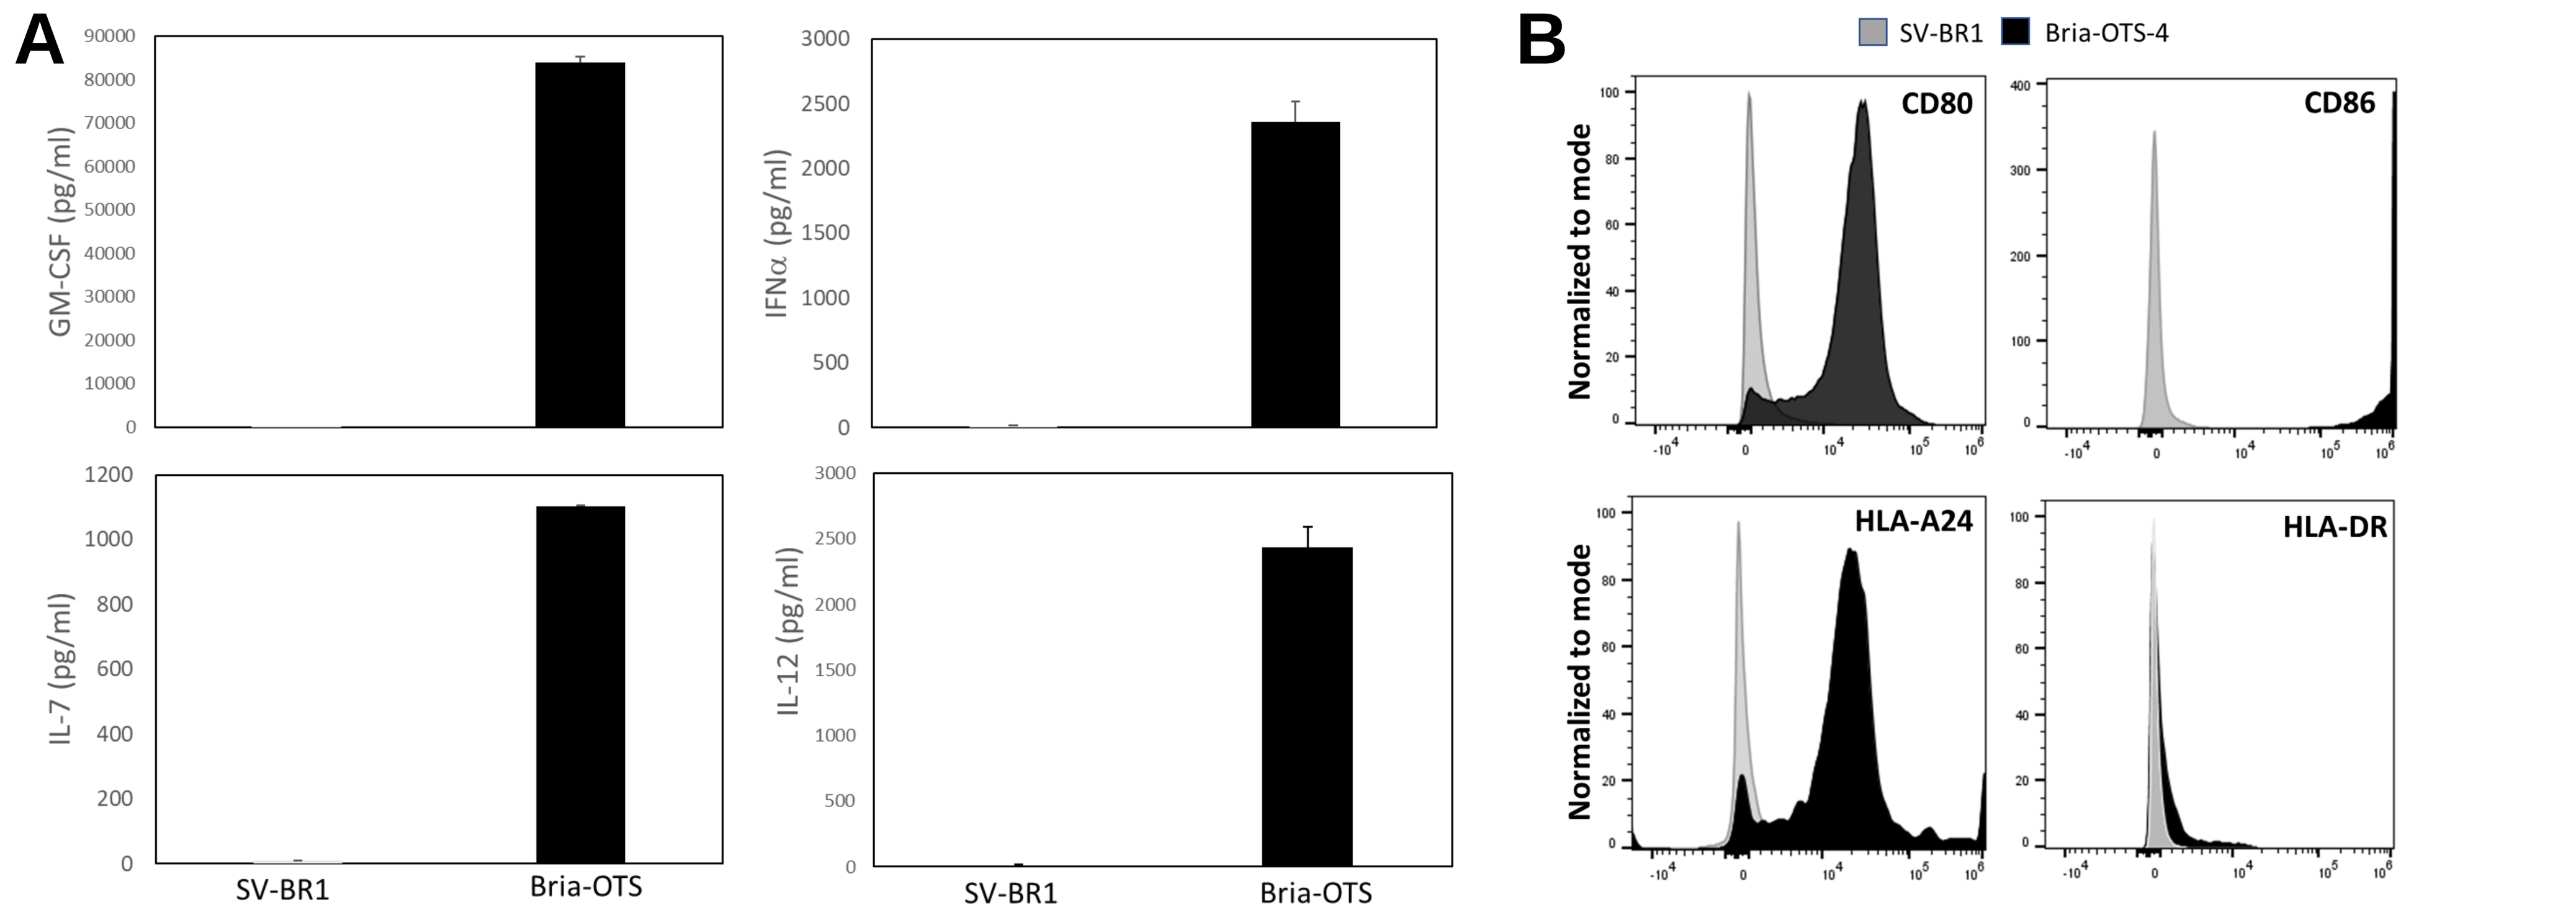
A
GM-CSF (pg/ml)
IFNα (pg/ml)
IL-7 (pg/ml)
IL-12 (pg/ml)
SV-BR1
Bria-OTS
B
SV-BR1
Bria-OTS-4
CD80
CD86
HLA-A24
HLA-DR
Normalized to mode

Figure 5. Expression of cytokines/co-stimulatory molecules in Bria-OTS-4 cells. *SV-BR-1-KO was sequentially transduced with lentiviruses expressing 8 different cytokines and co-stimulatory molecules* ***A)*** *Levels of secreted GM-CSF, INFα, IL-12, and IL-7 were measured by ELISA.* ***B)*** *Expression of surface receptors CD86 and CD80, HLA-A24 and HLA-DR in Bria-OTS cells as measured by flow cytometry.*

Conclusion: ***Tumor cell lines that express co-stimulatory molecules and immuno-modulatory cytokines have been developed.***

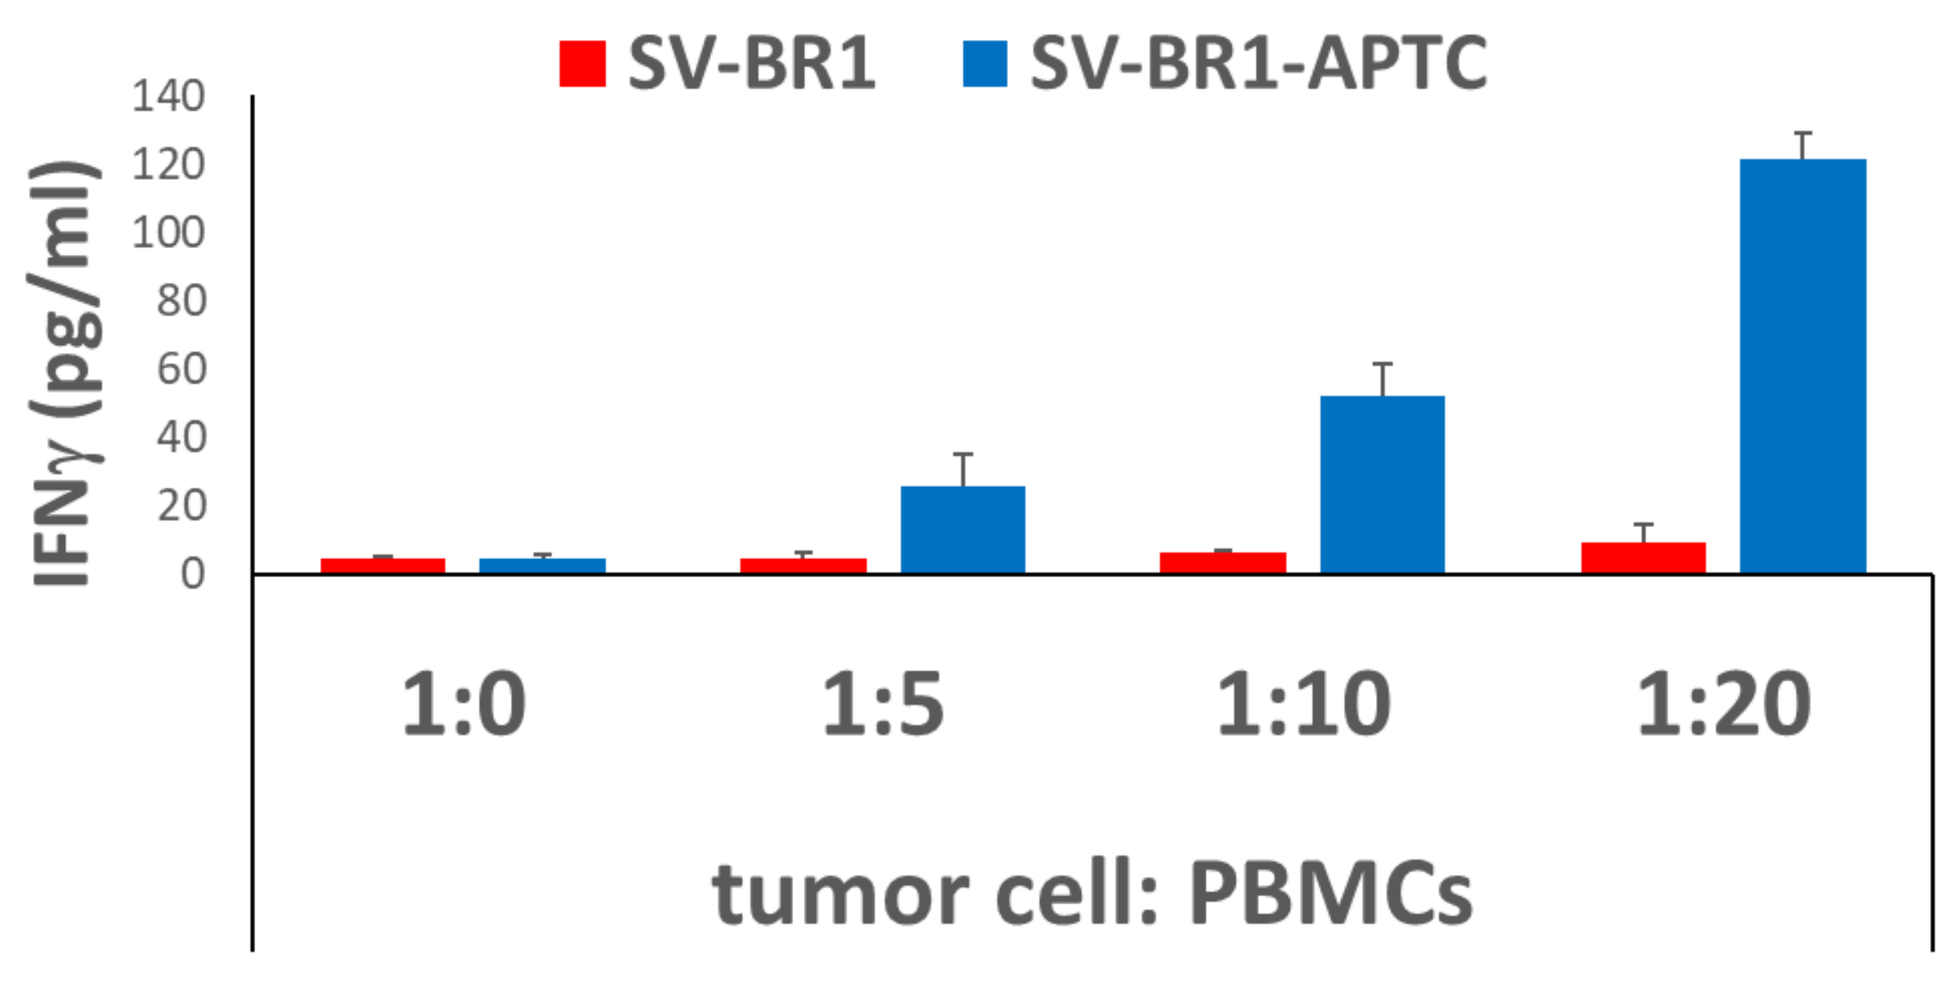
SV-BR1
SV-BR1-APTC
IFNγ (pg/ml)
1:0
1:5
1:10
1:20
tumor cell: PBMCs

Figure 6. Functional validation of SV-BR-1-derived Bria-APTC cells.
A) *SV-BR-1 or SV-BR-1-APTC cells (10,000/well in a 96 well plate) were incubated alone or with PBMCs at the indicated ratios for 24 hours. IFNγ levels were measured in the supernatant by ELISA. N = 3.*

Conclusion: ***Tumor cell lines that express co-stimulatory molecules and immuno-modulatory cytokines are able to activate/prime naïve T-cells***

DISCUSSION AND CONCLUSIONS

Cancer immunotherapy relies on the activation of the patient's immune system to eliminate cancer cells. Despite great advances, current immunotherapies are effective only on a very limited number of patients. Different types of cancer vaccines have been developed with moderate success, often hampered by lack of strong immunogenicity and complex manufacturing. BriaCell's Bria-OTS provides a solution by increasing vaccine immunogenicity and decreasing manufacturing costs while personalizing the therapy based on matching the patient's HLA type.

References:

1. Lacher MD et al, Front Immunol. 2018 May 15;9:776
2. Gragert L, Madbouly A, Freeman J, Maiers M. Six-locus high resolution HLA haplotype frequencies derived from mixed-resolution DNA typing for the entire US donor registry. *Hum Immunol.* 2013;74(10):1313-1320

This presentation is the intellectual property of the author/presenter. Contact them at info@briacell.com for permission to reprint and/or distribute.